Poster 1269

IDWeek 2020
22–25 October, 2020

Differences in Interpretative Breakpoints Between CLSI, FDA and EUCAST Impact Reporting of Susceptibility and Resistance to Cefiderocol

Y Yamano,[1] M Takemura,[1] C Longshaw,[2] R Echols,[3]

[1]Shionogi & Co. Ltd, Osaka, Japan; [2]Shionogi B.V., London, UK; [3]Infectious Disease Drug Development Consulting LLC, Easton, CT, USA

Contact information:
Yoshinori Yamano
3-1-1, Futaba-cho, Toyonaka,
Osaka, 561-0825, Japan
Phone: +81-6-6331-6293
Fax: +81-6-6331-8612
Email: yoshinori.yamano@shionogi.co.jp

Abstract (Revised)

Background: Cefiderocol (CFDC) is a siderophore cephalosporin with broad coverage of aerobic Gram-negative (GN) bacteria. Provisional breakpoints (BP) were approved by CLSI in 2018, with FDA and EUCAST providing clinical BP in 2019 and 2020, respectively. FDA breakpoints for Enterobacterales were recently updated. There still remains substantial differences between organizations, reflecting differences in labelling, interpretation of PK-PD data and availability of clinical study data during regulatory review. Here we compare susceptibility rates based on these different BPs.

Methods: Susceptibility rates for each bacterial species were determined using CFDC BP from each organization and the MICs of 28,629 GN clinical isolates from 3 consecutive years of SIDERO-WT surveillance studies (2014–17). The analysis used all isolates and sub-grouped isolates based on meropenem (MEM) susceptibility (CLSI BP) or carbapenemase production.

Results: Within the overall Enterobacterales group, ≥98.5% isolates were interpreted as susceptible to CFDC regardless of BP used. However, the proportion of susceptible differed significantly (82.5–98.6%) when applied to MEM-non-susceptible (NS) isolates. Similarly, against most carbapenemase producers, susceptibility ranged from 80 to 100%, however for NDM producers, only 51% of isolates were defined as susceptible by old FDA or EUCAST BP vs 84% using the updated FDA/CLSI BP. Against Pseudomonas aeruginosa including MEM-NS isolates, susceptibility was ≥94% despite different BPs recommended by FDA (1 μg/mL), EUCAST (2 μg/mL) and CLSI (4 μg/mL). This changed the proportion of IMP-producing isolates classified as susceptible from 100% (CLSI) and 81% (EUCAST) to only 19% (FDA). Against other non-fermenters, susceptibility was ≥91% irrespective of BP used.

Conclusions: Differences in BPs between FDA, CLSI and EUCAST could impact on the reporting of susceptibility or resistance to CFDC, particularly for MEM-NS isolates. PK/PD model simulations support 100% fT>MIC up to an MIC of 4 mg/L, and in Phase 3 trials the mean trough concentration of unbound cefiderocol was >4 mg/L. The potential impact of these differences on clinical decision making are important as the greatest clinical utility for CDFC is expected to be in patients with carbapenem-resistant GN infections due to limited treatment options.

Introduction

Cefiderocol is a novel siderophore cephalosporin with activity against a wide variety of Gram-negative bacteria, including carbapenem-resistant Enterobacterales and non-fermenters.

In 2018, the investigational breakpoint for cefiderocol was determined by Clinical and Laboratory Standards Institute (CLSI) [1].

Cefiderocol has been approved in the USA for the treatment of patients with complicated urinary tract infections (cUTI) caused by Gram-negative bacteria and in Europe for the treatment of infections due to Gram-negative pathogens with limited treatment options [2, 3].

Cefiderocol has also been approved in the USA for the trearment of the patients with hospital-acquired pneumonia and ventilator-associated pneumonia, together with the update of breakpoints [2].

We have conducted 3-years-concecutive multinational surveillance studies (SIDERO-WT) from 2014 to 2016 to evaluate the in vitro activity of cefiderocol against clinical isolates collected in Europe and North America and to evaluate the carbapenemase profile of the subset of meropenem-non-susceptible isolates from the same studies [4, 5].

In this study, we evaluated the in vitro activity of cefiderocol based on the different breakpoint between organization (CLSI, FDA and EUCAST) [1, 6, 7].

Materials and Methods

Bacterial strains

- A total of 28,629 isolates (19,119 Enterobacterales, 4,942 *Pseudomonas aeruginosa*, 3,231 *Acinetobacter baumannii* complex, 1,173 *Stenotrophomonas maltophilia* and 164 *Burkholeria cepacia* complex) from SIDERO-WT-2014 to -2016 were used. These isolates were collected from North America (United States and Canada) and European countries (Czech Republic, France, Germany, Greece, Hungary, Italy, Russia, Spain Sweden, Turkey and United Kingdom) from November 2014 to October 2017.
- MICs of cefiderocol and meropenem (MEM) were determined by International Health Management Associate (IHMA), Schaumburg, IL, USA, using the broth microdilution methodology recommended by CLSI. For MIC determination of cefiderocol, iron-depleted cation-adjusted Muller Hinton broth (ID-CAMHB) was used.

Materials and Methods (Continued)

Bacterial strains

- The carbapenem-non-susceptible (Carb-NS) isolates were defined using the CLSI breakpoints for MEM (>1 μg/mL for Enterobacterales and >2 μg/mL for *P. aeruginosa* and *Acinetobacter* spp.) [1].
- The carbapenemase profile of Carb-NS isolates were determined by IHMA using PCR methodology to identify KPC, OXA [including OXA-23, OXA-24, OXA-48 and OXA-58], GES, NDM, IMP, VIM, SPM, and GIM.

Determination of Susceptibility Rates

- The susceptibility rate was determined for each bacterial species and a subset of CarbNS isolates of each bacterial species by using the breakpoint approved by CLSI, FDA and EUCAST (**Table 1**). The BP for Enterobacterales by FDA has been updated, so both BP (previous BP and updated BP) were used for this analysis.
- The susceptibility ratio was also determined by a subset of each carbapenemase producer of each bacterial species.

Results

Enterobacterales

- Within the overall Enterobacterales group, ≥98.5% isolates were interpreted as susceptible to CFDC regardless of BP used (**Fig. 1**). However, the proportion of susceptible differed significantly (82.5–98.6%) when applied to Carb-NS isolates (**Fig. 2**). Similarly, against KPC, OXA-48 or VIM producers, susceptibility ranged from 80 to 100%, however for NDM producers, only 51% of isolates were defined as susceptible by EUCAST or previous FDA BP vs 84% using the CLSI/updated FDA BP (**Fig. 3**).

P. aeruginosa

- Within the overall *P. aeruginosa*, ≥98.5% isolates were interpreted as susceptible to CFDC regardless of BP used (**Fig. 4**). Even for Carb-NS *P. aeruginosa*, ≥94.5% isolates were interpreted as susceptible to CFDC regardless of BP used (**Fig. 5**). Similarly, against VIM producers, susceptibility ranged from 94.5 to 99.9%, however for IPM producers, the susceptibility rate was significantly different between 3 different BP; 100, 18.8 and 81.2% based on BP by CLSI, FDA and EUCAST, respectively (**Fig. 6**).

A. baumannii complex

- Within the overall *A. baumannii* complex, ≥90% isolates were interpreted as susceptible to CFDC regardless of BP used (**Fig. 7**). For Carb-NS *A. baumannii* complex, the susceptibility rate was different between 3 different BP; 94.9, 86.0 and 91.8% based on BP by CLSI, FDA and EUCAST, respectively (**Fig. 8**).

