Poster 1626

IDWeek 2020
22–25 October, 2020

Synergistic Effect of Cefiderocol with Other Antibiotics Against PER-Producing *Acinetobacter baumannii* Isolates from the Multinational SIDERO-WT Studies

Y Yamano,[1] M Takemura,[1] N Anan,[1] R Nakamura,[2] R Echols,[3]

[1]Shionogi & Co. Ltd, Osaka, Japan; [2]Shionogi Techno Advance Research, Osaka, Japan; [3]Infectious Disease Drug Development Consulting LLC, Easton, CT, USA

Contact information:
Yoshinori Yamano
3-1-1, Futaba-cho, Toyonaka,
Osaka, 561-0825, Japan
Phone: +81-6-6331-6293
Fax: +81-6-6331-8612
Email: yoshinori.yamano@shionogi.co.jp

Abstract (Revised)

Background: Cefiderocol (CFDC), a novel siderophore cephalosporin, showed potent activity at minimum inhibitory concentrations (MICs) of ≤4 μg/mL against ≥99% of Gram-negative isolates in the multinational SIDERO-WT studies. PER-producing *Acinetobacter baumannii*, mainly from Russia, showed high CFDC MICs of 8 to >64 μg/mL. This study evaluated the synergistic effects of CFDC combined with other antibiotics against *PER-producing A. baumannii* isolates with high CFDC MICs.

Methods: Two isolates of PER-producing *A. baumannii* with resistance to CFDC (MIC 16 μg/mL), meropenem (MEM; MIC 64 μg/mL), ceftazidime-avibactam (CZA; MIC 64/4 μg/mL), amikacin (AMK; MIC 32 or 64 μg/mL), and ciprofloxacin (CIP; MIC ≥64 μg/mL) were tested. Against ampicillin-sulbactam (SAM), one isolate was resistant (MIC 32/64 μg/mL) and another was susceptible (MIC 8/16 μg/mL). Effects of CFDC combined with other antibiotics were evaluated by checkerboard assay and chemostat model reproducing humanized antibiotic exposure. The checkerboard assay used a single agent (e.g. ceftazidime [CAZ], avibactam [AVI], ampicillin [AMP] or sulbactam [SUL]). Iron-depleted cation-adjusted Mueller–Hinton broth was used as the standard medium for CFDC, as recommended by the Clinical Laboratory and Standards Institute.

Results: Against both isolates, synergy with CFDC was seen for two β-lactamase inhibitors, AVI and SUL, with a fractional inhibitory concentration (FIC) index of 0.033–0.047 and 0.26–0.27, respectively. A synergistic to additive effect was seen for MEM and AMK, with an FIC index of 0.53–0.75 and 0.25–0.52, respectively. In the chemostat model, regrowth during 24-h treatment was observed with single agents (CFDC 2 g, q8h, 3-h infusion; MEM 2 g, q8h, 1-h infusion; CZA 2 g, q8h, 2-h infusion; SAM 3 g, q8h, 3-h infusion; AMK 15 mg/kg, q8h, 3-h infusion) for both isolates, including the SAM-susceptible isolate. However, no regrowth was seen when CFDC was combined with CZA, MEM, SAM or AMK.

Conclusions: The most potent synergy was seen between CFDC and AVI against PER-producing *A. baumannii* with decrease of MIC to ≤1 μg/mL for the 2 isolates tested, followed by SUL and MEM. Under humanized pharmacokinetic exposure, combination of CFDC and CZA, MEM, SAM or AMK is expected to be effective against PER-producing *A. baumannii* with high CFDC MICs.

Introduction

Cefiderocol (CFDC) is a novel siderophore cephalosporin with activity against a wide variety of Gram-negative bacteria, including carbapenem-resistant Enterobacterales and non-fermenters.

Cefiderocol has been approved in the USA for the treatment of patients with complicated urinary tract infections (cUTI) and hospital-acquired pneumonia and ventilator-associated pneumonia caused by Gram-negative bacteria and in Europe for the treatment of infections due to Gram-negative pathogens with limited treatment options [1, 2].

We have conducted consecutive multinational surveillance studies (SIDERO-WT) to evaluate the in vitro activity of CFDC against clinical isolates collected in Europe and North America [3], showing that ≥99% of the isolates were susceptible to cefiderocol with MIC of ≤4 μg/mL based on the CLSI investigational breakpoint [4].

In this study, we evaluated the in vitro bactericidal activity against high MIC isolates of CFDC in the combination with other anti-Gram-negative agents.

Materials and Methods

Bacterial strains

- Among a total of 9,205 Gram-negative pathogens isolated which were collected from North America and Europe from 2014 Nov to 2015 Oct (SIDERO-WT-2014 study), 39 isolates showed MIC of ≥8 μg/mL, which are non-susceptible to CFDC based on the CLSI investigational breakpoint (Fig. 1).
- Among these 39 isolates, 28 isolates were *Acinetobacter baumannii*, among which 25 isolates harbored *bla*$_{PER}$ gene. Among these 25 *bla*$_{PER}$ -possessing *A. baumannii*, 18 isolates were collected in Russia and 6 isolates were collected in Turkey [5] (Fig. 2).
- Two *bla*$_{PER}$ -possessing *A. baumannii* isolates from Russia and Turkey were used in this study. The β-lactamase genes were identified by whole genome sequencing analysis.

Materials and Methods (Continued)

Determination of MIC and fractional inhibitory concentration index (FIC index)

- MICs of CFDC, meropenem (MEM), ceftazidime-avibactam (CZA), ampicillin-sulbactam (SAM) and amikacin (AMK) were determined using the broth microdilution methodology recommended by CLSI. For MIC determination of CFDC, iron-depleted cation-adjusted Muller Hinton broth (ID-CAMHB) was used. The susceptibility interpretive criteria determined by CLSI was used.
- To evaluate the combination effect between CFDC and each antibiotic, FIC index was determined by using checkerboard methods using ID-CAMHB. In case of CZA and SAM, the FIC index was determined between CFDC and each of single compound (ceftazidime, avibactam, ampicillin or sulbactam).

In vitro chemostat models

- The bactericidal activity against 2 isolates under humanized PK exposure was evaluated using chemostat models. The PK profiles which occurred by the dosing regimen shown in **Table 1** were reproduced in this study.
- The bacterial suspension was incubated under the humanized PK exposure for 24 hours with the initial inoculum of 3 × 10^5 to 1 × 10^6 CFU/mL, and the viable cells were counted every 2 hours.
- The susceptibility ratio was also determined by a subset of each carbapenemase producer of each bacterial species.

Results

- Both of 2 isolates of *bla*$_{PER}$ possessing *A. baumannii* from Russia and Turkey showed resistance to CFDC, CZA, MEM and AMK. Against SAM, one isolate 1121552 from Russia was susceptible with MIC of 8 μg/mL and isolate 1247951 was resistant with MIC of 32 μg/mL (**Table 2**).
- The most potent synergistic effect was observed in the combination use with avibactam with FIC index of ≤0.05, followed by the combination use with sulbactam with FIC index of 0.26 (**Table 3**).
- The additive effect was observed in the combination use with AMK or MEM although synergistic effect was observed for 1 isolate in the combination use with AMK (**Table 3**).
- The ≥2-log reduction of viable bacterial cells of both isolates were observed for initial 6 hours under the humanized PK exposure with single use of CFDC, CZA, MEM and AMK. However, the bacterial reduction was not observed even for a susceptible isolate under the human PK exposure of SAM. In all cases, bacterial regrowth was observed during the incubation under human PK exposure for 24 hours (**Fig. 3-1**, **Fig. 4-1**).
- The potent synergistic effects were observed for each case of the combinations with CFDC. Under the human PK exposure in the combination use of CFDC plus CZA, SAM, MEM or AMK caused ≥3-log reduction of bacterial cells within 6 hours and no significant regrowth was not observed for 24 hours (**Fig. 3-2**, **Fig. 4-2**).

