557

Impact of Concomitant Hydroxychloroquine Use on Safety and Efficacy of Remdesivir in Moderate COVID-19 Patients

Jose Ramon Arribas,[1] Arun J. Sanyal,[2] Alex Soriano Viladomiu,[3] Bum Sik Chin,[4] Shirin Kalimuddin,[5] Stefan Schreiber,[6] Emon Elboudwarej,[7] Yuan Tian,[7] Robert H. Hyland,[7] Devi SenGupta,[7] Anand Chokkalingam,[7] Anu O. Osinusi,[7] Diana M. Brainard,[7] Christoph Lübbert,[8] David Chien Boon Lye,[9] Judith A. Aberg,[10] Enrique Navas Elorza,[11] Karen T. Tashima,[12] Mark McPhail[13]

[1]Hospital Universitario La Paz, Madrid, Spain; [2]VCU Medical Center, Richmond, VA; [3]Hospital Clínic de Barcelona, Spain; [4]National Medical Center, Seoul, Republic of Korea; [5]Singapore General Hospital, Singapore; [6]Universitätsklinikum Schleswig-Holstein, Kiel, Germany; [7]Gilead Sciences, Inc., Foster City, CA; [8]Klinik für Infektiologie/Tropenmedizin, Nephrologie und Rheumatologie, Klinikum St. Georg, Leipzig, Germany; [9]National Centre for Infectious Diseases, Tan Tock Seng Hospital, Singapore; [10]Icahn School of Medicine at Mount Sinai, New York, NY; [11]Hospital Universitario Ramón y Cajal, Madrid; [12]The Miriam Hospital, Providence, RI; [13]King's College Hospital, London, UK

Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
800-445-3235

Introduction

- Remdesivir (RDV) has demonstrated potent in vitro and in vivo activity against severe acute respiratory syndrome coronavirus 2 (SARS-CoV-2), and favorable clinical efficacy and tolerability in patients with COVID-19[1]
- Results from the Phase 3 SIMPLE-Moderate study (GS-US-540-5774; NCT04292730) showed that hospitalized patients with COVID-19 not requiring O_2 support treated with RDV experienced better clinical status and good safety outcomes compared with standard of care (SOC)[2,3]
- Hydroxychloroquine (HCQ) is an experimental treatment for COVID-19 that may interfere with conversion of RDV to the active triphosphate (RDV-TP)

Preliminary In Vitro RDV and Chloroquine Data in RSV[4]

