561

Safety of Remdesivir vs Standard of Care in Patients With Moderate COVID-19

Gerard Criner,[1] Mi Young Ahn,[2] Gregory Huhn,[3] Aruna Subramanian,[4] Carlos Lumbreras,[5] Stefan Schmiedel,[6] Robert H. Hyland,[7] Vithika Suri,[7] Huyen Cao,[7] Hongyuan Wang,[7] Devi SenGupta,[7] Anand Chokkalingam,[7] Anu Osinusi,[7] Diana M. Brainard,[7] Yao-Shen Chen,[8] Huldrych Günthard,[9] D. Jose Sanz-Moreno,[10] Judith A. Aberg,[11] Emanuele Nicastri,[12] Tak Yin Owen Tsang[13]

[1]Temple University Hospital, Philadelphia, PA; [2]Seoul Medical Center, Seoul, Republic of Korea; [3]John H. Stroger, Jr. Hospital of Cook County, Chicago, IL; [4]Stanford University School of Medicine, Stanford, CA; [5]Hospital Universitario 12 de Octubre, Madrid, Spain; [6]Universitätsklinikum Hamburg-Eppendorf, Hamburg, Germany; [7]Gilead Sciences, Inc., Foster City, CA; [8]Kaohsiung Veterans General Hospital, Kaohsiung City, Taiwan; [9]Universitätsspital Zürich, Switzerland; [10]Hospital Universitario Principe de Asturias, Madrid; [11]Icahn School of Medicine at Mount Sinai, New York, NY; [12]U.O.C. Malattie Infettive ad Alta Intensità di Cure ed Altamente Contagiose, Istituto Nazionale Malattie Infettive Lazzaro Spallanzani, Roma, Italy; [13]Princess Margaret Hospital, Hong Kong, People's Republic of China

Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
800-445-3235

Introduction

- Remdesivir (RDV) is a broad-spectrum nucleotide analog prodrug that inhibits viral RNA polymerases, and has demonstrated potent in vitro and in vivo activity against severe acute respiratory syndrome coronavirus-2 (SARS-CoV-2)[1-3]
- Results from the Phase 3 SIMPLE-Moderate study (GS-US-540-5774; NCT04292730) showed that hospitalized COVID-19 patients not requiring O_2 support who were treated with 5 days of RDV had higher odds for improvement compared with those treated with standard of care (SOC) at Day 11[4]
- The US Food & Drug Administration Emergency Use Authorization for RDV was broadened to include hospitalized patients irrespective of O_2 support based on these data

Objective

- To assess safety in patients with moderate COVID-19 who received RDV for 5 or 10 days vs SOC in the SIMPLE-Moderate study