Table 1. Dosing regimen which were used to recreate humanized PK in chemostat models

Drugs	Dosing regimens used in this study
Cefiderocol	2 g, q8h, 3h-infusion
Ceftazidime/avibactam	CAZ 2 g/AVI 0.5 g, q8h, 2h-infusion
Ampicillin/sulbactam	ABPC 3 g/SUL 1.5 g, q8h, 1h-infusion
Meropenem	MEM 2 g, q8h, 1h-infusion
Amikacin	AMK 1050mg (15 mg/kg), q8h, 1h-infusion

References

1. Fetroja [cefiderocol] for injection, for intravenous use. Prescribing Information. Shionogi Inc., Florham Park, NJ, USA. 2020.
2. Fetcroja [cefiderocol]. 1 g powder for concentrate for solution for infusion. Summary of Product Characteristics. Shionogi B.V., Kingsfordweg 151, 1043 GR, Amsterdam, Netherlands. 2020.
3. Hackel MA, et al. Antimicrob Agents Chemother 2017;61:pii:e00093-17.
4. CLSI. CLSI supplement M100: M100–S30. Wayne, PA: CLSI; 2020.
5. Kohira N et al. J Glob Antimicrob Re 2020; 22: 738.

Results

Figure 1. MIC distribution of each bacterial species isolates collected in SIDERO-WT-2014 study

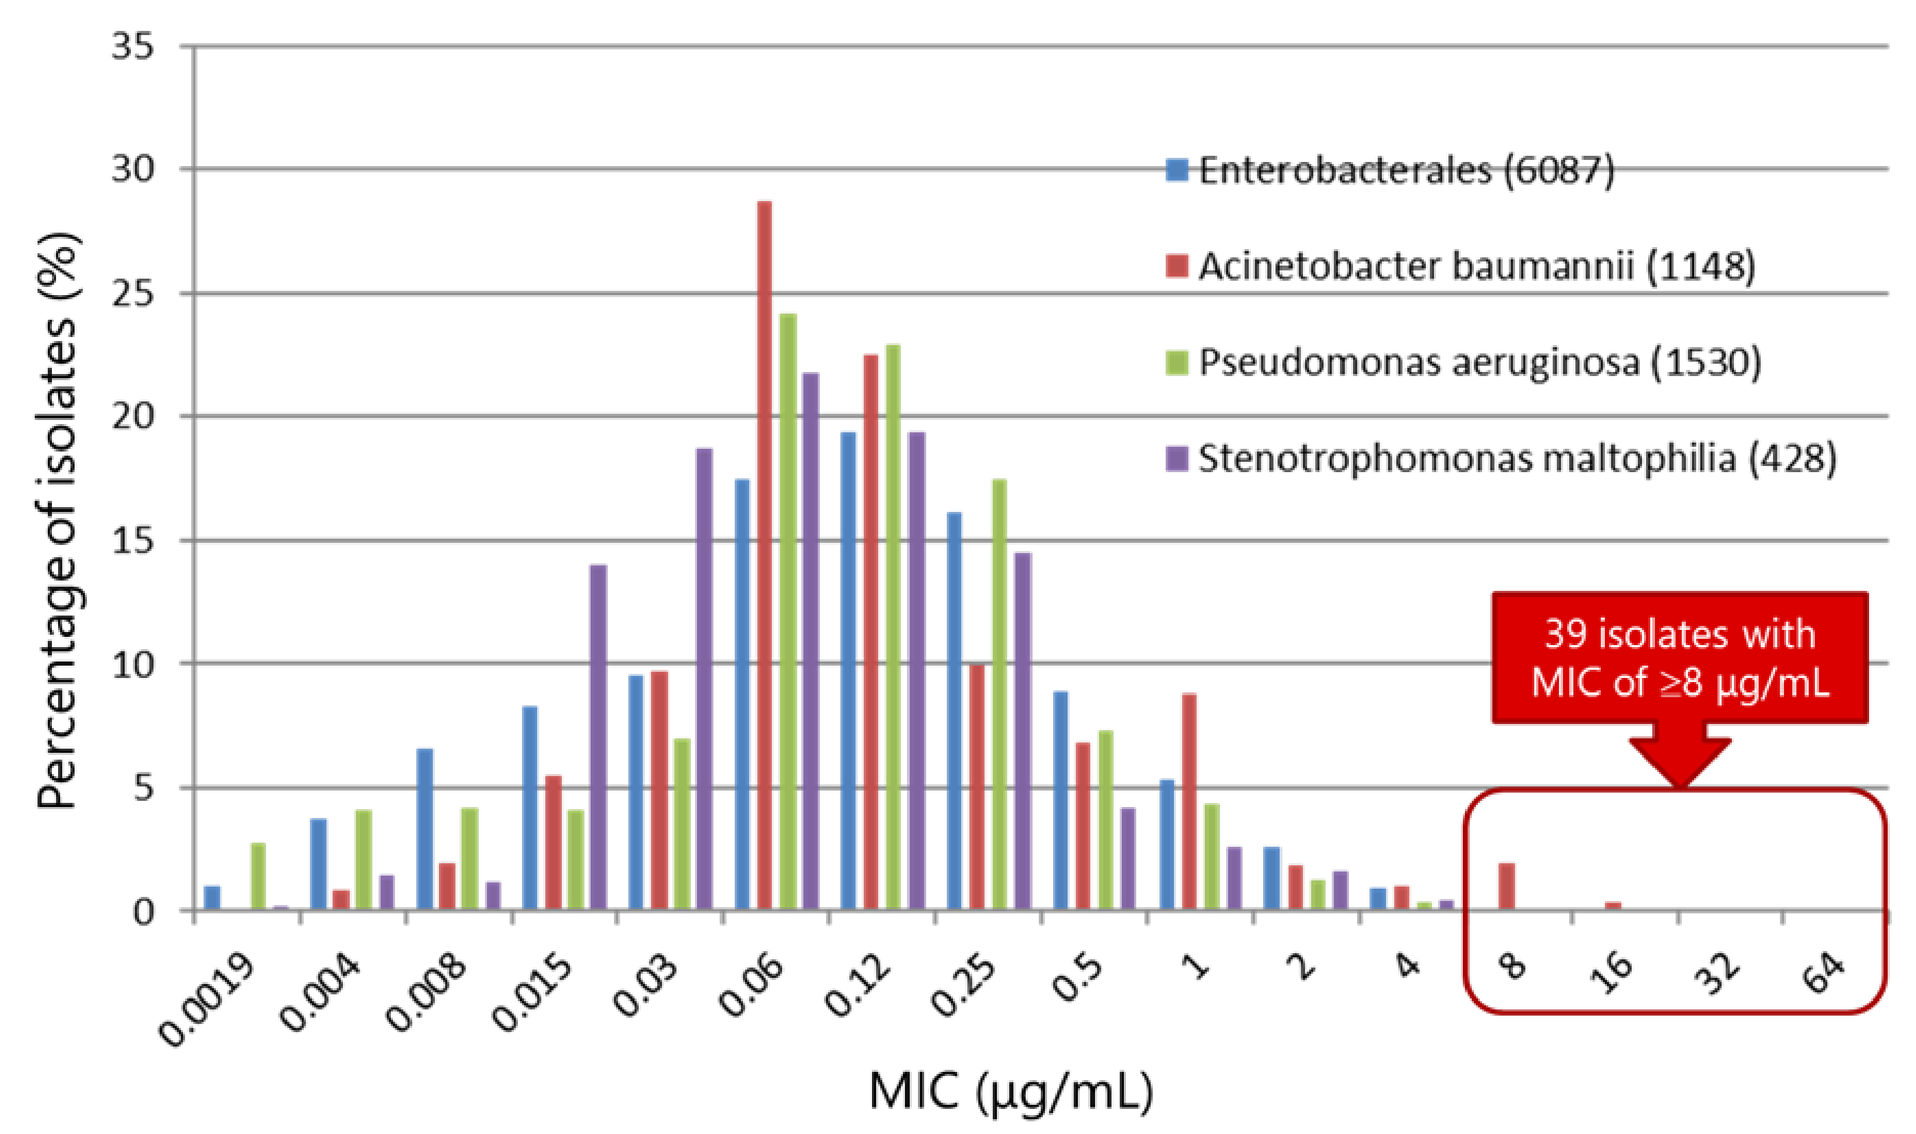

Percentage of isolates (%)
MIC (μg/mL)
Enterobacterales (6087)
Acinetobacter baumannii (1148)
Pseudomonas aeruginosa (1530)
Stenotrophomonas maltophilia (428)
39 isolates with MIC of ≥8 μg/mL
0.0019
0.004
0.008
0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
32
64