Table 1. Susceptible Breakpoint Approved by each Organization

	FDA			CLSI			EUCAST	
	S	I	R	S	I	R	S	R
Enterobacterales	(2)	(4)	(8)	4	8	16	2	4
Updated	4	8	16					
P. aeruginosa	1	2	4	4	8	16	2	4
A. baumannii Updated	1	2	4	4	8	16	2*	4*
S. maltophilia	NA			4	8	16	2*	4*
B. cepacia	NA			NA			2*	4*

NA: Not available
*: PK-PD (non-species-related) breakpoints were applied

Results

Figure 1. MIC Distribution and Susceptibility Rate against All Enterobacterales (N = 19,119)

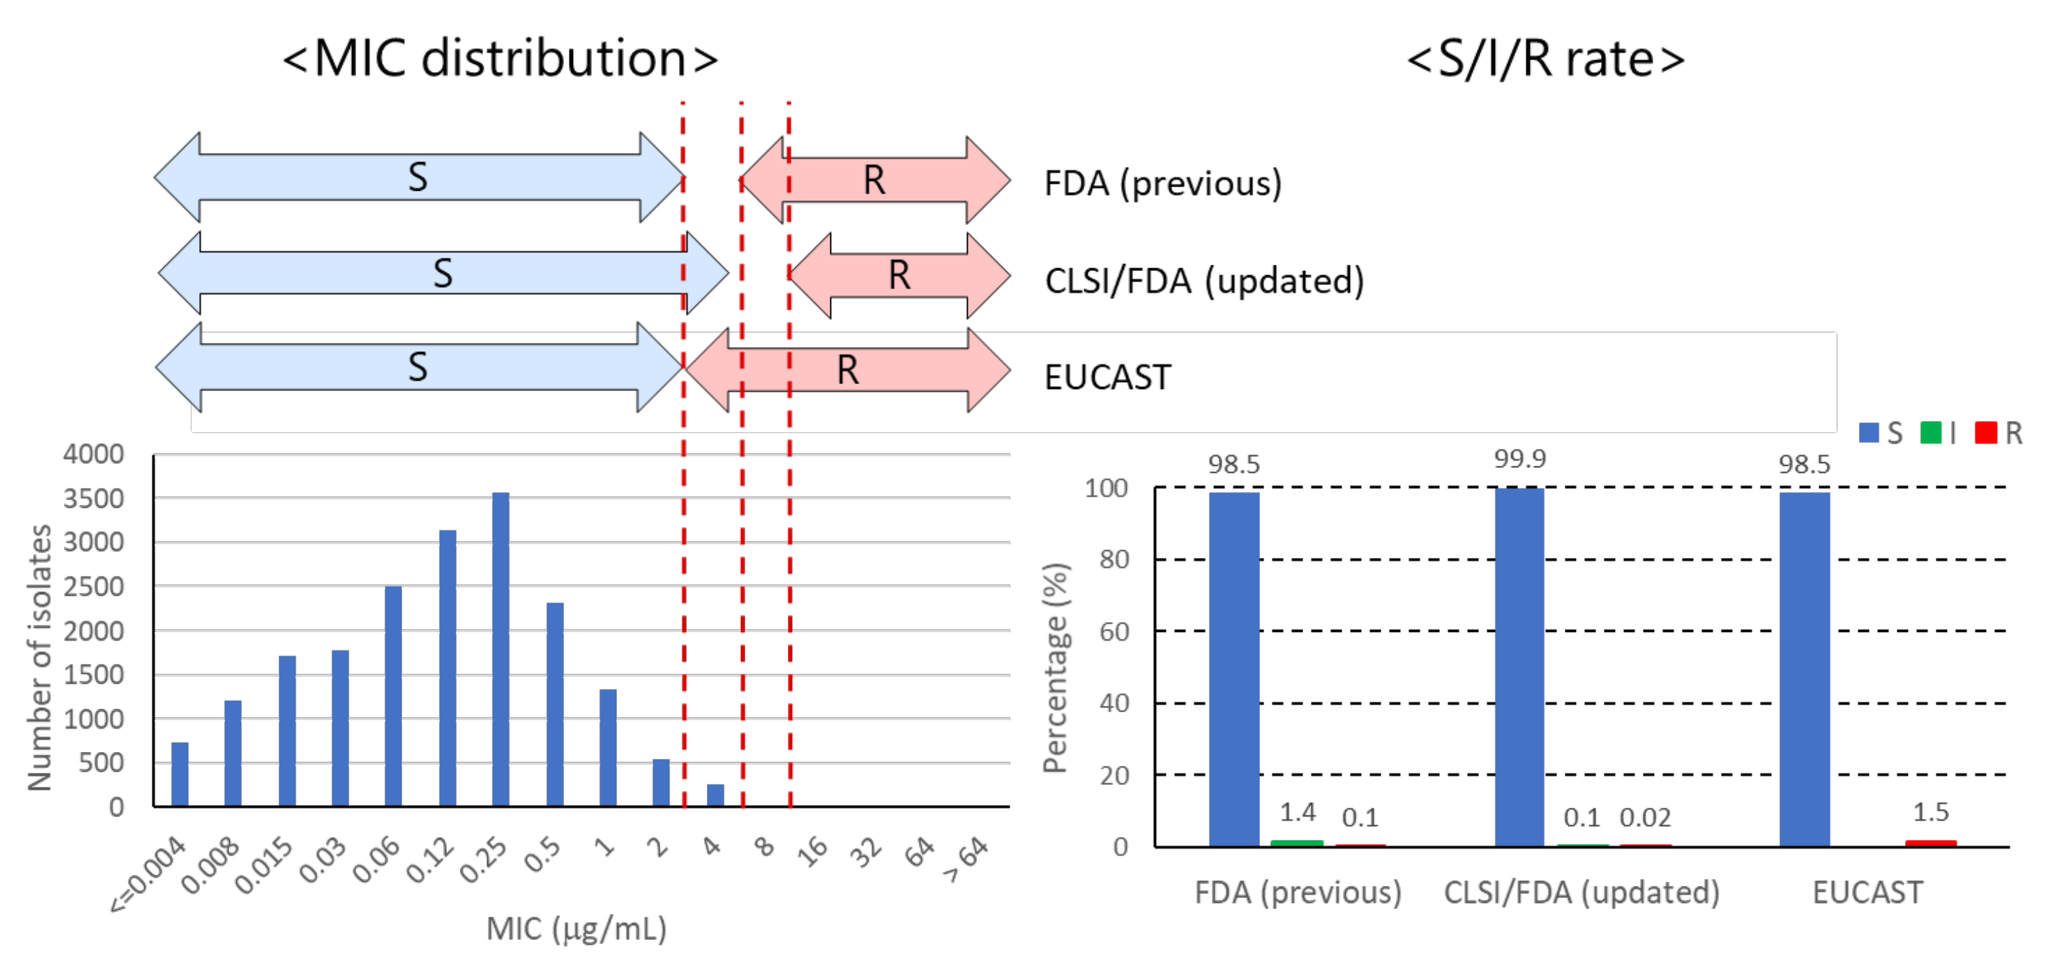

<MIC distribution>
<S/I/R rate>
FDA (previous)
CLSI/FDA (updated)
EUCAST
Number of isolates
MIC (μg/mL)
Percentage (%)
98.5
99.9
98.5

Figure 2. MIC Distribution and Susceptibility Rate against CarbNS Enterobacterales (N = 640)