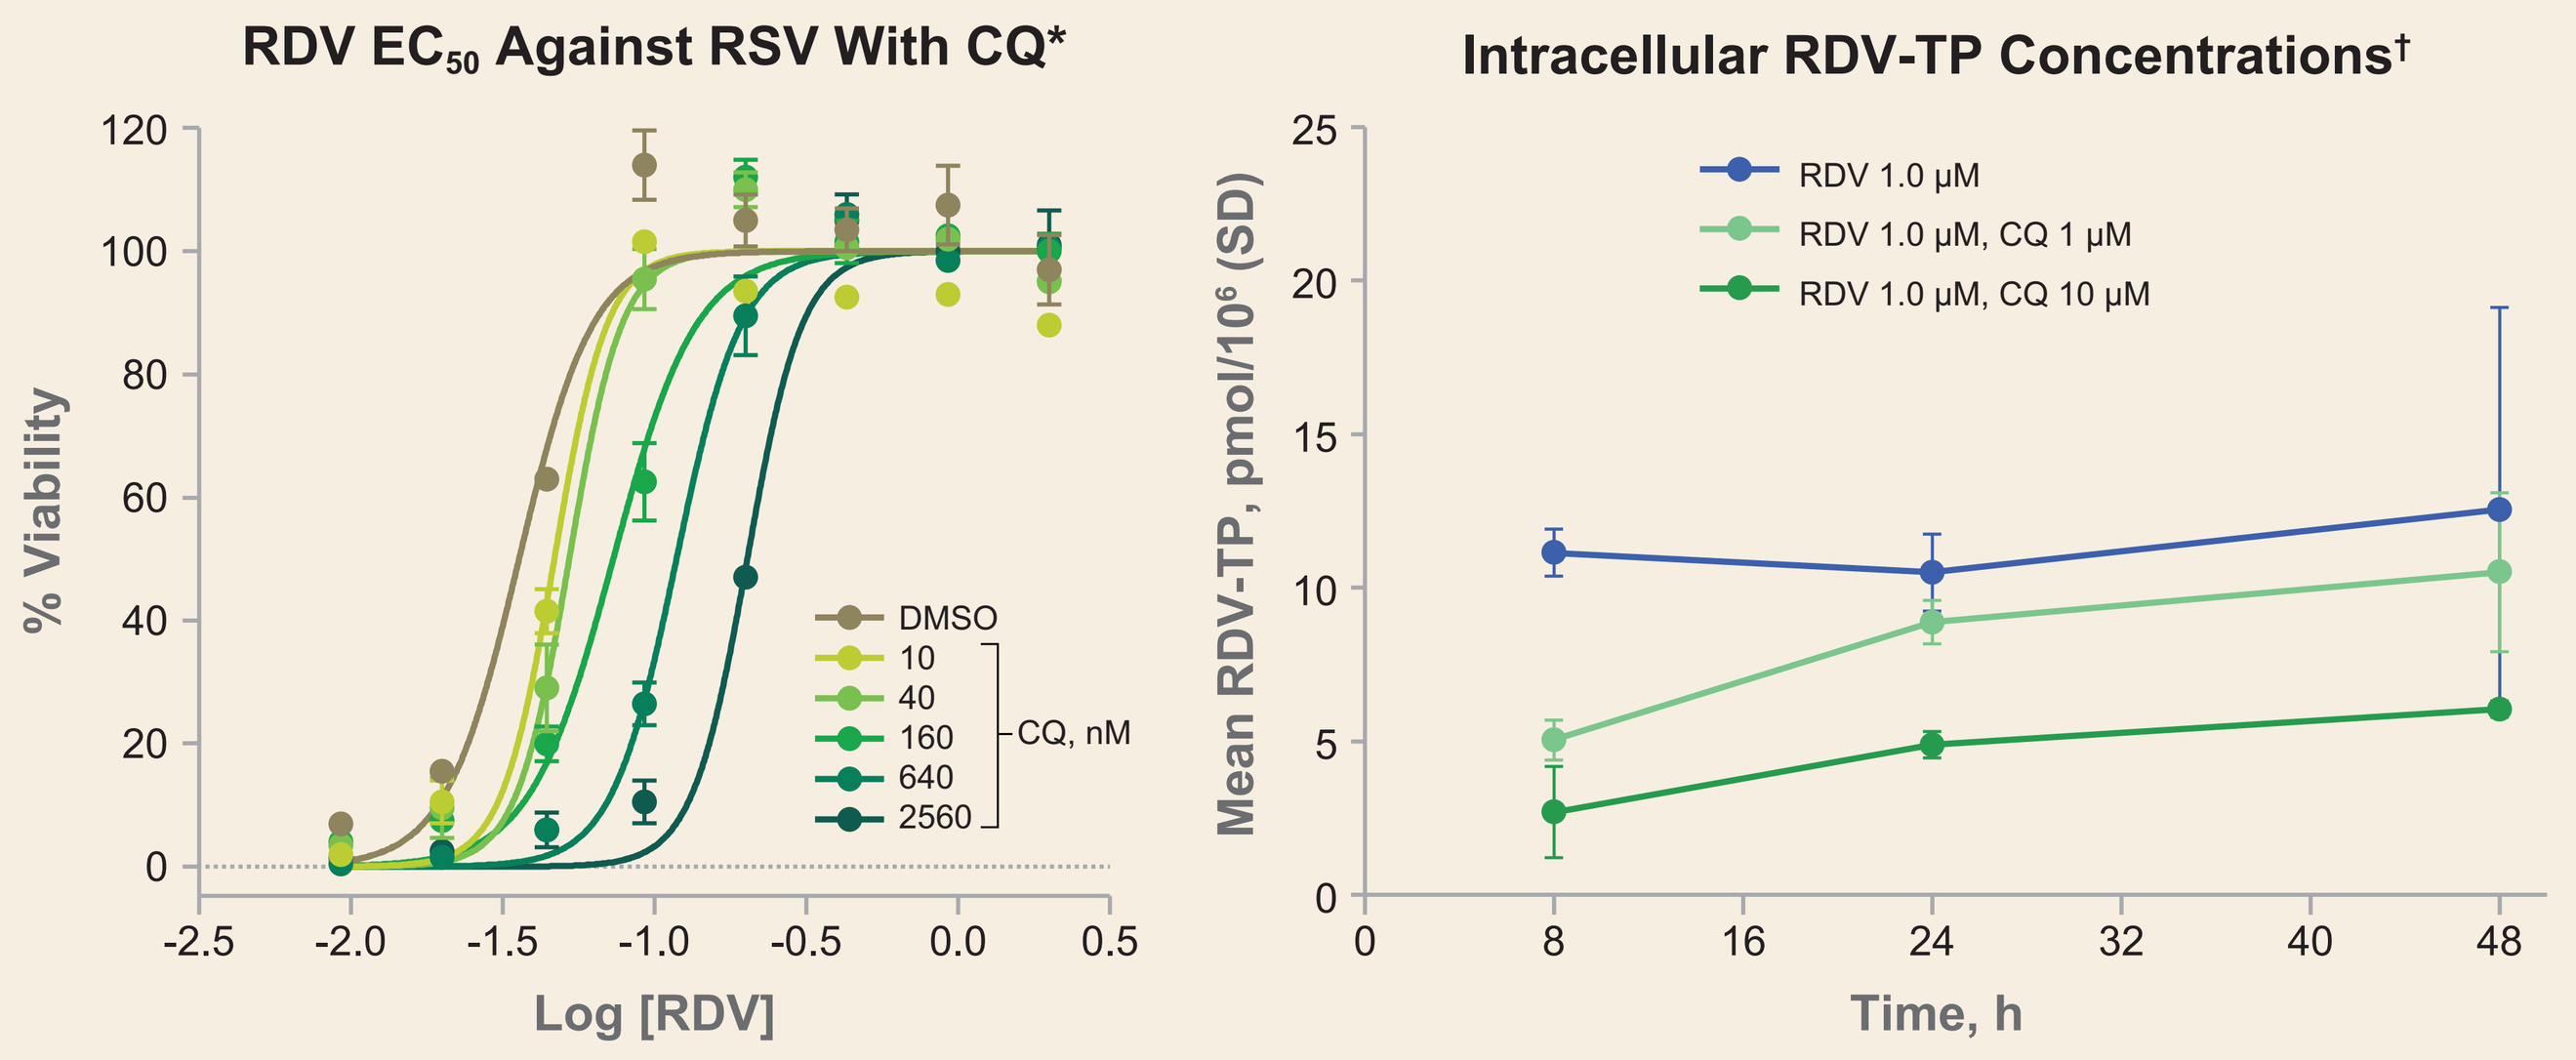

RDV EC50 Against RSV With CQ*
% Viability
Log [RDV]
DMSO
10
40
160
640
2560
CQ, nM
Intracellular RDV-TP Concentrations†
Mean RDV-TP, pmol/10^6 (SD)
Time, h
RDV 1.0 µM
RDV 1.0 µM, CQ 1 µM
RDV 1.0 µM, CQ 10 µM

*Respiratory syncytial virus (RSV) antiviral experiments were performed in human epithelial type 2 cells; †RDV metabolism studies were performed in normal human bronchial epithelial cells. CQ, chloroquine; DMSO, dimethyl sulfoxide; EC_{50}, half-maximal effective concentration; SD, standard deviation.

- CQ antagonizes RDV RSV antiviral activity in a dose-dependent manner in vitro
- Co-incubation of RDV and CQ shows dose-dependent inhibition of RDV-TP formation

Objectives

- To assess the impact of concomitant HCQ with RDV use on clinical outcomes and safety of RDV in the SIMPLE-Moderate Study

Methods

Phase 3, Randomized, Open-label, Multicenter Study

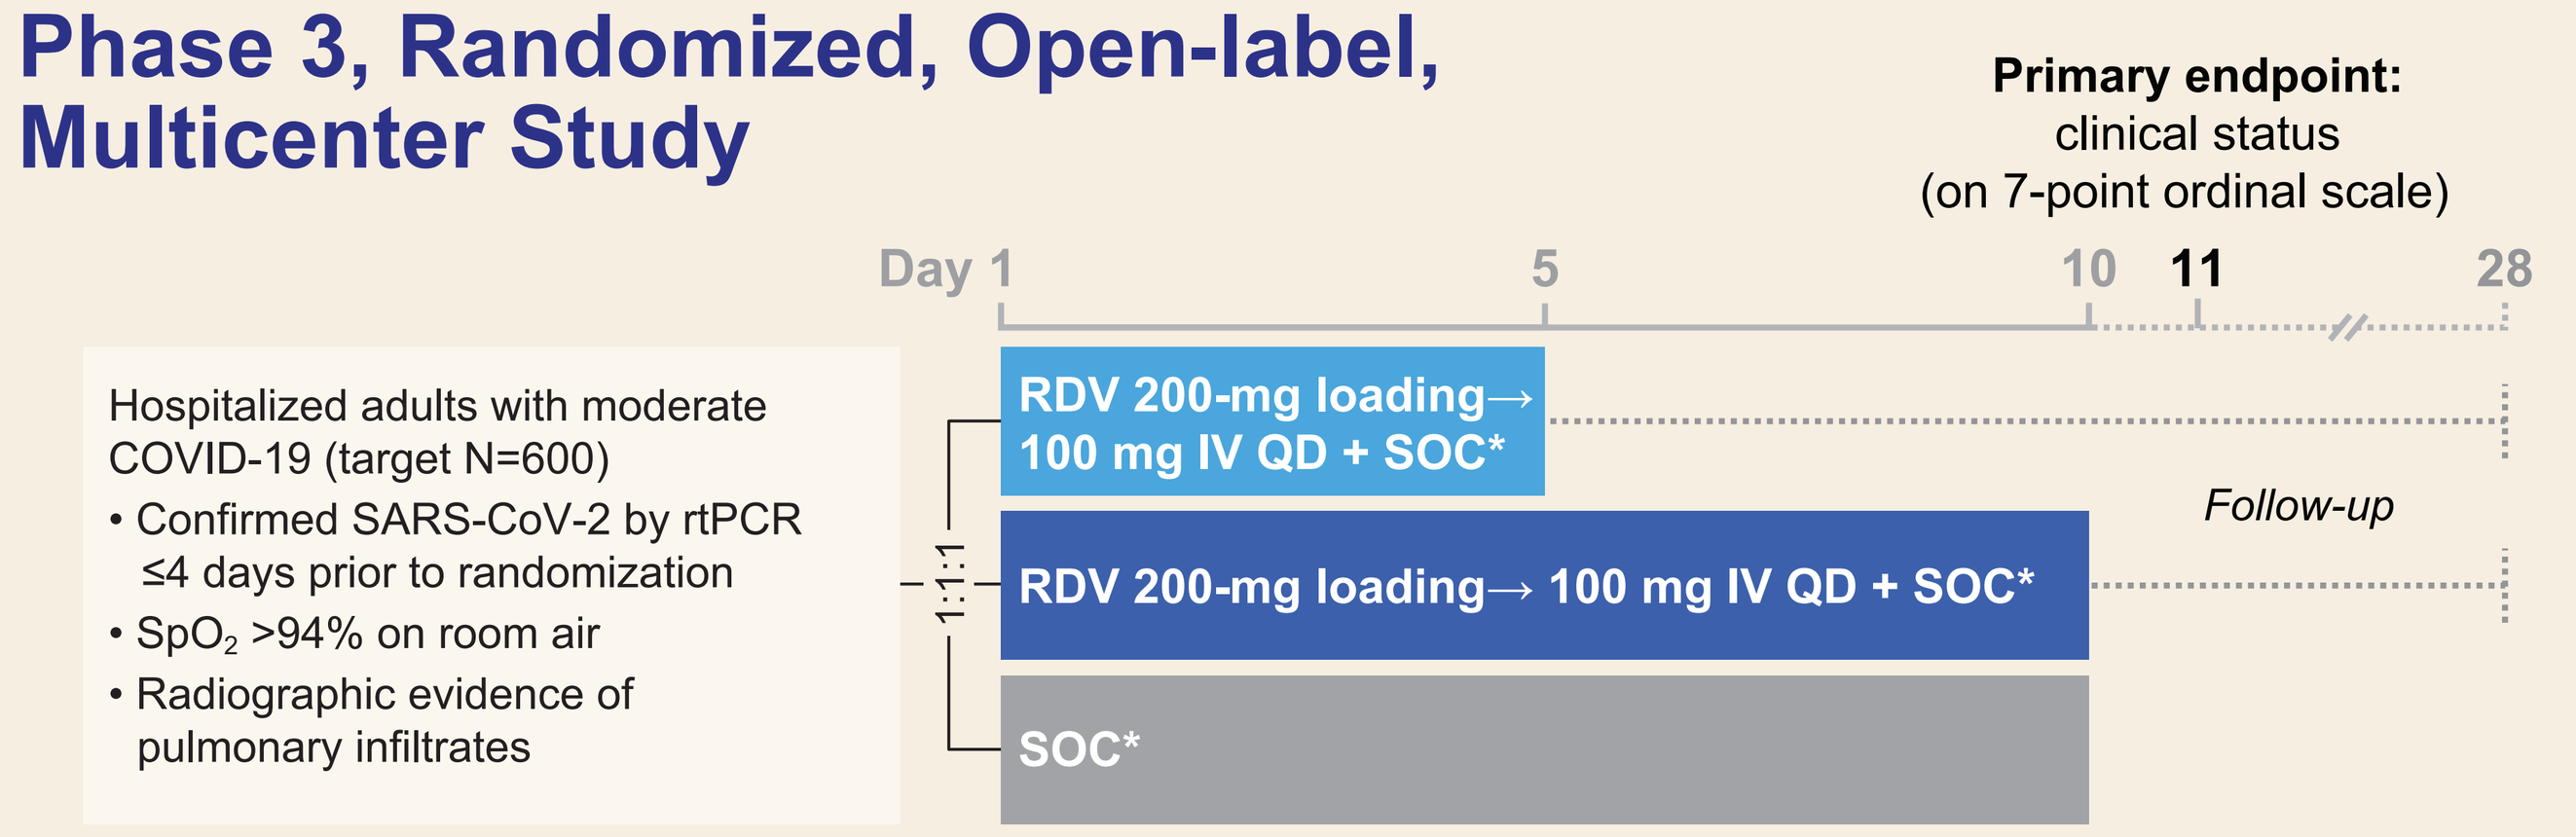

Primary endpoint:
clinical status
(on 7-point ordinal scale)
Day 1
5
10
11
28
Hospitalized adults with moderate COVID-19 (target N=600)
• Confirmed SARS-CoV-2 by rtPCR ≤4 days prior to randomization
• SpO2 >94% on room air
• Radiographic evidence of pulmonary infiltrates
1:1:1
RDV 200-mg loading→ 100 mg IV QD + SOC*
RDV 200-mg loading→ 100 mg IV QD + SOC*
SOC*
Follow-up

*SOC continued throughout study duration; agents with actual or possible direct-acting antiviral activity against SARS-CoV-2 were not allowed. rtPCR, reverse-transcriptase–polymerase chain reaction assay; SpO_2, peripheral oxygen saturation.