Figure 2. The profile of the isolates with CFDC MIC of ≥8 μg/mL

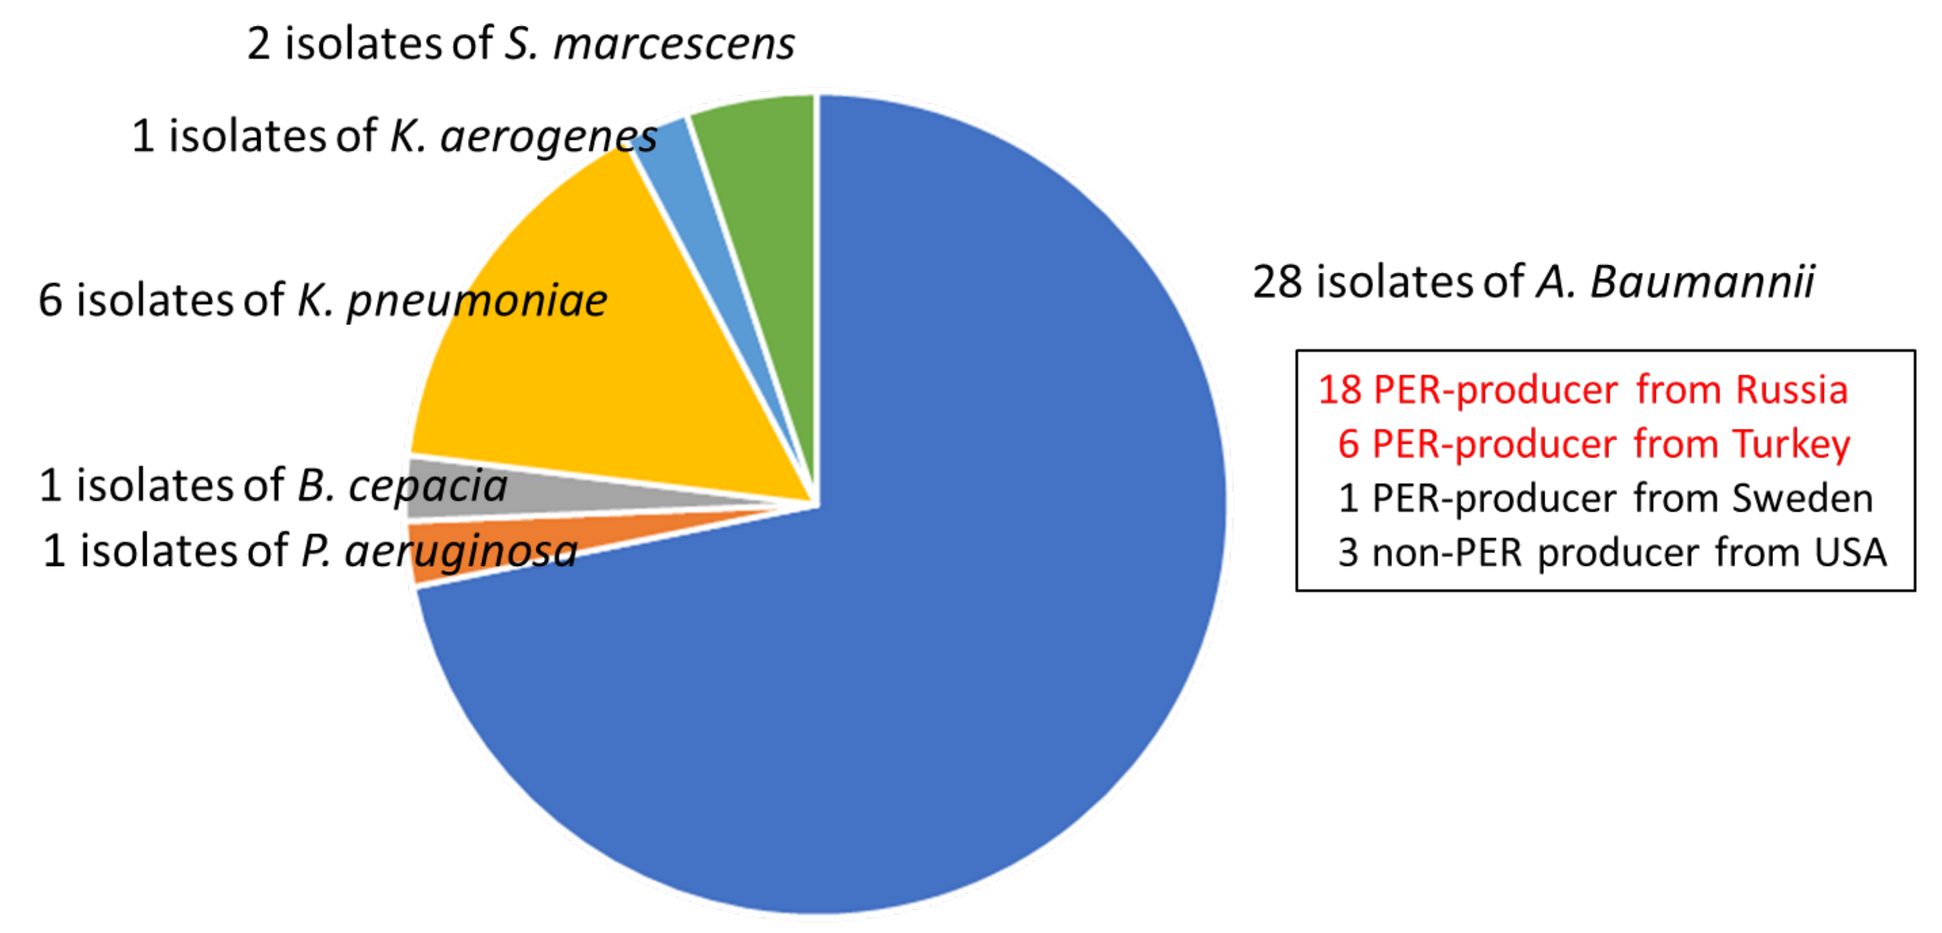

2 isolates of *S. marcescens*
1 isolates of *K. aerogenes*
6 isolates of *K. pneumoniae*
1 isolates of *B. cepacia*
1 isolates of *P. aeruginosa*
28 isolates of *A. Baumannii*
18 PER-producer from Russia
6 PER-producer from Turkey
1 PER-producer from Sweden
3 non-PER producer from USA

Table 2. MIC of the test isolates

Strain (Source)	MIC (μg/mL) of					β-lactamase profile
	cefiderocol	meropenem	Ampicillin/ sulbactam	Ceftazidime/ avibactam	amikacin	
1247951 (Turkey)	16	64	32	64	32	PER-1, ADC-25, OXA-23, OXA-66
1121552 (Russia)	16	64	8	64	64	PER-1, ADC-25, OXA-23, OXA-66

Table 3. Combination effect between CFDC and other anti-Gram-negative antibiotics

Strains	FIC index between cefiderocol and					
	Ceftazidime	Avibactam	Ampicillin	Sulbactam	Meropenem	Amikacin
1247951	2	0.047	2	0.266	0.531	0.25
1121552	2	0.033	2	0.265	0.75	0.516

Figure 3-1. Bactericidal activity against PER1-producing *A. baumannii* 1247951 under human PK of each antibiotic