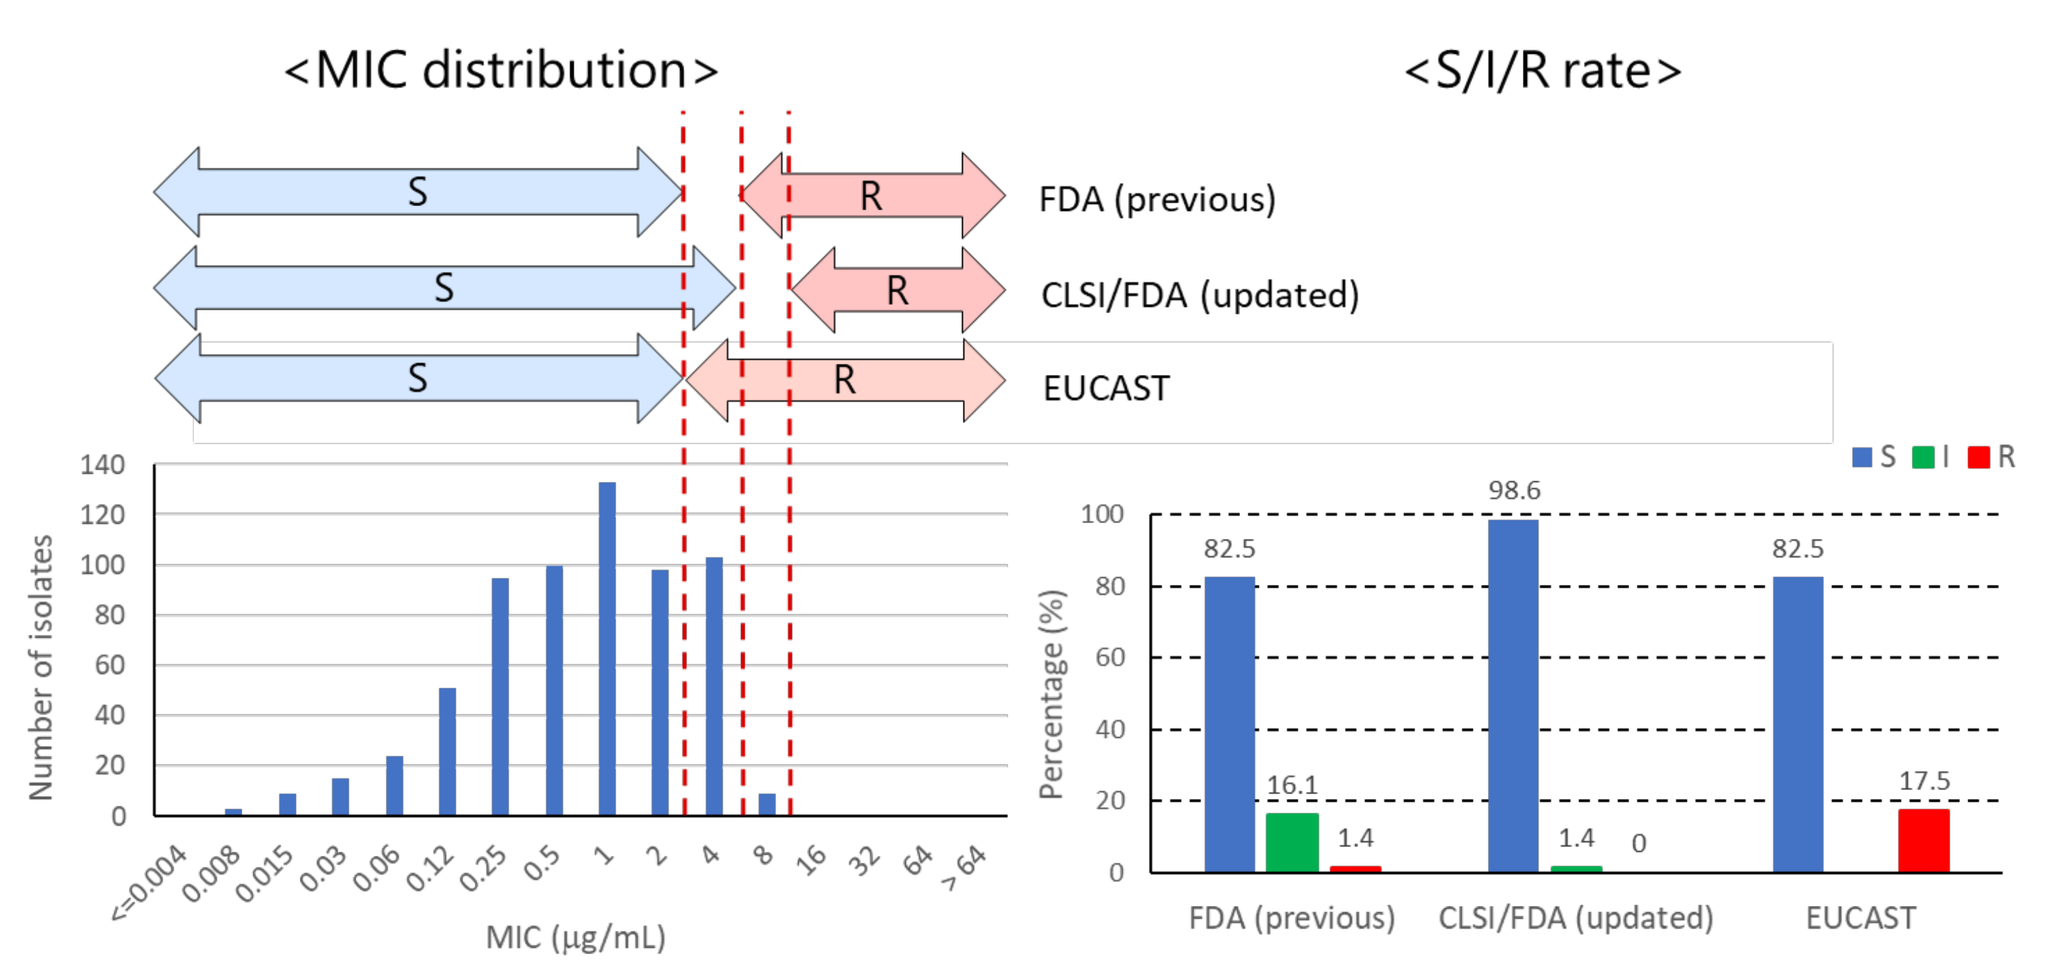

<MIC distribution>
<S/I/R rate>
FDA (previous)
CLSI/FDA (updated)
EUCAST
82.5
16.1
98.6
82.5
17.5

Figure 3. Susceptible Rate against each Carbapenemase-Positive Isolates of Enterobacterales