- Study enrolled March 15–April 18, 2020; date of final follow-up: May 20
- 105 centers in 12 countries: Asia (Hong Kong, Republic of Korea, Singapore, and Taiwan), Europe (France, Germany, Italy, Netherlands, Spain, Switzerland, and UK), and North America (USA)
- Key exclusion criteria:
 - Alanine aminotransferase (ALT) or aspartate aminotransferase (AST) >5x upper limit of normal; creatinine clearance <50 mL/min
 - Use of any experimental treatment for COVID-19, including HCQ, ≤24 h prior to dosing
 - Any requirement for invasive mechanical ventilation (IMV) at screening
- HCQ includes WHODrug BMAR20 preferred name hydroxychloroquine, hydroxychloroquine sulfate, chloroquine, and aminoquinolines
 - Classification of each medication in the HCQ group and all other COVID-19 medications was determined by a Gilead medical monitor

Clinical Outcomes Were Measured on 7-Point Ordinal Scale[3]

	1	Death
Hospitalized +	2	IMV or ECMO
	3	Noninvasive ventilation or high-flow O_2
	4	Low-flow O_2
	5	Room air, ongoing medical care (COVID-19 related or otherwise)
	6	Room air, no ongoing medical care (other than per-protocol RDV administration)
	7	Discharged

ECMO, extracorporeal membrane oxygenation.

Statistical Methods

- Within each treatment group (RDV for 5 or 10 days vs SOC), patients who received concomitant HCQ (≥1 day of exposure) were compared with those who did not
 - Concomitant use excluded patients who stopped HCQ prior to study Day -21 or started after study Day 5
- Comparison of continuous covariates by HCQ status was tested using Wilcoxon rank-sum test; categorical variables were tested using Cochran Mantel-Haenszel test
- Clinical recovery, ≥2-point clinical improvement, and all-cause mortality were evaluated using Cox proportional hazards
 - Clinical recovery and ≥2-point clinical improvement were analyzed with death as a competing risk
- For adverse events (AEs) and death by Day 28, HCQ differences were evaluated using logistic regression
- Potential covariates for adjustment included:
 - Baseline (BL) demographics: age, sex, race, and region (Italy vs outside Italy)
 - Disease characteristics: symptom duration and ordinal scale score (clinical status)
 - Comorbidities: cardiovascular disease (CVD), diabetes, and obesity
- Results were considered significant at p <0.05

Results

Baseline Demographics and Clinical Characteristics

		All RDV: RDV Alone n=274	All RDV: RDV + HCQ n=110	All RDV: p-Value	SOC: SOC Alone n=107	SOC: SOC + HCQ n=93	SOC: p-Value
Median age, year (range)		57 (12–94)	57 (20–86)	0.38	57 (24–89)	57 (23–95)	0.59
≥65 years, n (%)		79 (29)	22 (20)	0.08	29 (27)	29 (31)	0.53
Men, n (%)		167 (61)	65 (59)	0.74	70 (65)	55 (59)	0.36
Median body mass index, kg/m² (range)		27.8 (16.1–63.2)	26.0 (17.3–76.9)	0.002	26.6 (16.6–53.9)	27.0 (15.9–52.7)	0.88
Obesity: ≥30 kg/m², n (%)		91 (35)	21 (20)	0.004	33 (32)	22 (25)	0.29
Region, n (%)	Italy	15 (5)	38 (35)	<0.001	2 (2)	24 (26)	<0.001
	Rest of world	259 (95)	72 (65)		105 (98)	69 (74)	
Race, n (%)	Black*	67 (24)	5 (5)	<0.001	18 (17)	9 (10)	<0.001
	White	124 (45)	92 (84)		46 (43)	66 (71)	
	Asian	63 (23)	2 (2)		32 (30)	5 (5)	
	Other	20 (7)	11 (10)		11 (10)	13 (14)	
Comorbidities, n (%)	CVD	177 (65)	45 (41)	<0.001	60 (56)	47 (51)	0.43
	Diabetes	129 (47)	27 (25)	<0.001	47 (44)	29 (31)	0.07
Median duration of symptoms before study Day 1, day (range)		7 (1–48)	9 (2–40)	<0.001	8 (2–26)	9 (1–34)	0.60
Clinical status, n (%)	High-flow O_2	1 (<1)	2 (2)	<0.001	0	2 (2)	0.005
	Low-flow O_2	23 (8)	29 (26)		13 (12)	23 (25)	
	Room air	250 (91)	79 (72)		94 (88)	68 (73)	

*33% (85/260) of all US patients.

- Patients treated with HCQ were commonly in Italy, had fewer comorbidities, and were more frequently receiving O_2 support