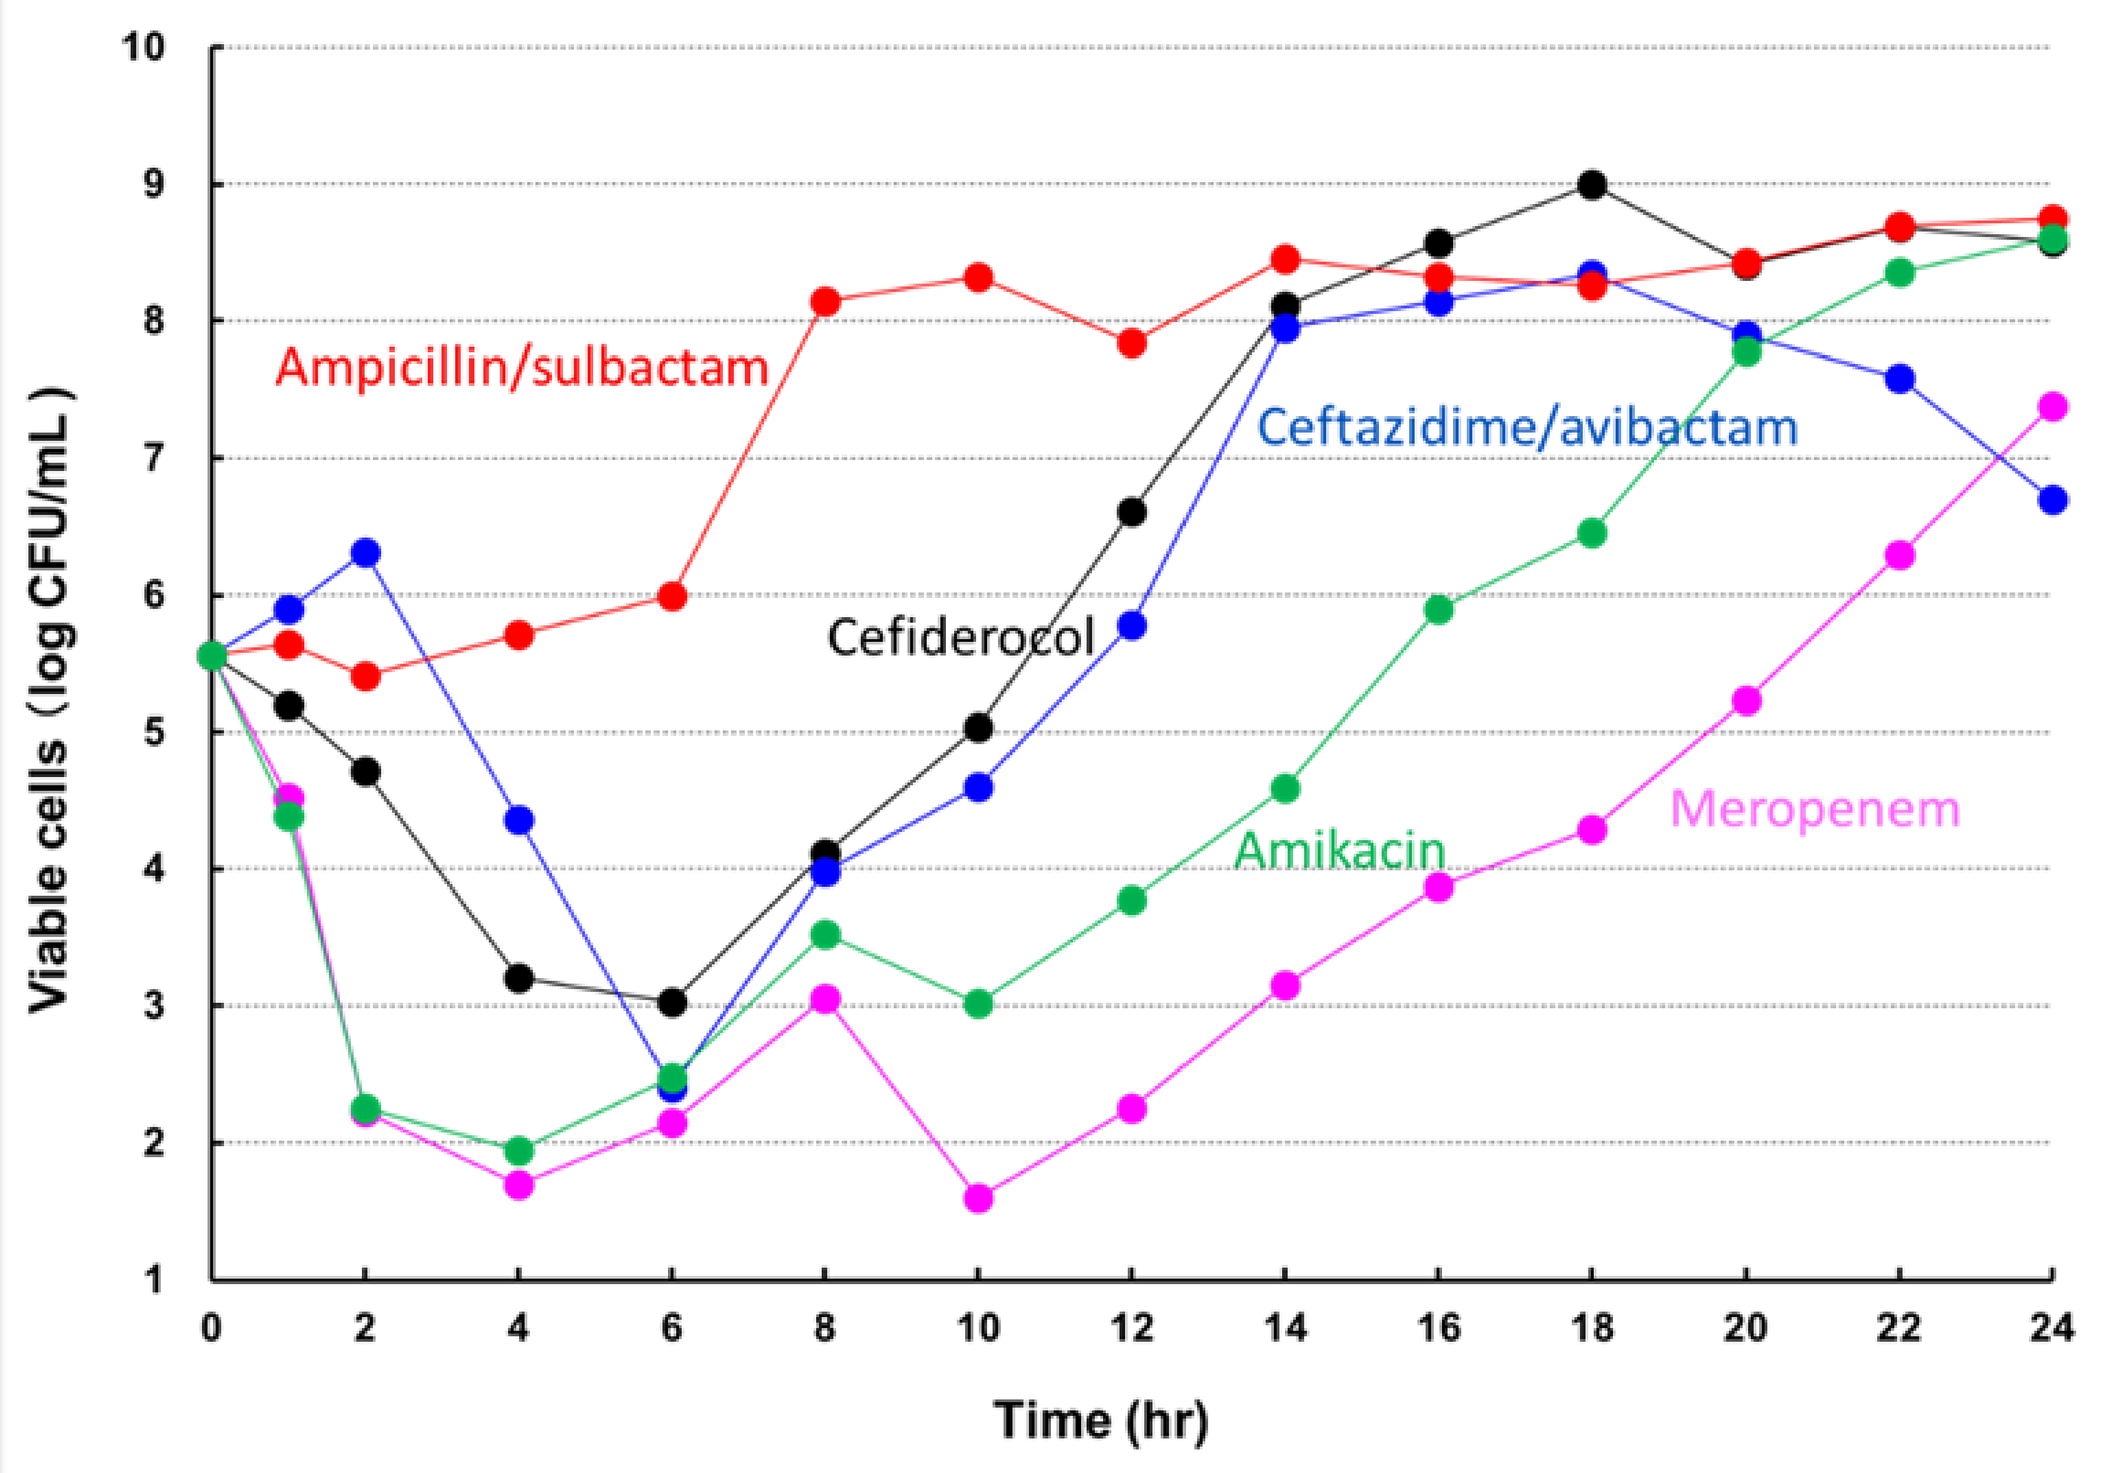

Viable cells (log CFU/mL)
Time (hr)
Ampicillin/sulbactam
Ceftazidime/avibactam
Cefiderocol
Amikacin
Meropenem