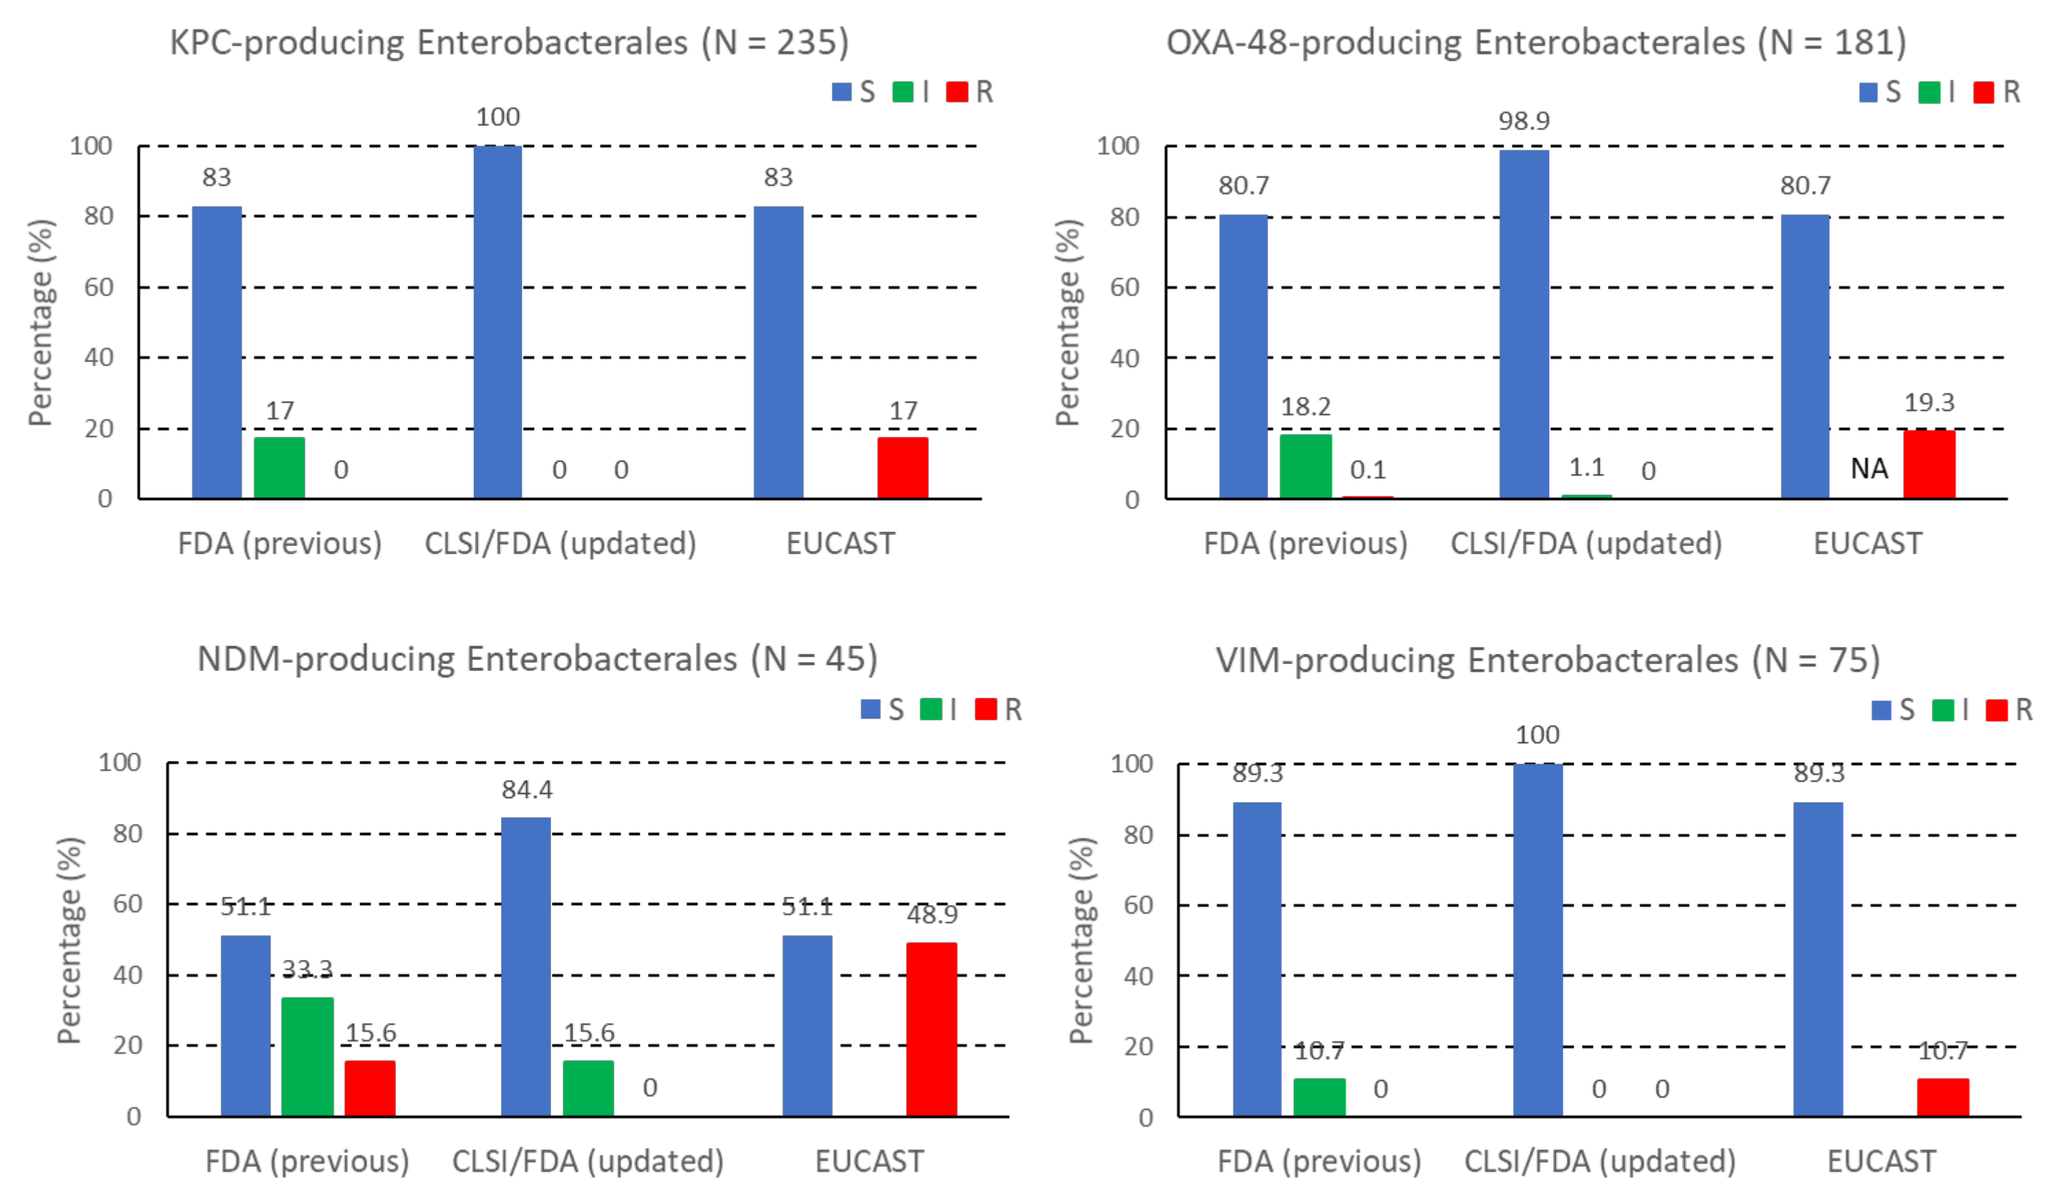

KPC-producing Enterobacterales (N = 235)
OXA-48-producing Enterobacterales (N = 181)
NDM-producing Enterobacterales (N = 45)
VIM-producing Enterobacterales (N = 75)

Figure 4. MIC Distribution and Susceptibility Rate against All *P. aeruginosa* (N = 4,942)