Baseline Clinical Status

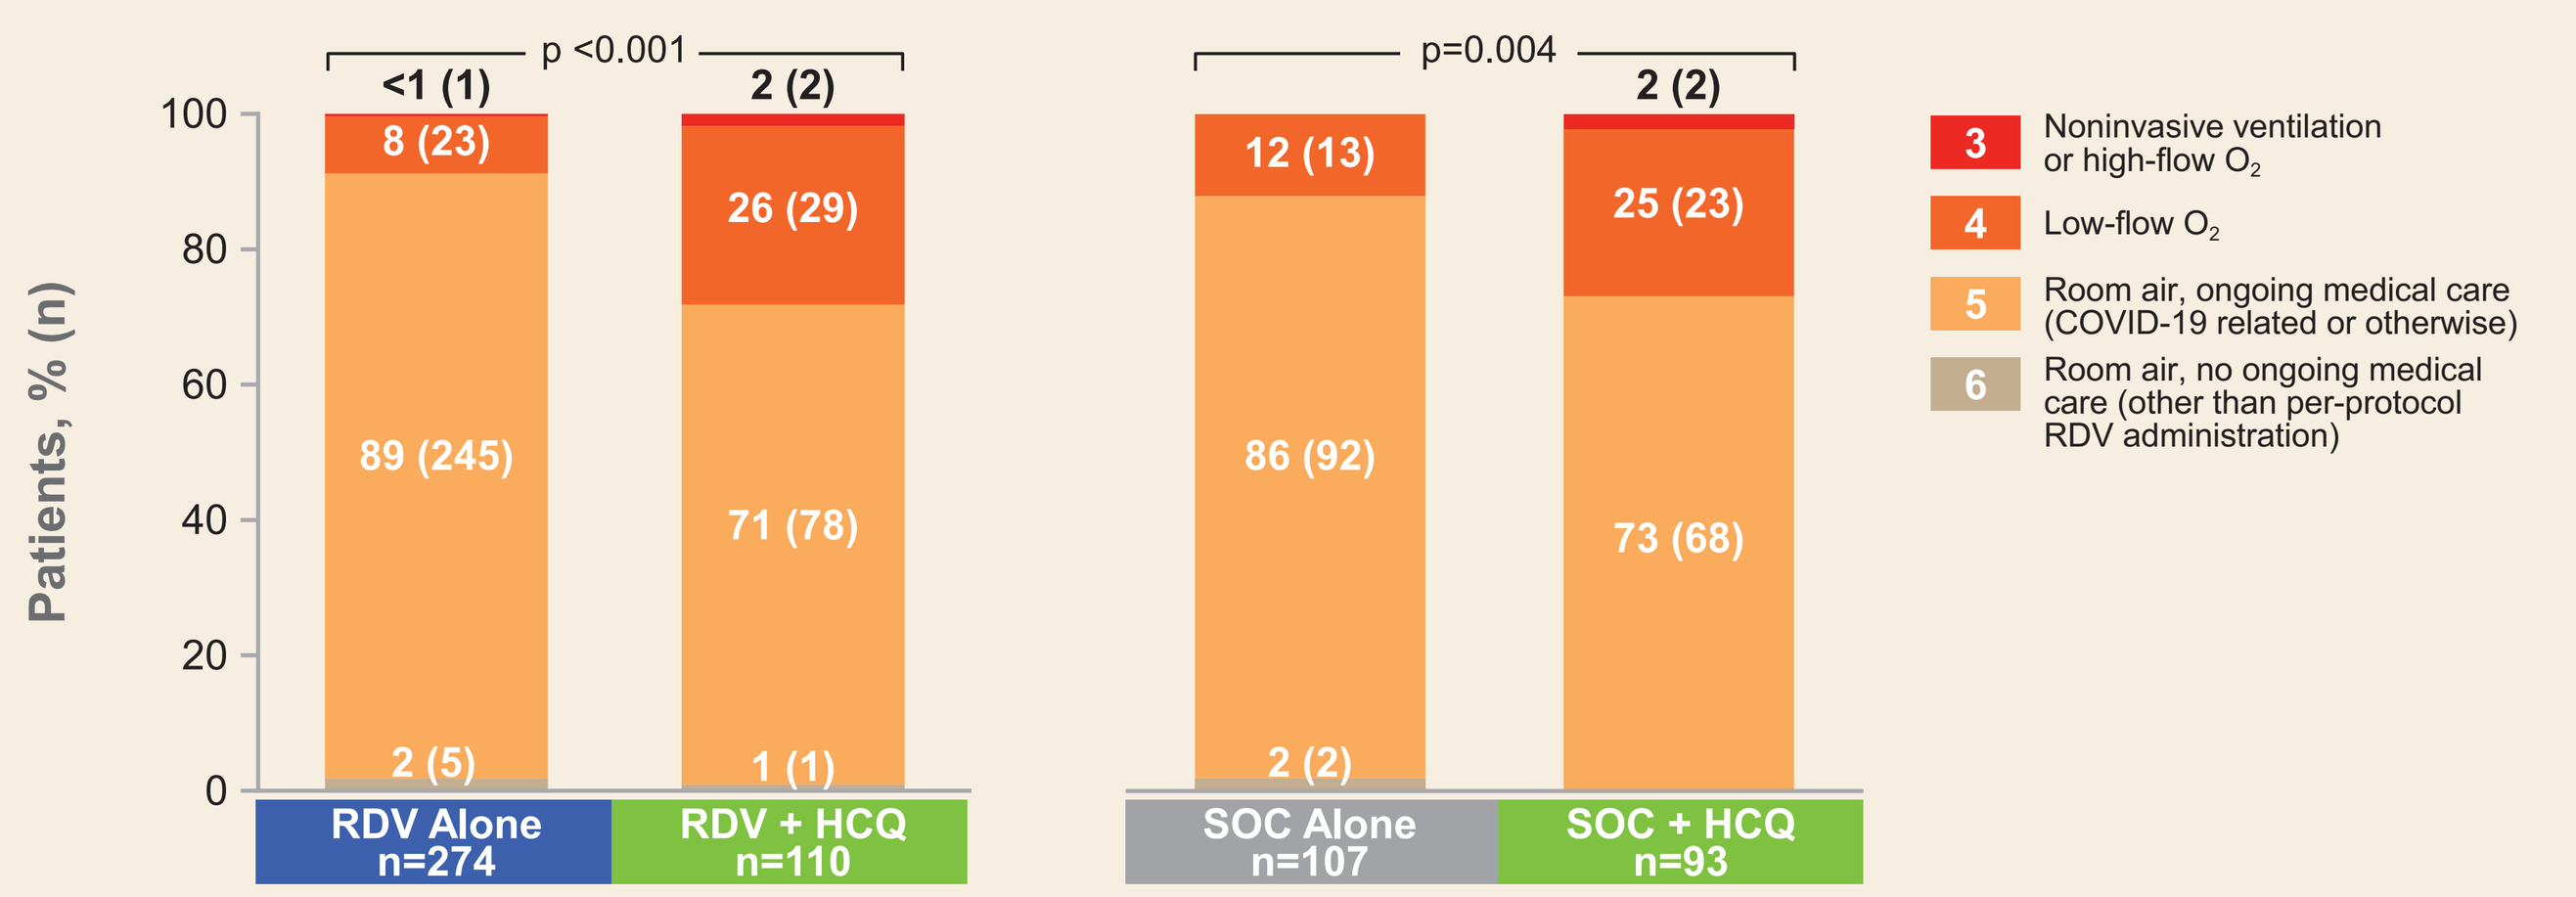

Patients, % (n)
p <0.001
<1 (1)
8 (23)
89 (245)
2 (5)
2 (2)
26 (29)
71 (78)
1 (1)
p=0.004
12 (13)
86 (92)
2 (2)
2 (2)
25 (23)
73 (68)
RDV Alone n=274
RDV + HCQ n=110
SOC Alone n=107
SOC + HCQ n=93
3 Noninvasive ventilation or high-flow O2
4 Low-flow O2
5 Room air, ongoing medical care (COVID-19 related or otherwise)
6 Room air, no ongoing medical care (other than per-protocol RDV administration)

- Compared with patients not on HCQ, more patients on HCQ were receiving O_2 support at BL (3–4 on ordinal scale)

HCQ Exposure Relative to RDV Dosing (n=110)