Figure 3-2. Bactericidal activity against PER1-producing *A. baumannii* 1247951 of the combination use of CFDC and other antibiotics

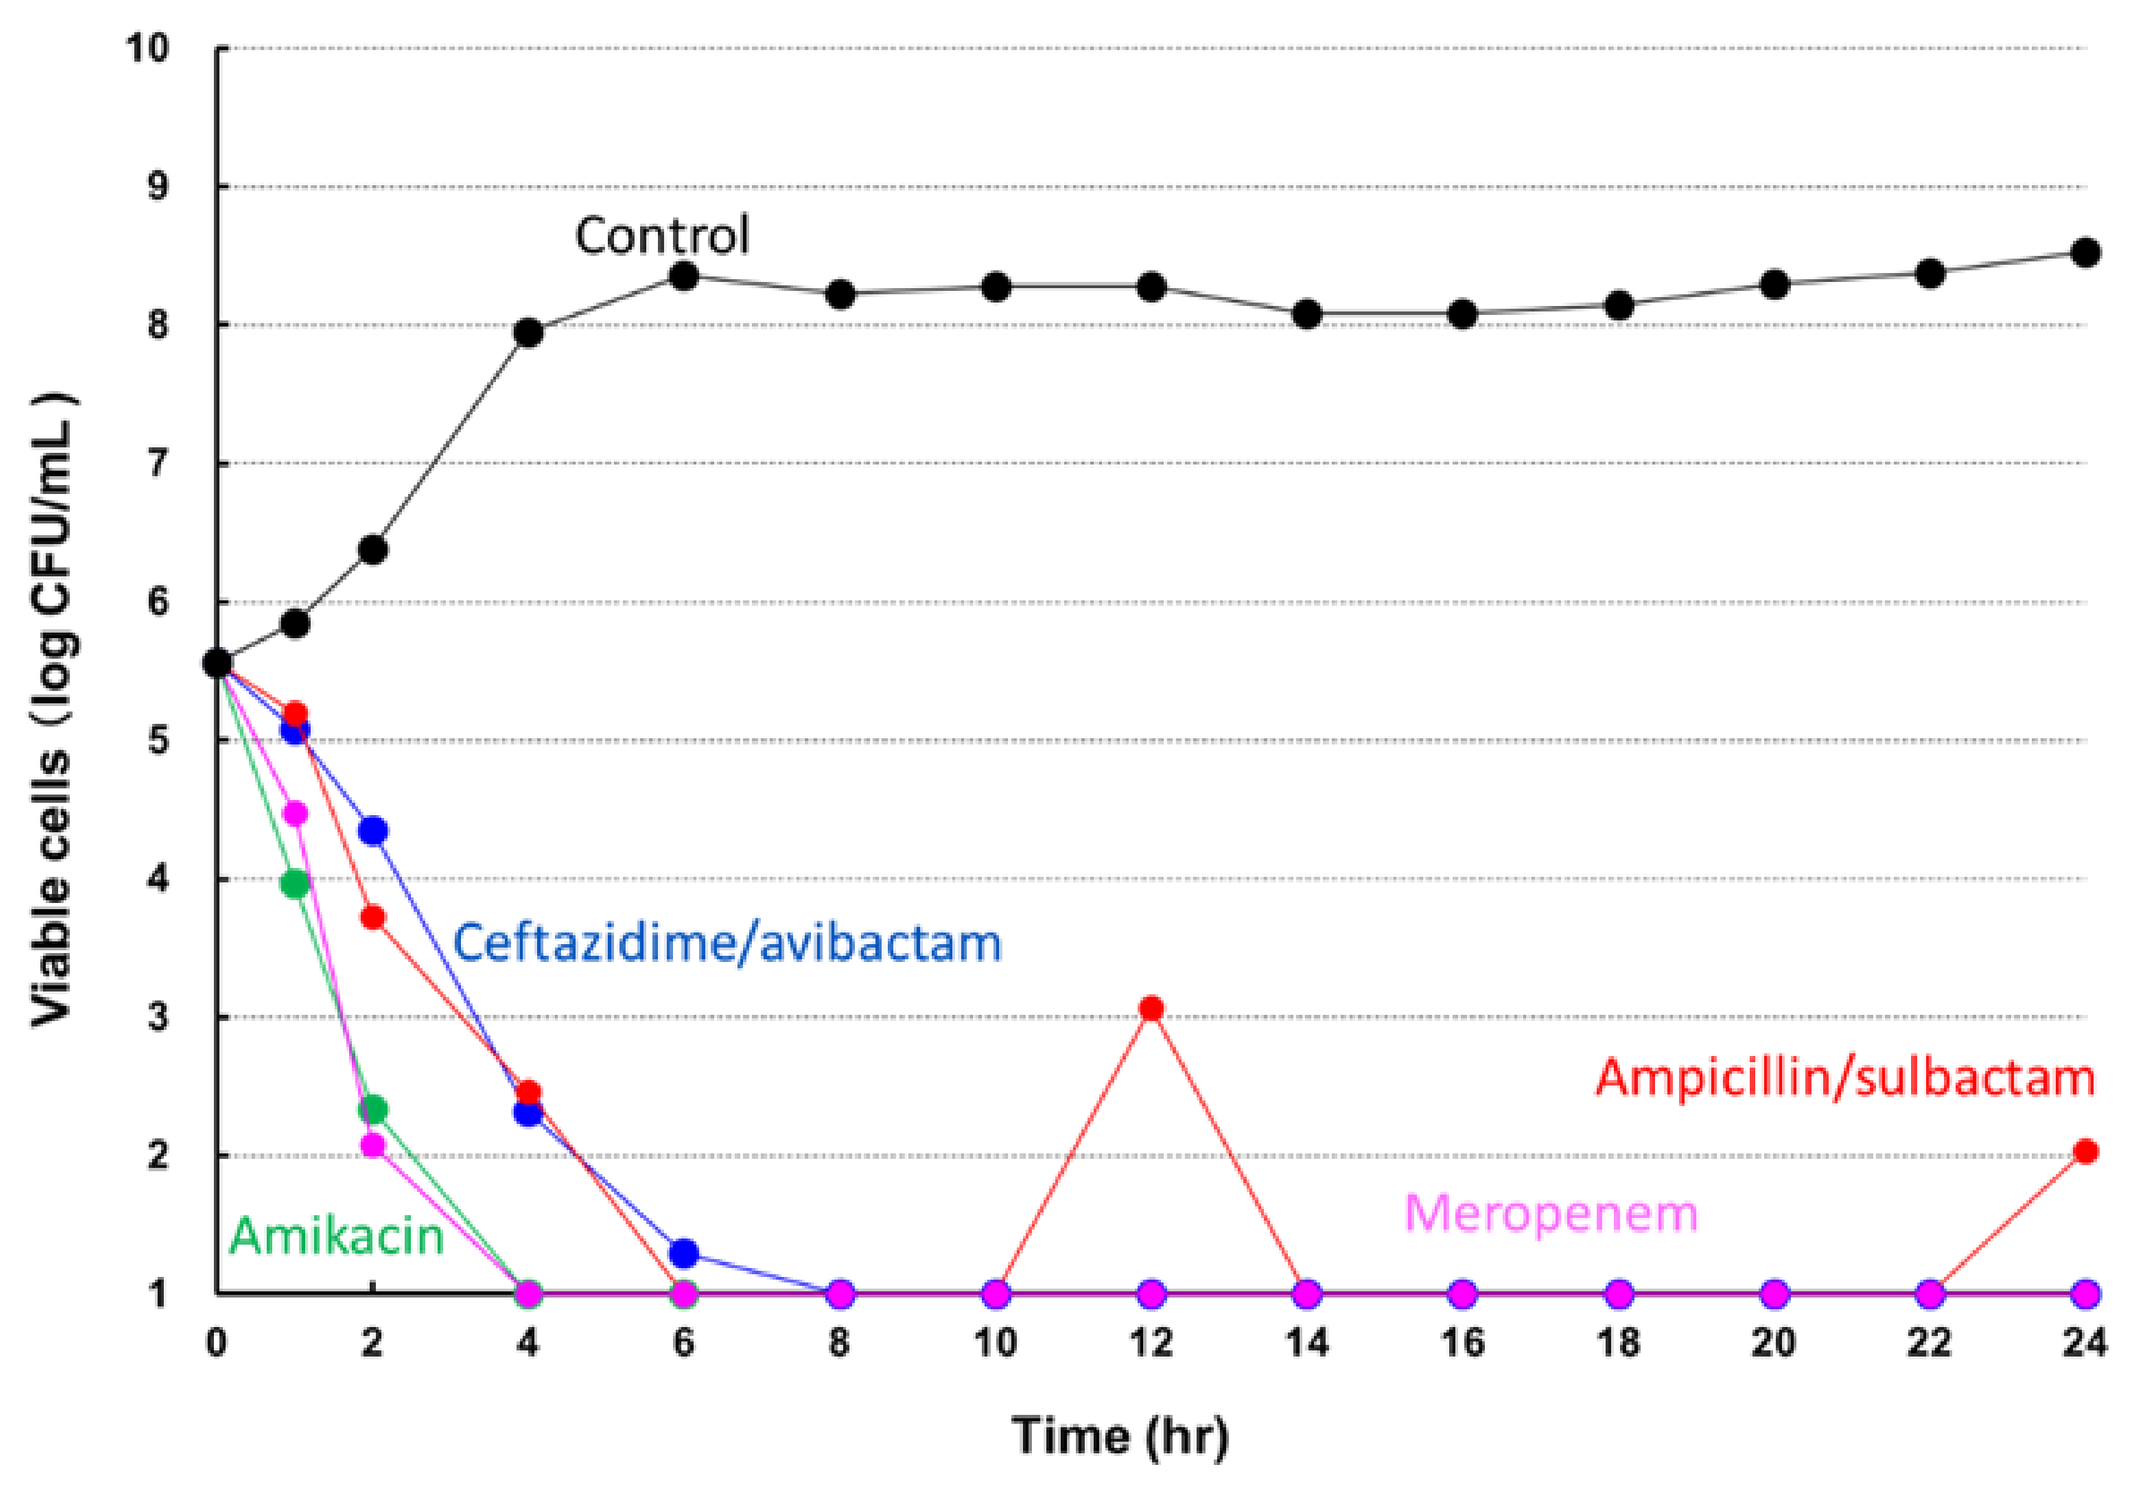

Viable cells (log CFU/mL)
Time (hr)
Control
Ceftazidime/avibactam
Ampicillin/sulbactam
Meropenem
Amikacin

Figure 4-1. Bactericidal activity against PER1-producing *A. baumannii* 1121552 under human PK of each antibiotic