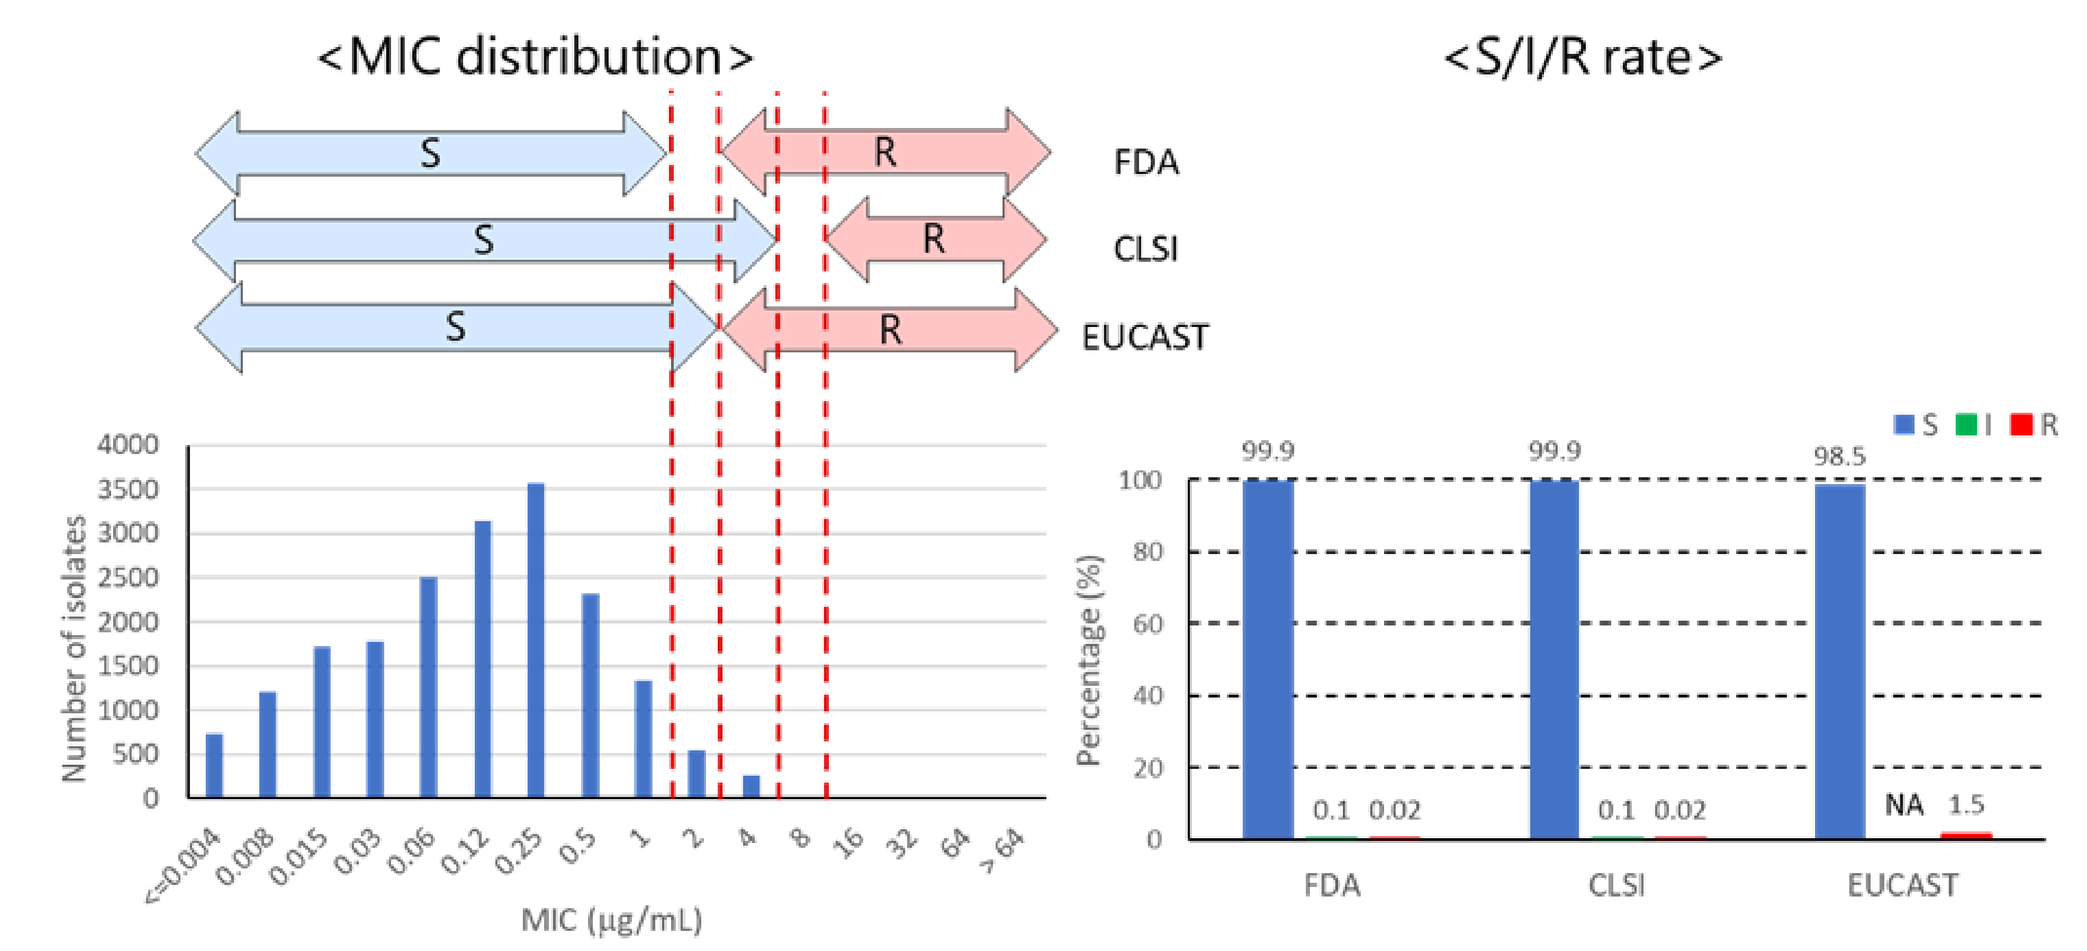

<MIC distribution>
<S/I/R rate>
FDA
CLSI
EUCAST
99.9
99.9
98.5

Figure 5. MIC distribution and Susceptibility rate against CarbNS *P. aeruginosa* (N = 1,154)