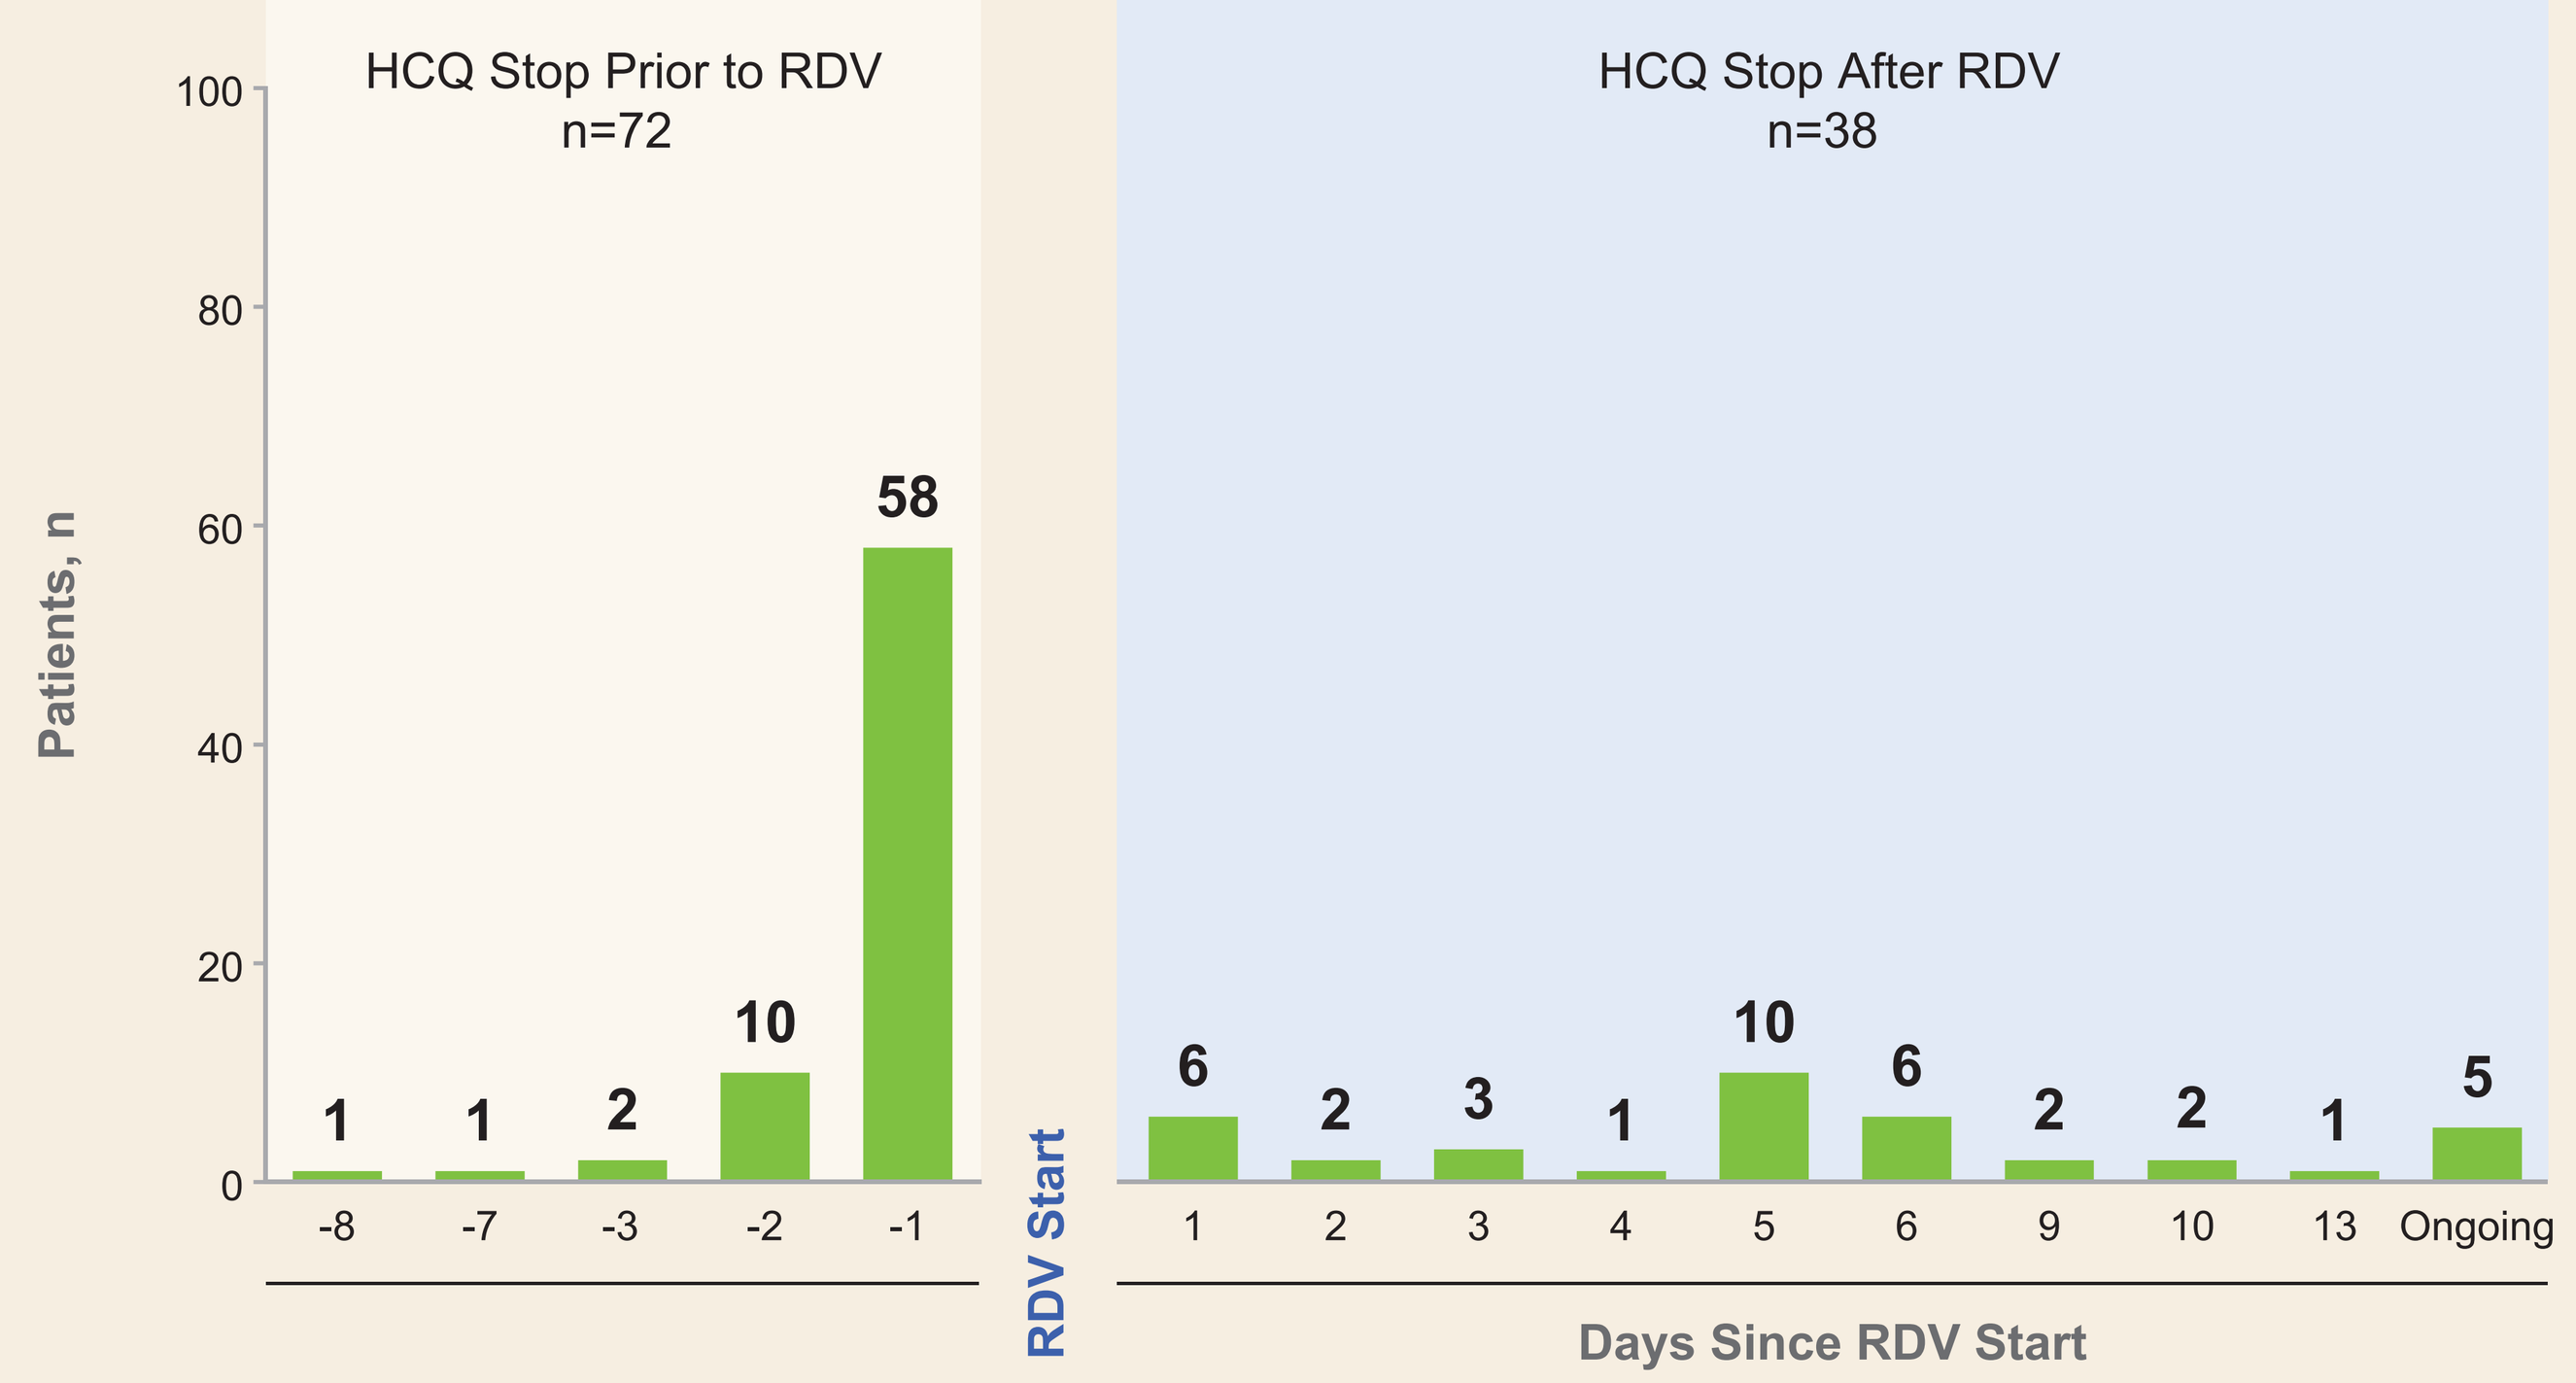

HCQ Stop Prior to RDV
n=72
HCQ Stop After RDV
n=38
Patients, n
1
1
2
10
58
-8
-7
-3
-2
-1
RDV Start
6
2
3
1
10
6
2
2
1
5
1
2
3
4
5
6
9
10
13
Ongoing
Days Since RDV Start

- Most patients treated with RDV + HCQ (65%) were discontinued from HCQ prior to start of RDV

HCQ Exposure Relative to Randomization in SOC Arm (n=93)