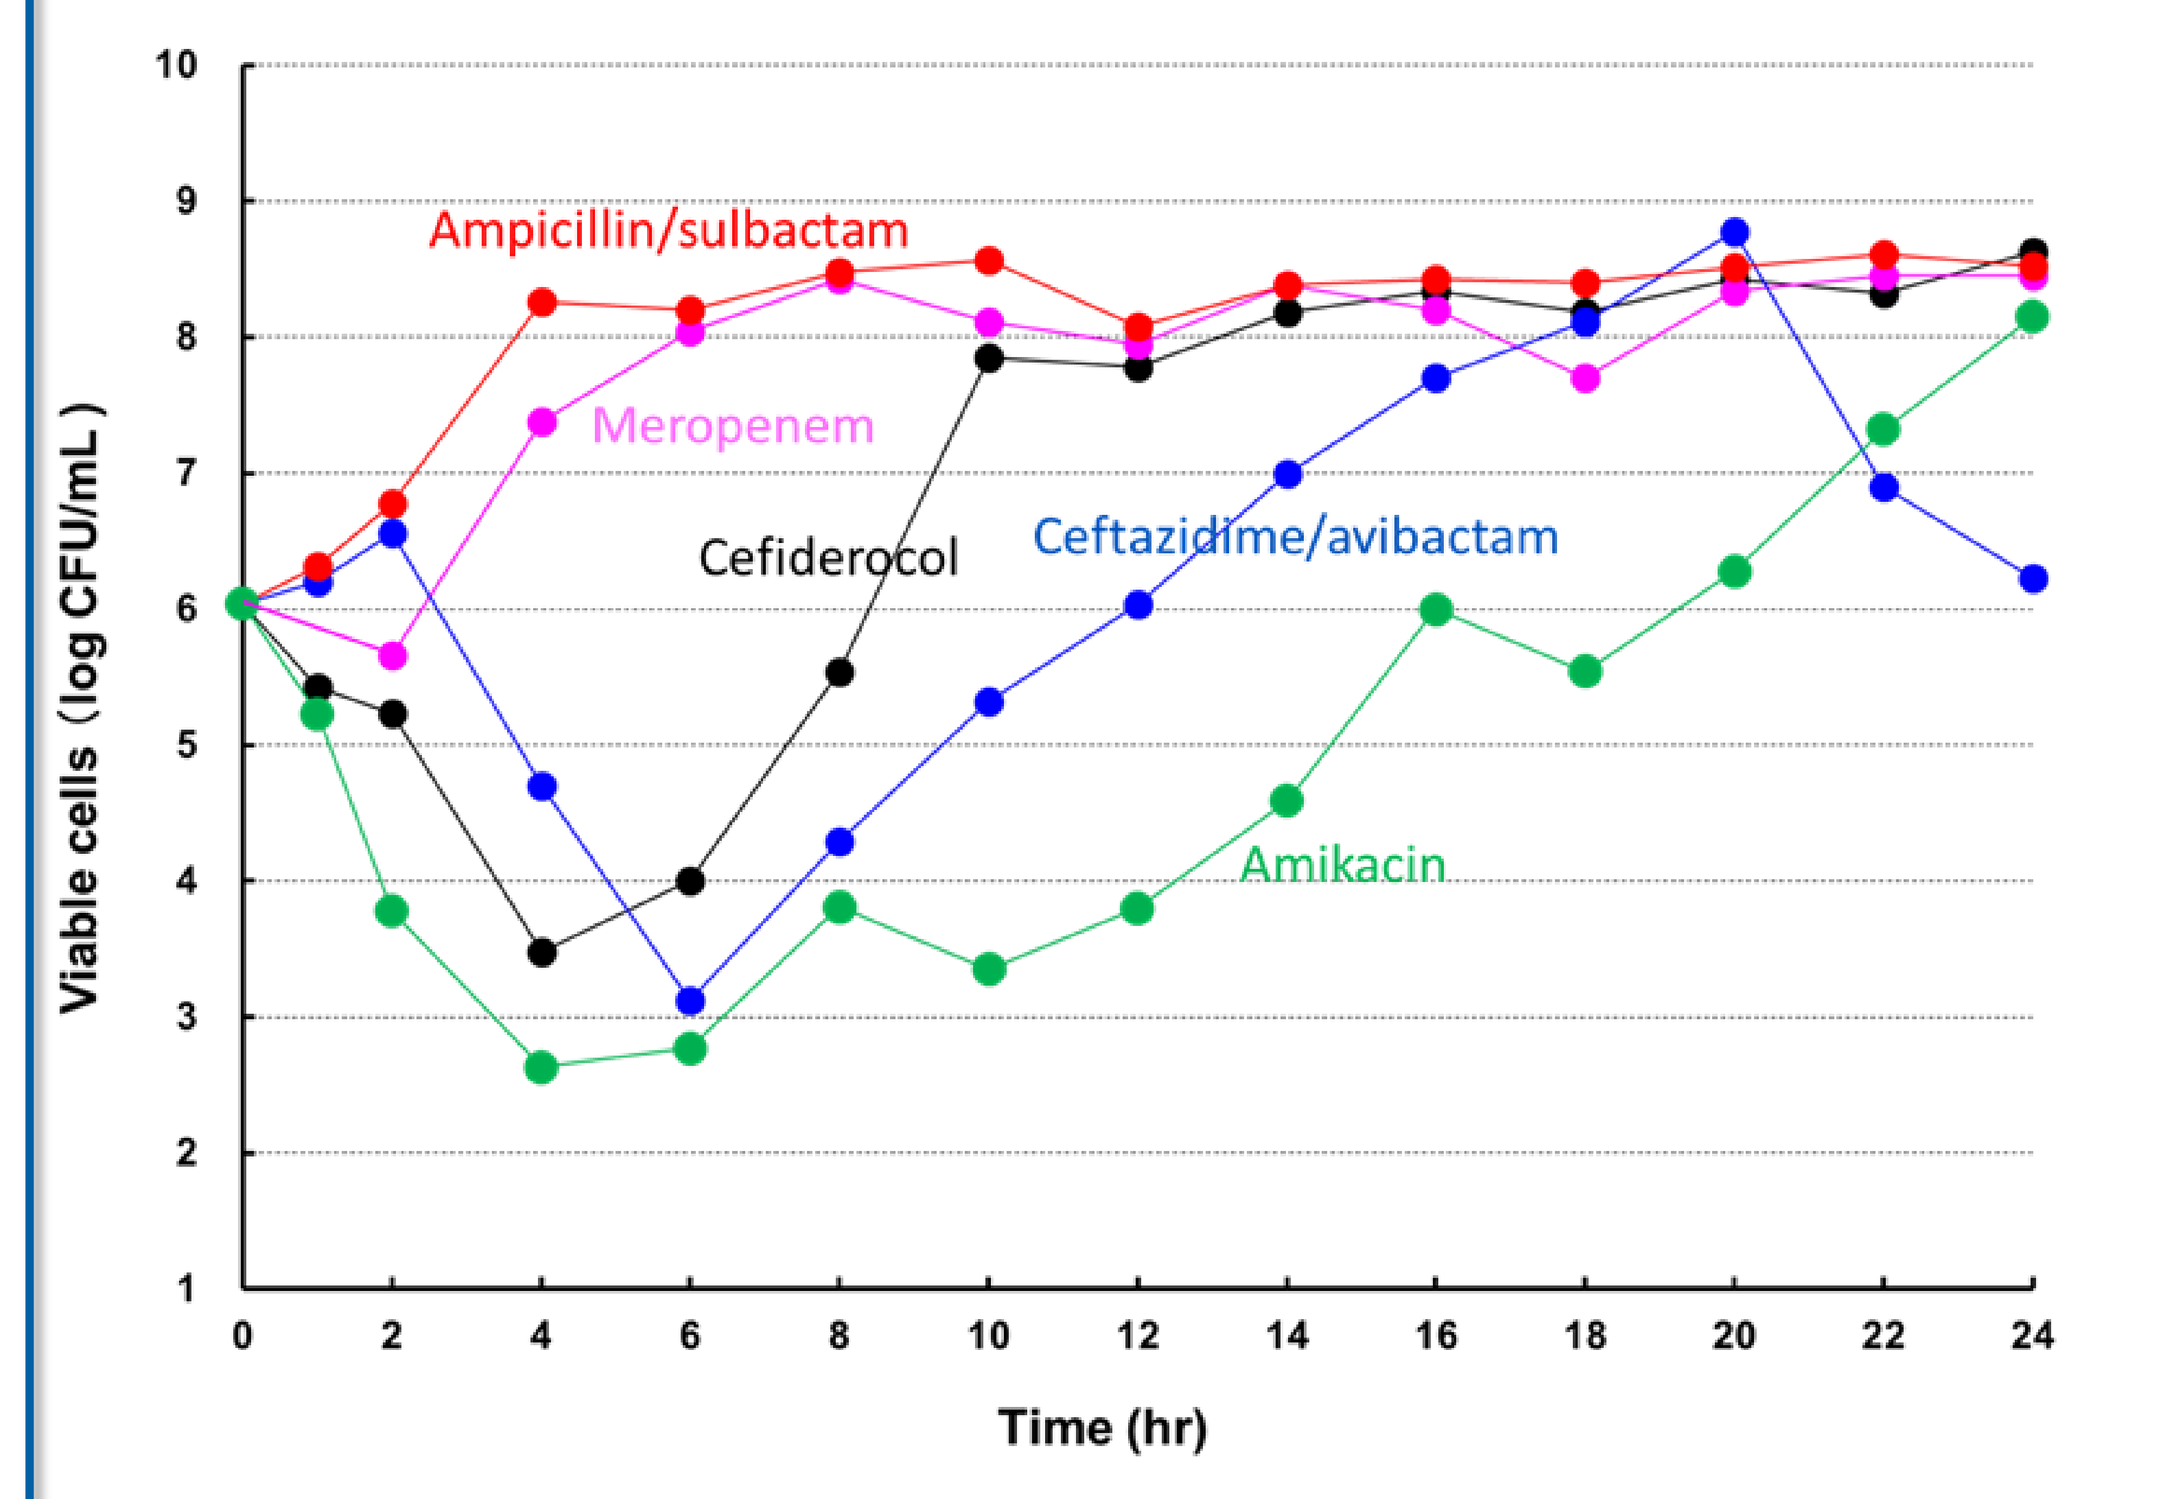

Viable cells (log CFU/mL)
Time (hr)
Ampicillin/sulbactam
Meropenem
Cefiderocol
Ceftazidime/avibactam
Amikacin

Figure 4-2. Bactericidal activity against PER1-producing *A. baumannii* 1121552 of the combination use of CFDC and other antibiotics