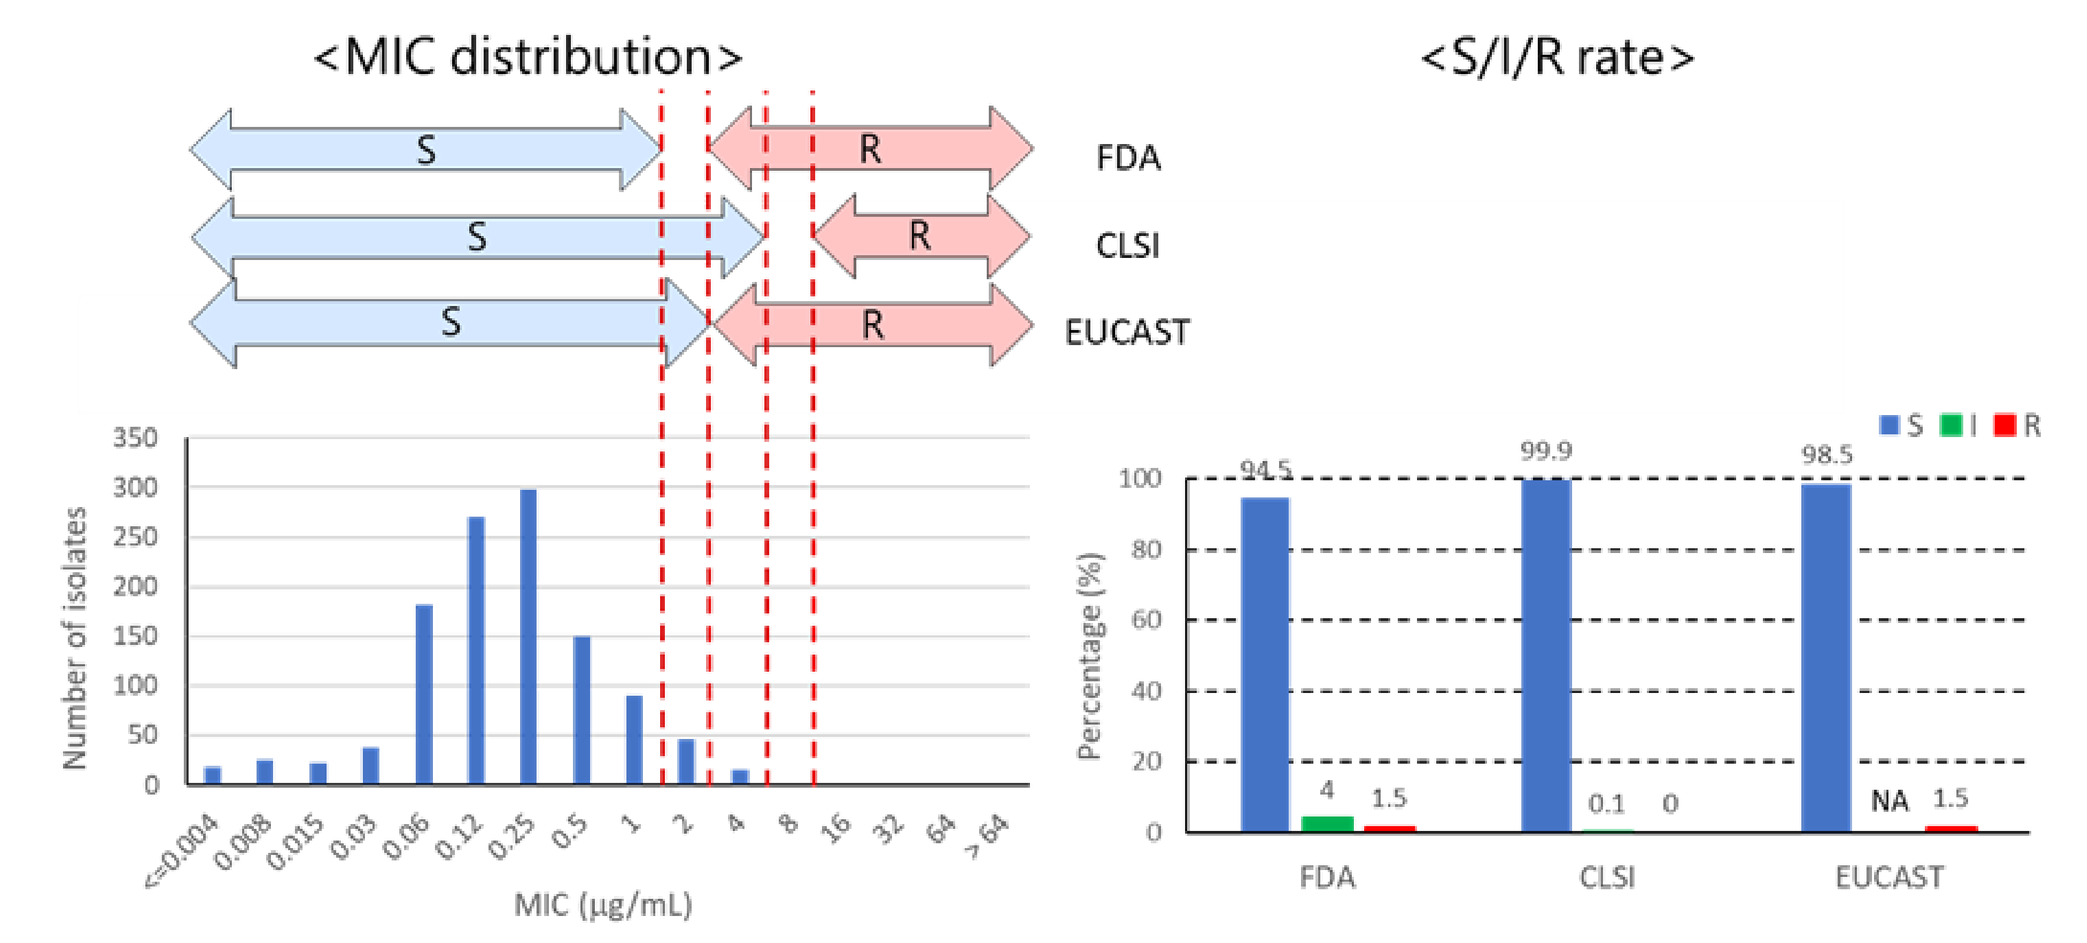

<MIC distribution>
<S/I/R rate>
FDA
CLSI
EUCAST
94.5
99.9
98.5

Figure 6. Susceptible Rate against each Carbapenemase-Positive Isolates of *P. aeruginosa*

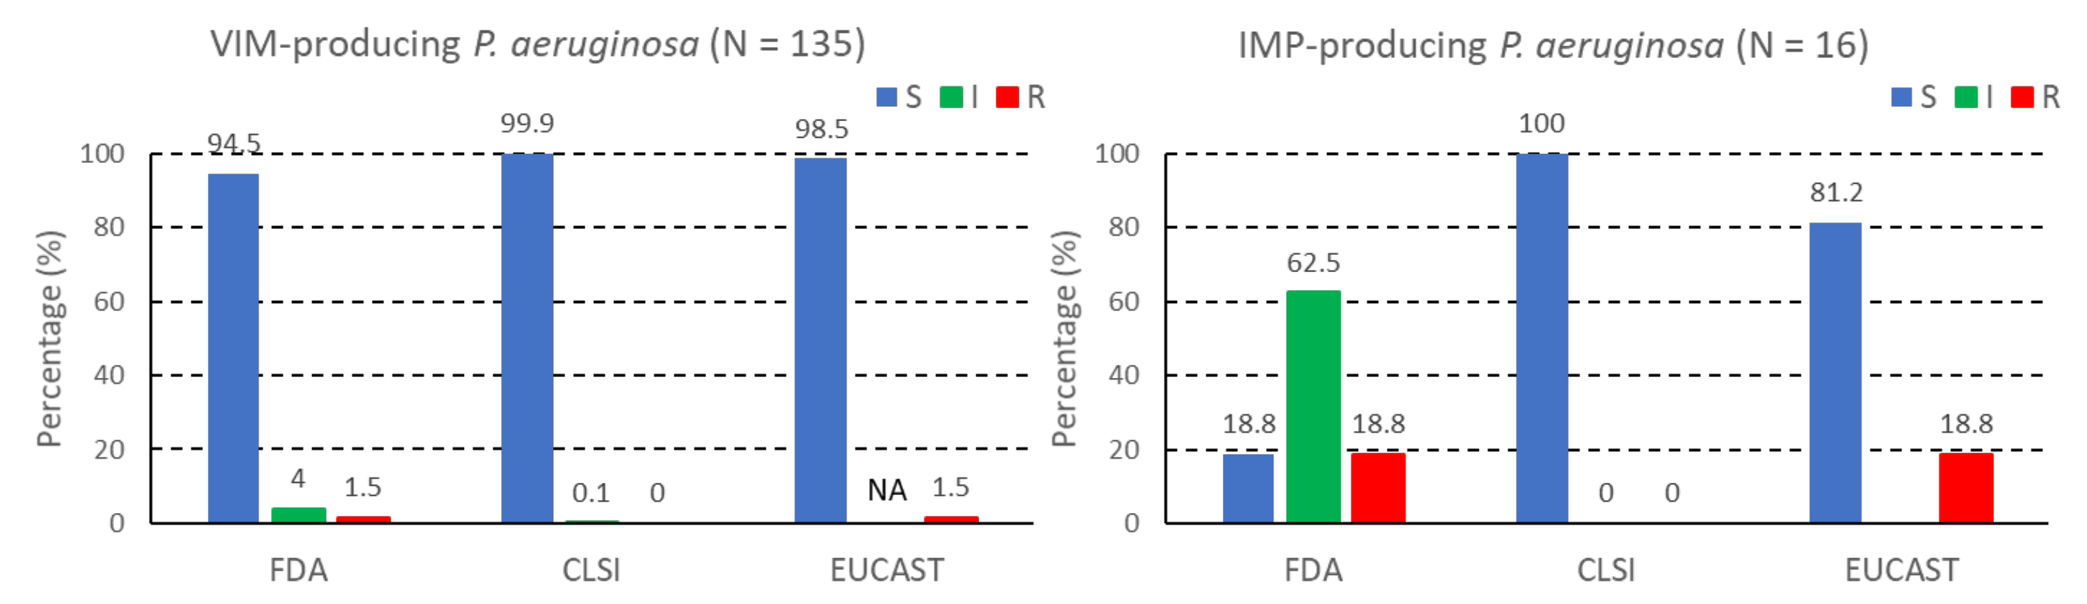

VIM-producing P. aeruginosa (N = 135)
IMP-producing P. aeruginosa (N = 16)