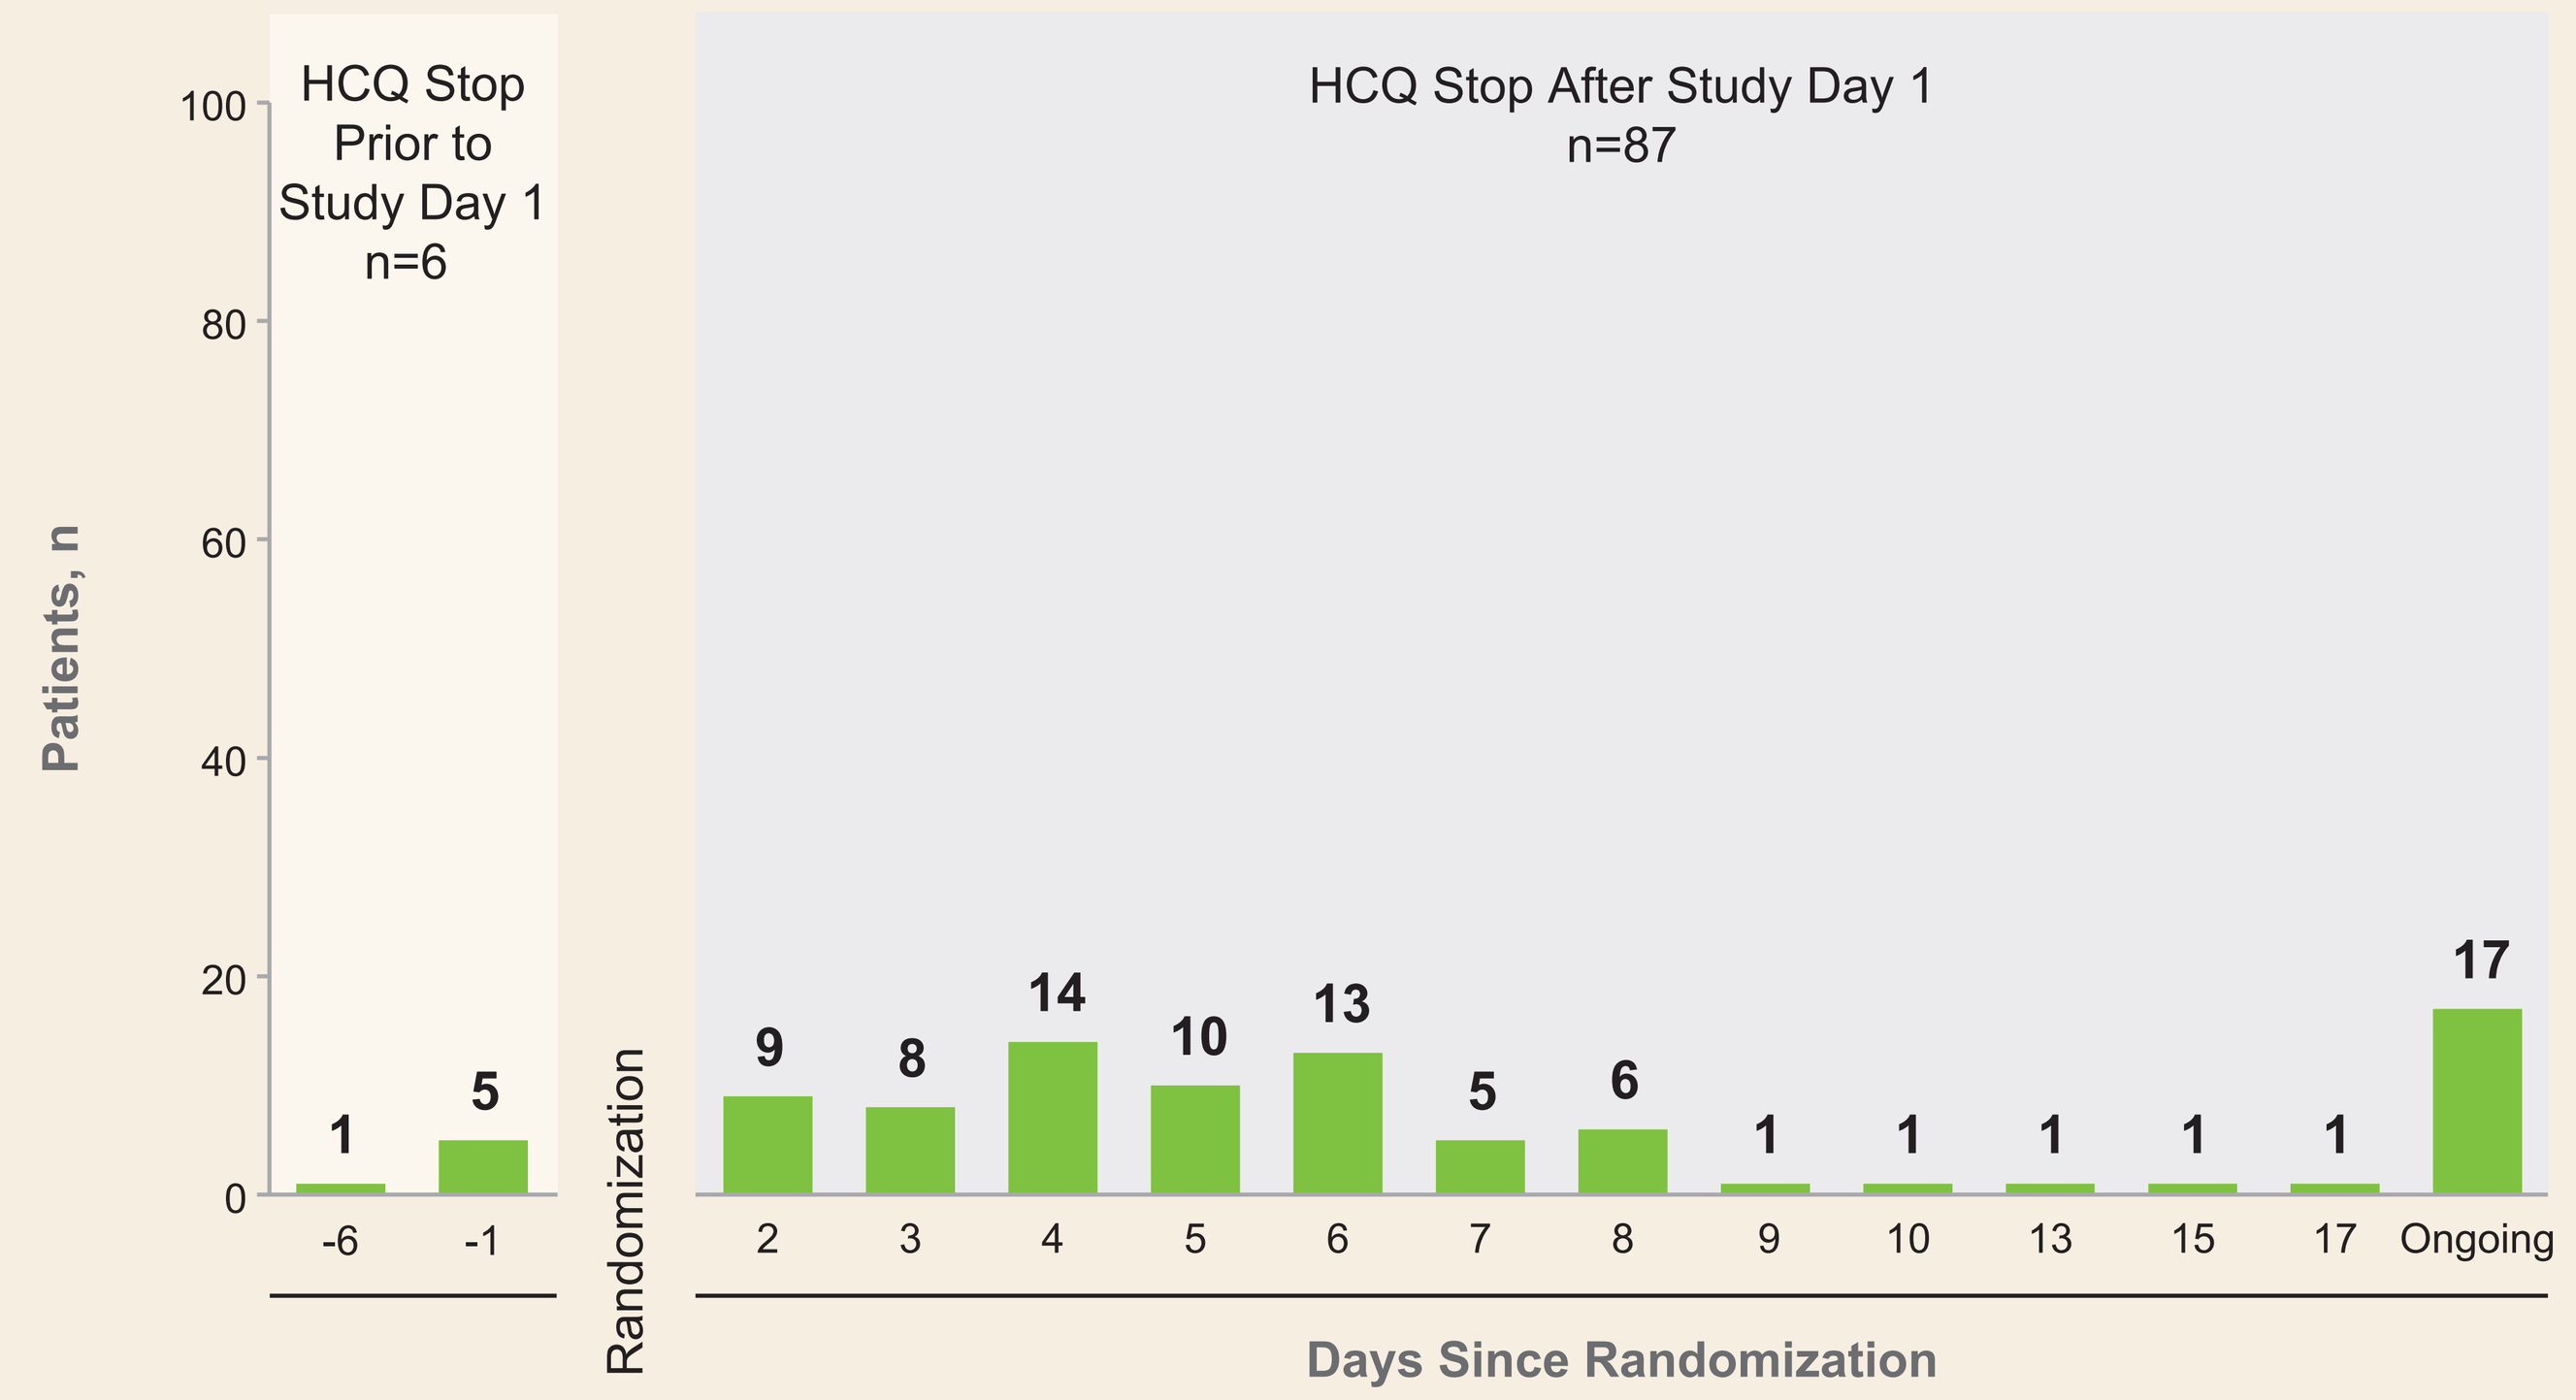

HCQ Stop Prior to Study Day 1
n=6
HCQ Stop After Study Day 1
n=87
Patients, n
1
5
-6
-1
Randomization
9
8
14
10
13
5
6
1
1
1
1
1
17
2
3
4
5
6
7
8
9
10
13
15
17
Ongoing
Days Since Randomization