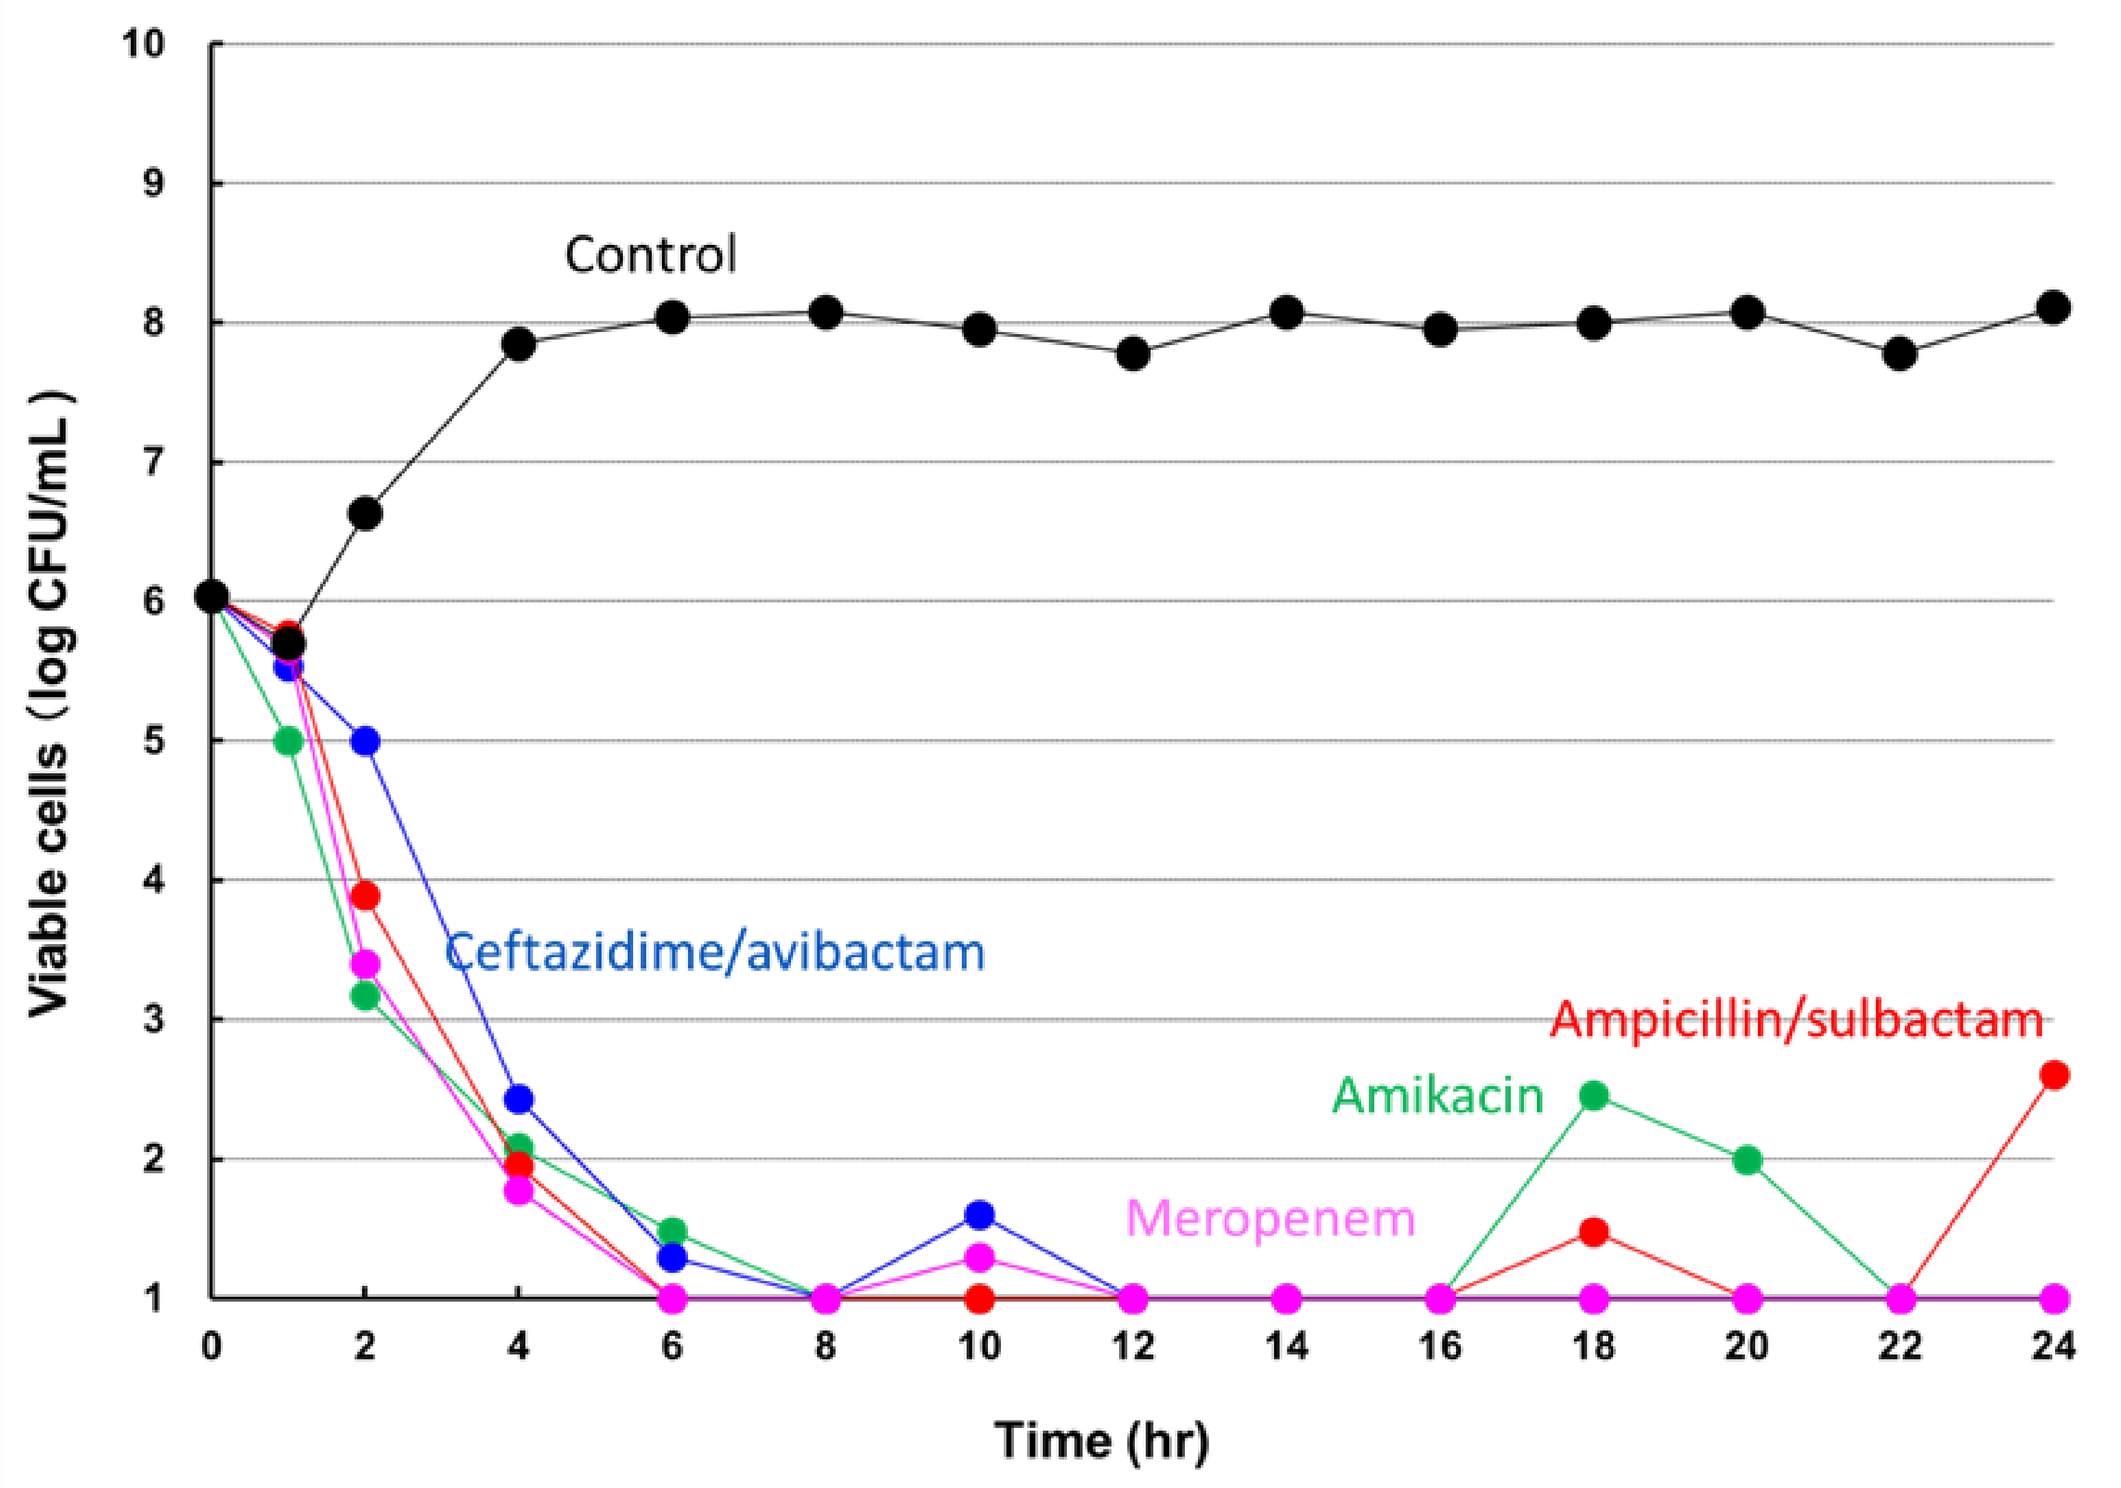

Viable cells (log CFU/mL)
Time (hr)
Control
Ceftazidime/avibactam
Ampicillin/sulbactam
Amikacin
Meropenem

Conclusions

- β-Lactamase inhibitors such as avibactam and sulbactam restore the activity of CFDC against *bla*$_{PER}$-possessing *A. baumannii*. Increased efficacy was also observed for the combination use of CFDC and a non-β-lactamase inhibitor such as MEM and AMK.
- These combination treatment regimens could lead to improved efficacy even against *bla*$_{PER}$ possessing *A. baumannii* with high CFDC MIC.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from IDWeek 2020 and the authors of this poster.