Figure 7. MIC Distribution and Susceptibility Rate against All *A. baumannii* complex (N = 3,231)

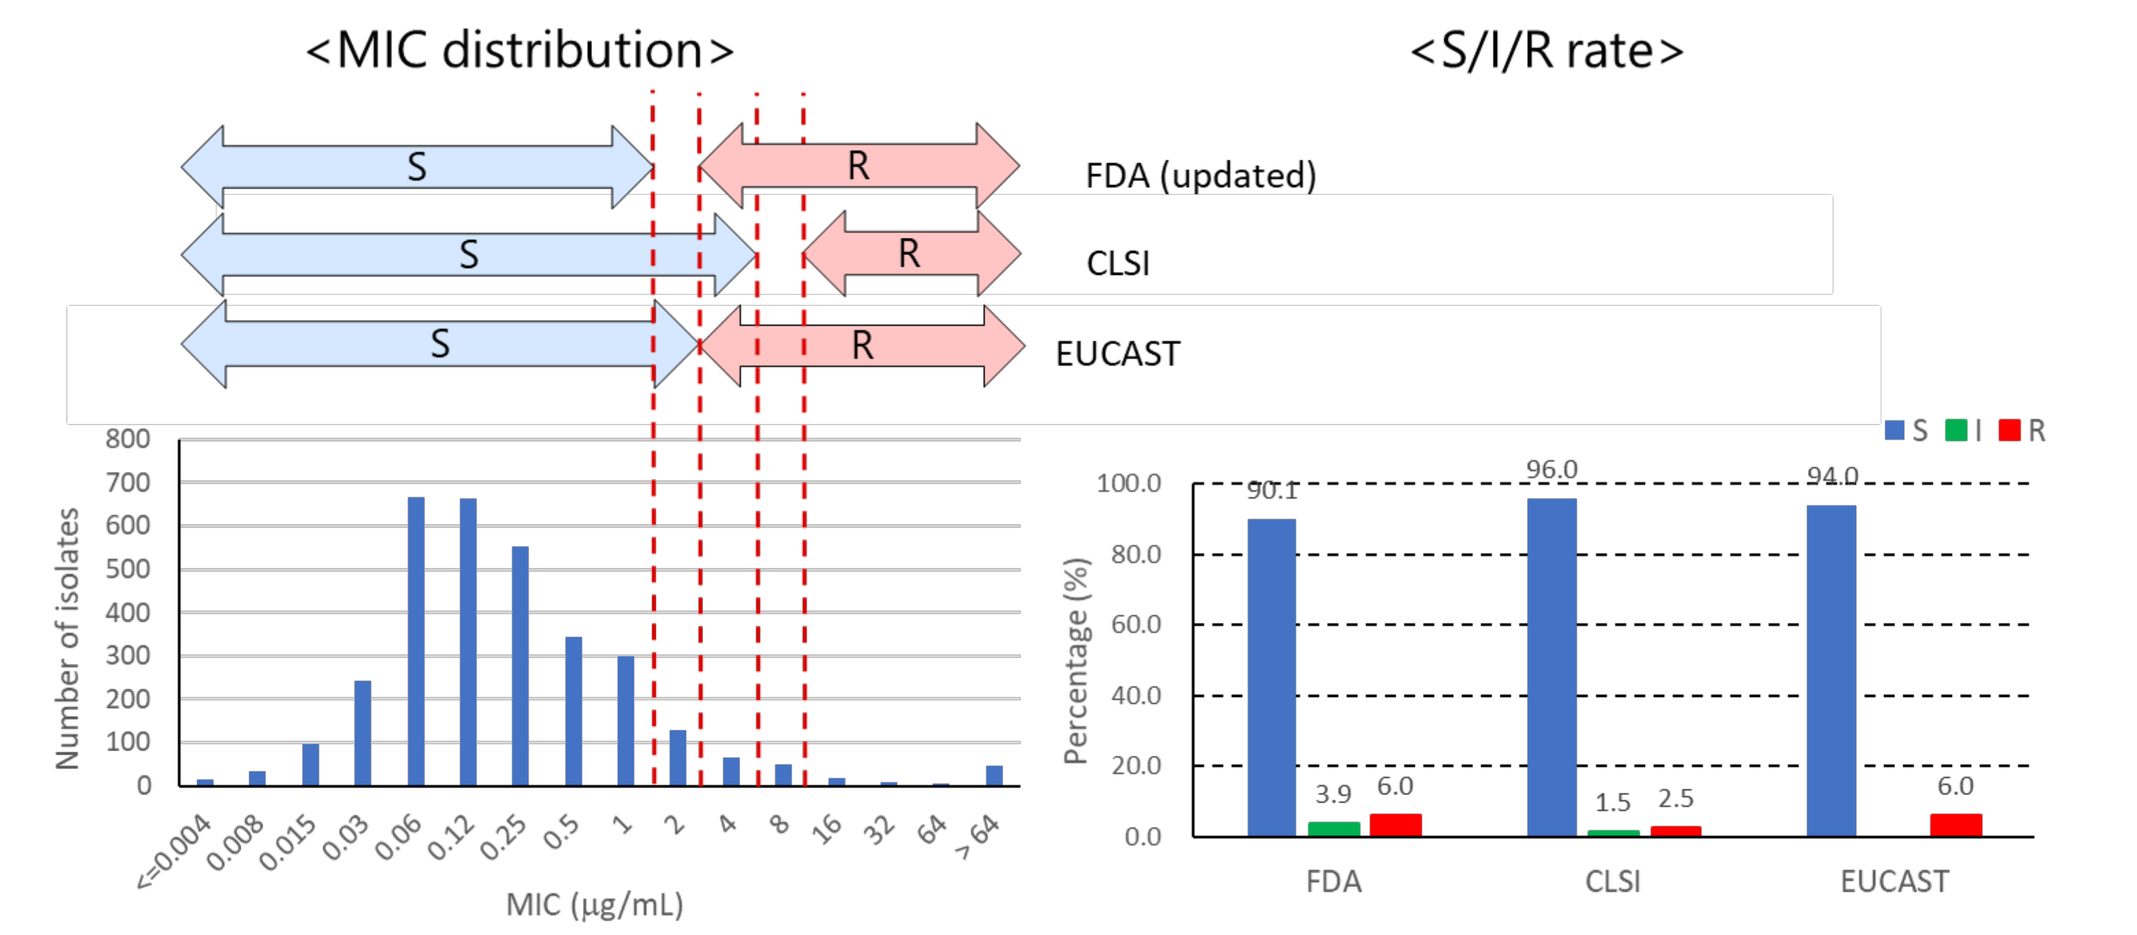

<MIC distribution>
<S/I/R rate>
FDA (updated)
CLSI
EUCAST
90.1
96.0
94.0

Figure 8. MIC Distribution and Susceptibility Rate against CarbNS *A. baumannii* complex (N = 1,899)