- Most patients treated with SOC + HCQ (94%) were continued on HCQ therapy after randomization (94%)

Safety

n (%)	All RDV: RDV Alone n=274	All RDV: RDV + HCQ n=110	All RDV: p-Value*	SOC: SOC Alone n=107	SOC: SOC + HCQ n=93	SOC: p-Value*
AE	145 (53)	66 (60)	0.38	46 (43)	47 (51)	0.45
Grade ≥3	34 (12)	10 (9)	0.24	11 (10)	13 (14)	0.78
Serious AE	13 (5)	6 (5)	0.4	7 (7)	11 (12)	0.27
RDV related	1 (0)	0	NE	—	—	—
AE leading to discontinuation	7 (3)	5 (5)	NE	—	—	—
Death	4 (1)	1 (1)	NE	1 (1)	3 (3)	NE
Any treatment-emergent toxicity	173/253 (68)	86/106 (81)	0.03	69/96 (72)	67/90 (74)	0.95
White blood cell decrease	12/251 (5)	10/106 (9)	0.09	2/95 (2)	6/89 (7)	0.12
ALT increase	70/250 (28)	48/106 (45)	0.07	37/92 (40)	34/90 (38)	0.41
AST increase	74/248 (30)	38/104 (37)	0.74	28/92 (30)	32/90 (36)	0.78
Creatinine clearance decrease	45/248 (18)	26/106 (25)	0.26	25/94 (27)	30/89 (34)	0.35

*From logistic-regression model adjusting for BL clinical status (high- or low-flow O_2 vs room air), age (< vs ≥65 years), sex, race, and ethnicity. NE, not evaluable due to few AEs.

- Patients treated with RDV alone had fewer AEs and treatment-emergent toxicities than those treated with RDV + HCQ
- Patients treated with SOC alone and SOC + HCQ had similar rates of AEs and treatment-emergent toxicities

Time to Recovery
Patients on RDV With and Without HCQ

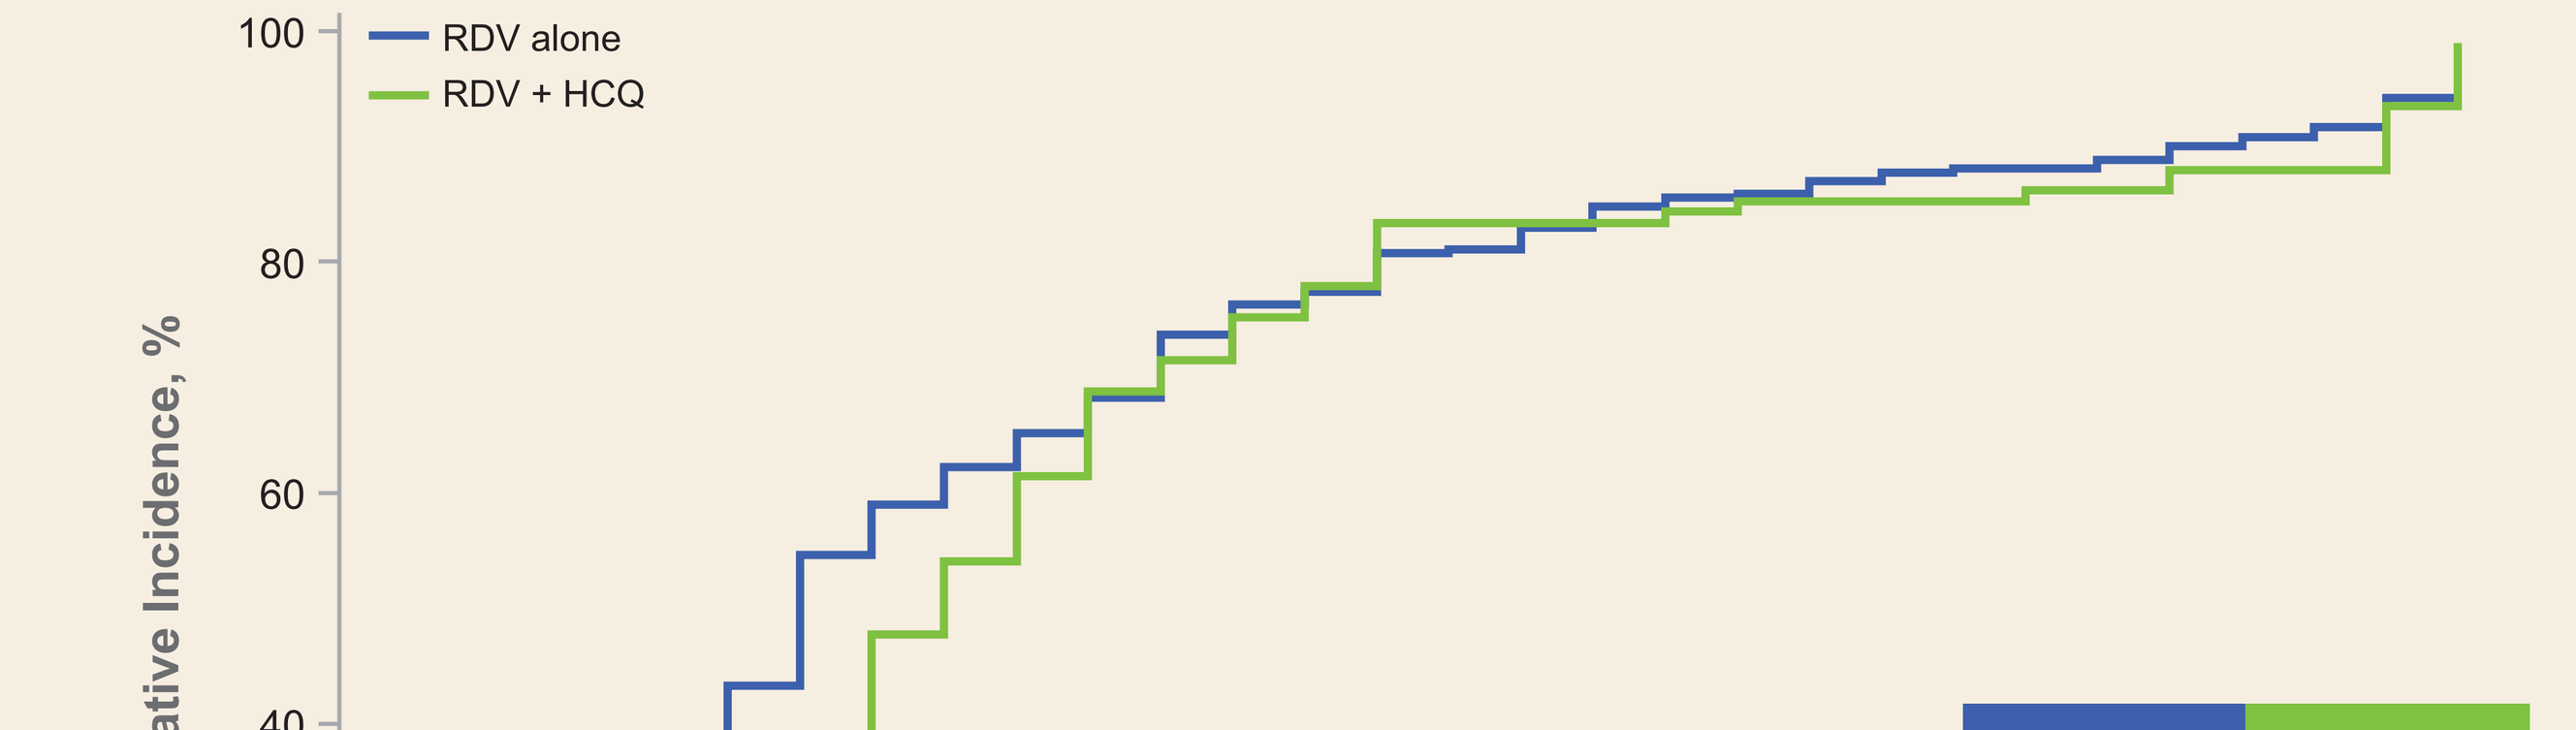

RDV alone
RDV + HCQ
Cumulative Incidence, %
Study Day

	RDV Alone n=274	RDV + HCQ n=110
Recovery, n (%)	255 (93)	104 (95)
Recovery rate ratio (95% CI)	0.88 (0.68, 1.14)	
Median time to recovery, day (Q1, Q3)	6 (4, 12)	8 (6, 12)

n at risk	BL	2	4	6	8	10	12	14	16	18	20	22	24	26	28	30
RDV alone	274	272	217	154	110	93	70	60	50	40	37	32	30	24	18	0
RDV + HCQ	110	108	103	85	57	41	30	23	17	17	15	15	14	12	12	0

CI, confidence interval; Q, quartile.

- After adjusting for BL covariates, there was no statistically significant difference in the rate of recovery between RDV patients on HCQ and RDV alone
- Similar results were observed in direction and magnitude for the rate of ≥2-point improvement

Time to Recovery
Patients on SOC With and Without HCQ

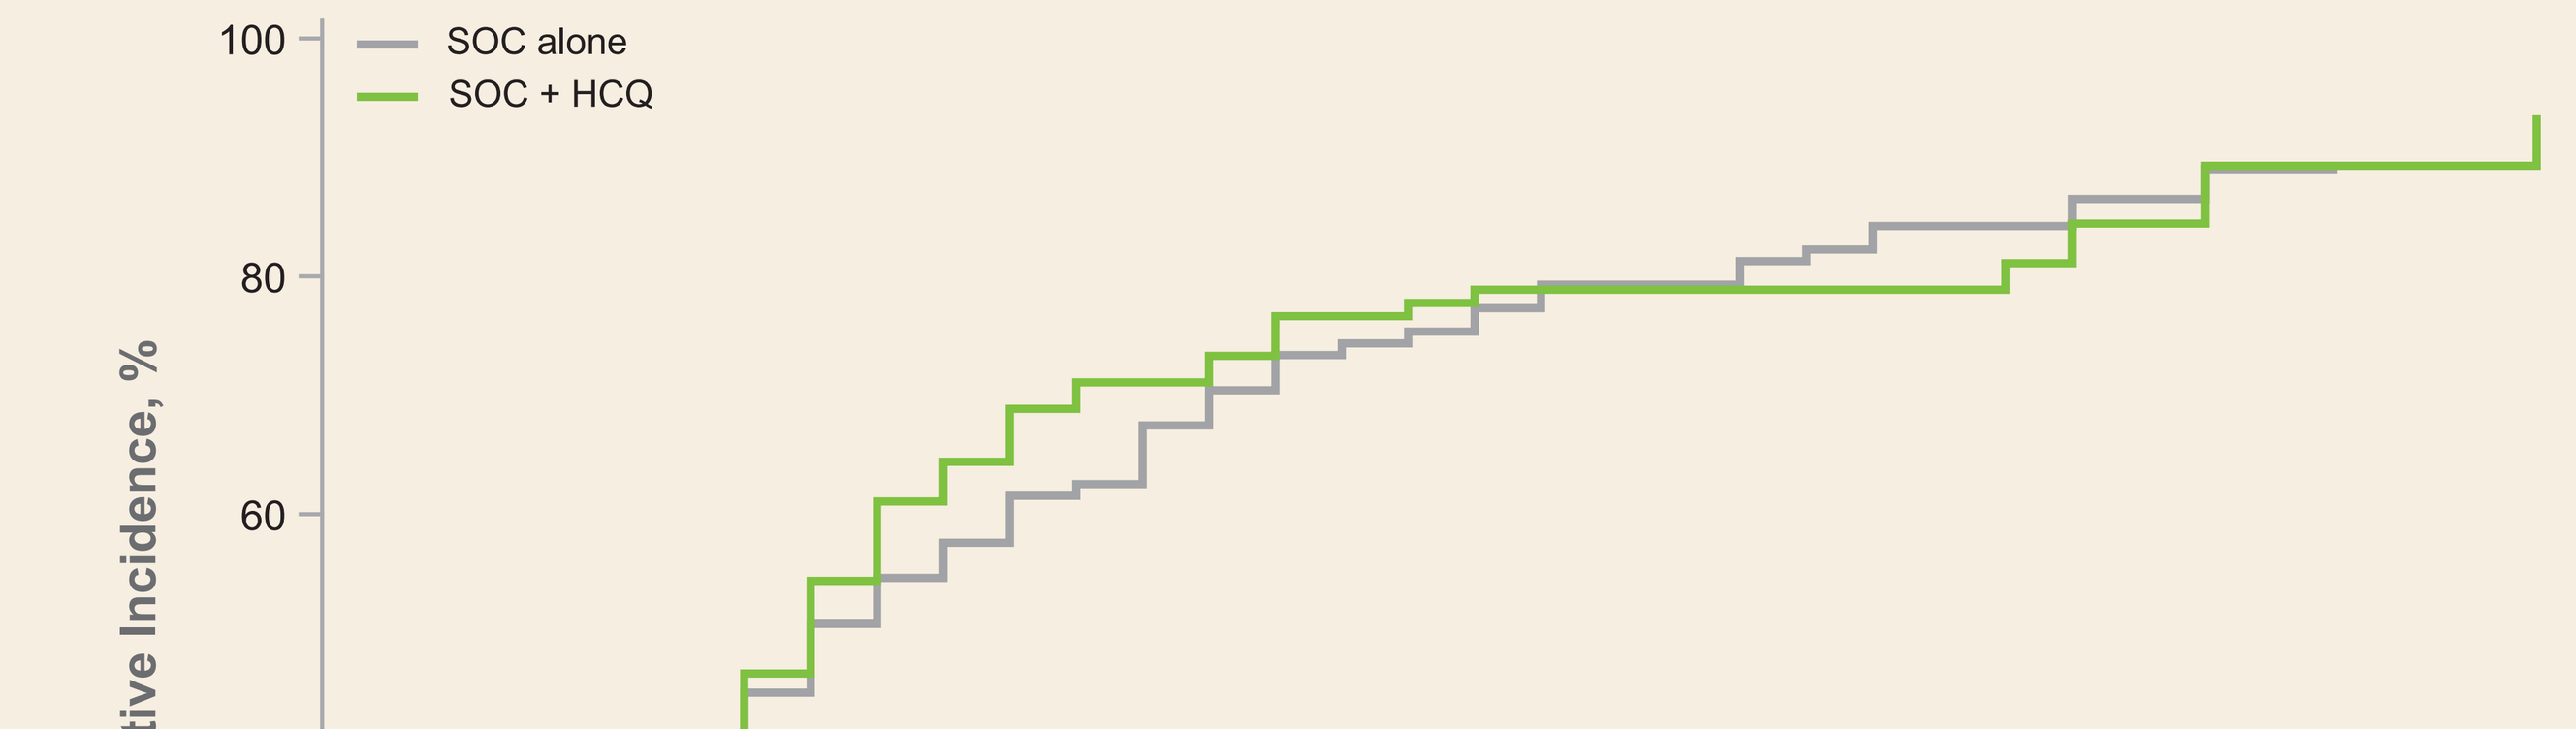

SOC alone
SOC + HCQ
Cumulative Incidence, %
Study Day

	SOC Alone n=107	SOC + HCQ n=93
Recovery, n (%)	91 (85)	82 (88)
Recovery rate ratio (95% CI)	1.13 (0.82, 1.56)	
Median time to recovery, day (Q1, Q3)	7 (4, 16)	7 (5, 14)

n at risk	BL	2	4	6	8	10	12	14	16	18	20	22	24	26	28	30	32
SOC alone	107	103	82	63	51	42	37	29	25	22	20	18	14	13	10	2	
SOC + HCQ	93	93	81	58	39	30	24	22	19	17	17	17	17	15	11	2	2

- After adjusting for BL covariates, there was no statistically significant difference in the rate of recovery between SOC patients on HCQ and SOC alone
- Similar results were observed in direction and magnitude for the rate of ≥2-point improvement

Conclusions

- In patients with moderate COVID-19, concomitant HCQ may delay recovery on RDV and showed no impact on recovery with SOC alone
- Patients treated with HCQ had more safety events than those not receiving HCQ irrespective of RDV treatment

References: 1. Gordon CJ, et al. J Biol Chem 2020;295:6785-97; 2. Goldman JD, et al. N Engl J Med 2020 May 27;NEJMoa2015301; 3. Spinner CD, et al. JAMA 2020;324:1048-57; 4. US Food and Drug Administration. Fact Sheet for Patients and Parent/Caregivers Emergency Use Authorization (EUA) of Hydroxychloroquine Sulfate for Treatment of COVID-19 in Certain Hospitalized Patients; 3/28/20; revoked 6/15/20.

Acknowledgments: We express our solidarity with those who are or have been ill with COVID-19, their families, and the healthcare workers on the frontlines of this pandemic. We extend our thanks and appreciation to the GS-US-540-5774 study participants, their partners and families, the frontline healthcare workers caring for them, the study staff, and the study investigators. This study was funded by Gilead Sciences, Inc.

Presented at IDWeek 2020, October 21–25, 2020

©2020 Gilead Sciences, Inc. All rights reserved.