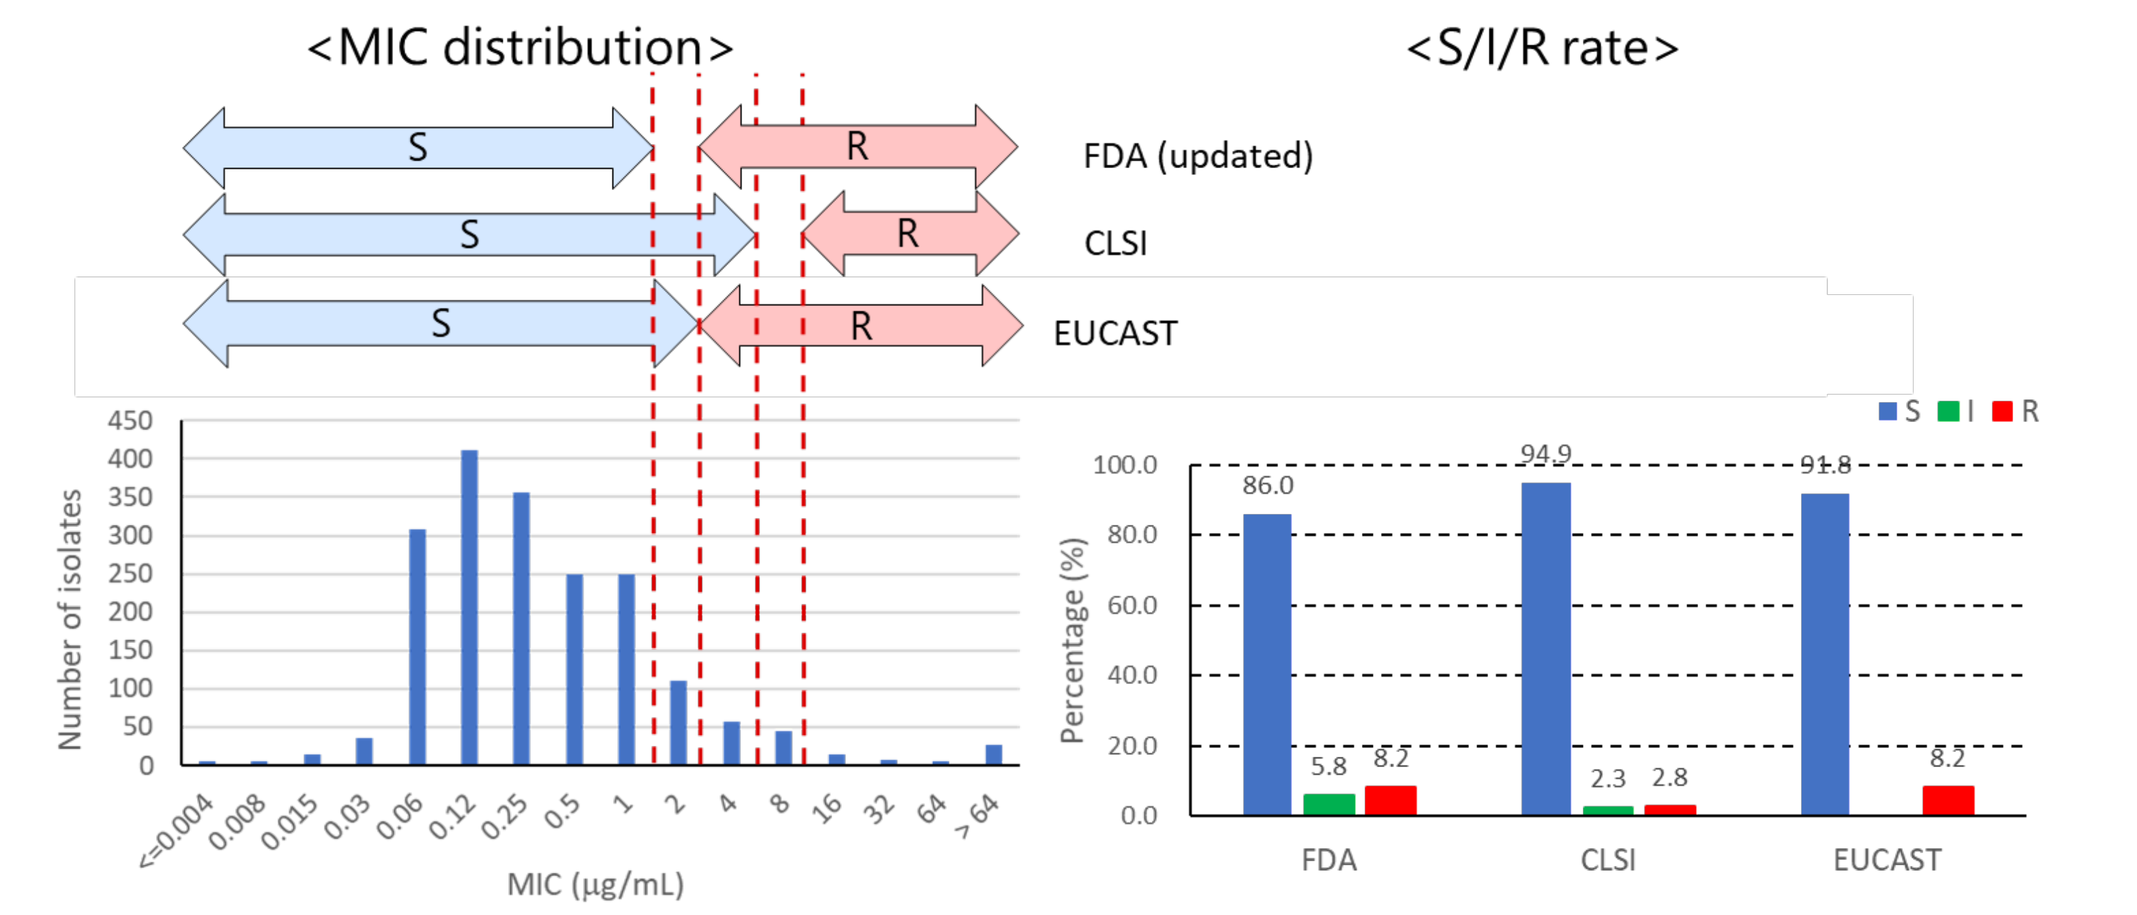

<MIC distribution>
<S/I/R rate>
FDA (updated)
CLSI
EUCAST
86.0
94.9
91.8

Conclusions

- Differences in BPs among FDA, CLSI and EUCAST may affect the reporting of susceptibility or resistance to CFDC, particularly for NDM-producing Carb-NS Enterobacterales and IMP-producing *P. aeruginosa*.
- The potential impact of these differences on clinical decision making cannot be ignored as the greatest clinical utility for CFDC is expected to be in patients with carbapenem-resistant GN infections, particularly metallo-carbapenemase producing organisms, as treatment options are limited in infections with these organisms.
- PK/PD model simulations support 100% fT>MIC up to an MIC of 4 μg/mL for CFDC; the mean trough concentration of unbound CFDC was >4 μg/mLin Phase 3 trials .
- Harmonization of CFDC breakpoints is desired and possible with further clinical data accumulation.

References

1. CLSI. CLSI supplement M100: M100–S30. Wayne, PA: CLSI; 2020.
2. Fetroja [cefiderocol] for injection, for intravenous use. Prescribing Information. Shionogi Inc., Florham Park, NJ, USA. 2020.
3. Fetcroja [cefiderocol]. 1 g powder for concentrate for solution for infusion. Summary of Product Characteristics. Shionogi B.V., Kingsfordweg 151, 1043 GR, Amsterdam, Netherlands. 2020.
4. Hackel MA, et al. Antimicrob Agents Chemother 2017;61:pii:e00093-17.
5. Karlowsky JA, et al. Int J Antimicrob Agents 2019;53:456–66.
6. The European Committee on Antimicrobial Susceptibility Testing – EUCAST Clinical Breakpoints 2019. Available at http://www.eucast.org/clinical_breakpoints/.
7. Antibacterial Susceptibility Test Interpretive Criteria. FDA. Available at https://www.fda.gov/drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from IDWeek 2020 and the authors of this poster.