Methods

Phase 3, Randomized, Open-label, Multicenter Study

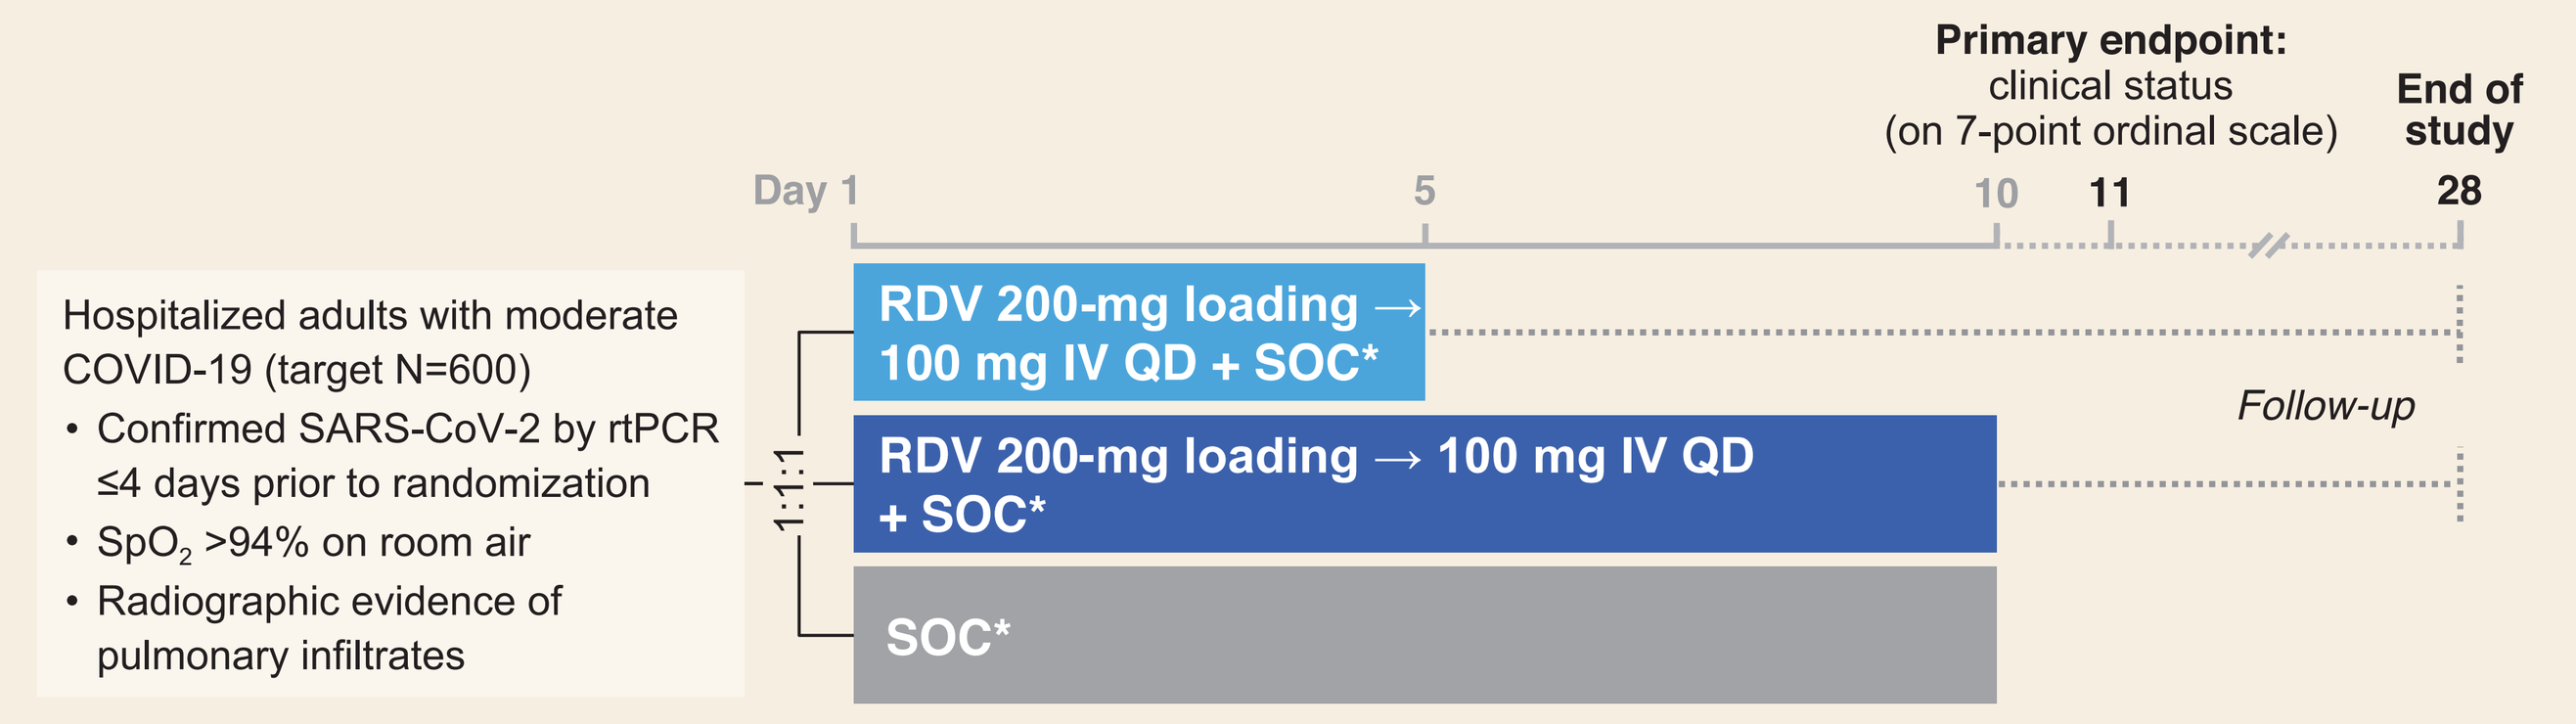

Primary endpoint:
clinical status
(on 7-point ordinal scale)
End of study
Day 1
5
10
11
28
Hospitalized adults with moderate COVID-19 (target N=600)
• Confirmed SARS-CoV-2 by rtPCR ≤4 days prior to randomization
• SpO2 >94% on room air
• Radiographic evidence of pulmonary infiltrates
1:1:1
RDV 200-mg loading → 100 mg IV QD + SOC*
RDV 200-mg loading → 100 mg IV QD + SOC*
SOC*
Follow-up

*SOC continued throughout study duration; agents with actual or possible direct-acting antiviral activity against SARS-CoV-2 were not allowed. rtPCR, reverse-transcriptase–polymerase chain reaction assay; SpO_2, peripheral oxygen saturation.

- Study enrolled March 15–April 18, 2020; date of final follow-up May 20
- 105 centers in 12 countries: Asia (Hong Kong, Republic of Korea, Singapore, and Taiwan), Europe (France, Germany, Italy, Netherlands, Spain, Switzerland, and UK), and North America (USA)
- Key exclusion criteria:
 - Alanine aminotransferase (ALT) or aspartate aminotransferase (AST) >5x upper limit of normal (ULN); creatinine clearance <50 mL/min
 - Use of any experimental treatment for COVID-19 ≤24 hours prior to dosing
 - Any requirement for invasive mechanical ventilation (IMV) at screening
- Primary endpoint of distribution of clinical status on a 7-point ordinal scale at Day 11 was previously reported[4]

Clinical Outcomes Measured on 7-Point Ordinal Scale[4]

	1	Death
Hospitalized +	2	IMV or ECMO
Hospitalized +	3	Noninvasive ventilation or high-flow O_2
Hospitalized +	4	Low-flow O_2
Hospitalized +	5	Room air, ongoing medical care (COVID-19 related or otherwise)
Hospitalized +	6	Room air, no ongoing medical care (other than per-protocol RDV administration)
	7	Discharged

ECMO, extracorporeal membrane oxygenation.

- Patients were assessed by physical examination, respiratory status (respiratory rate, type of O_2 support, blood O_2 saturation, and radiographic findings), adverse events (AEs), and concomitant medications
- On Days 1, 3, 5, 8, 10, and 14, blood samples were obtained for blood counts, serum creatinine, glucose, total bilirubin, and liver transaminases

Results

Patient Disposition

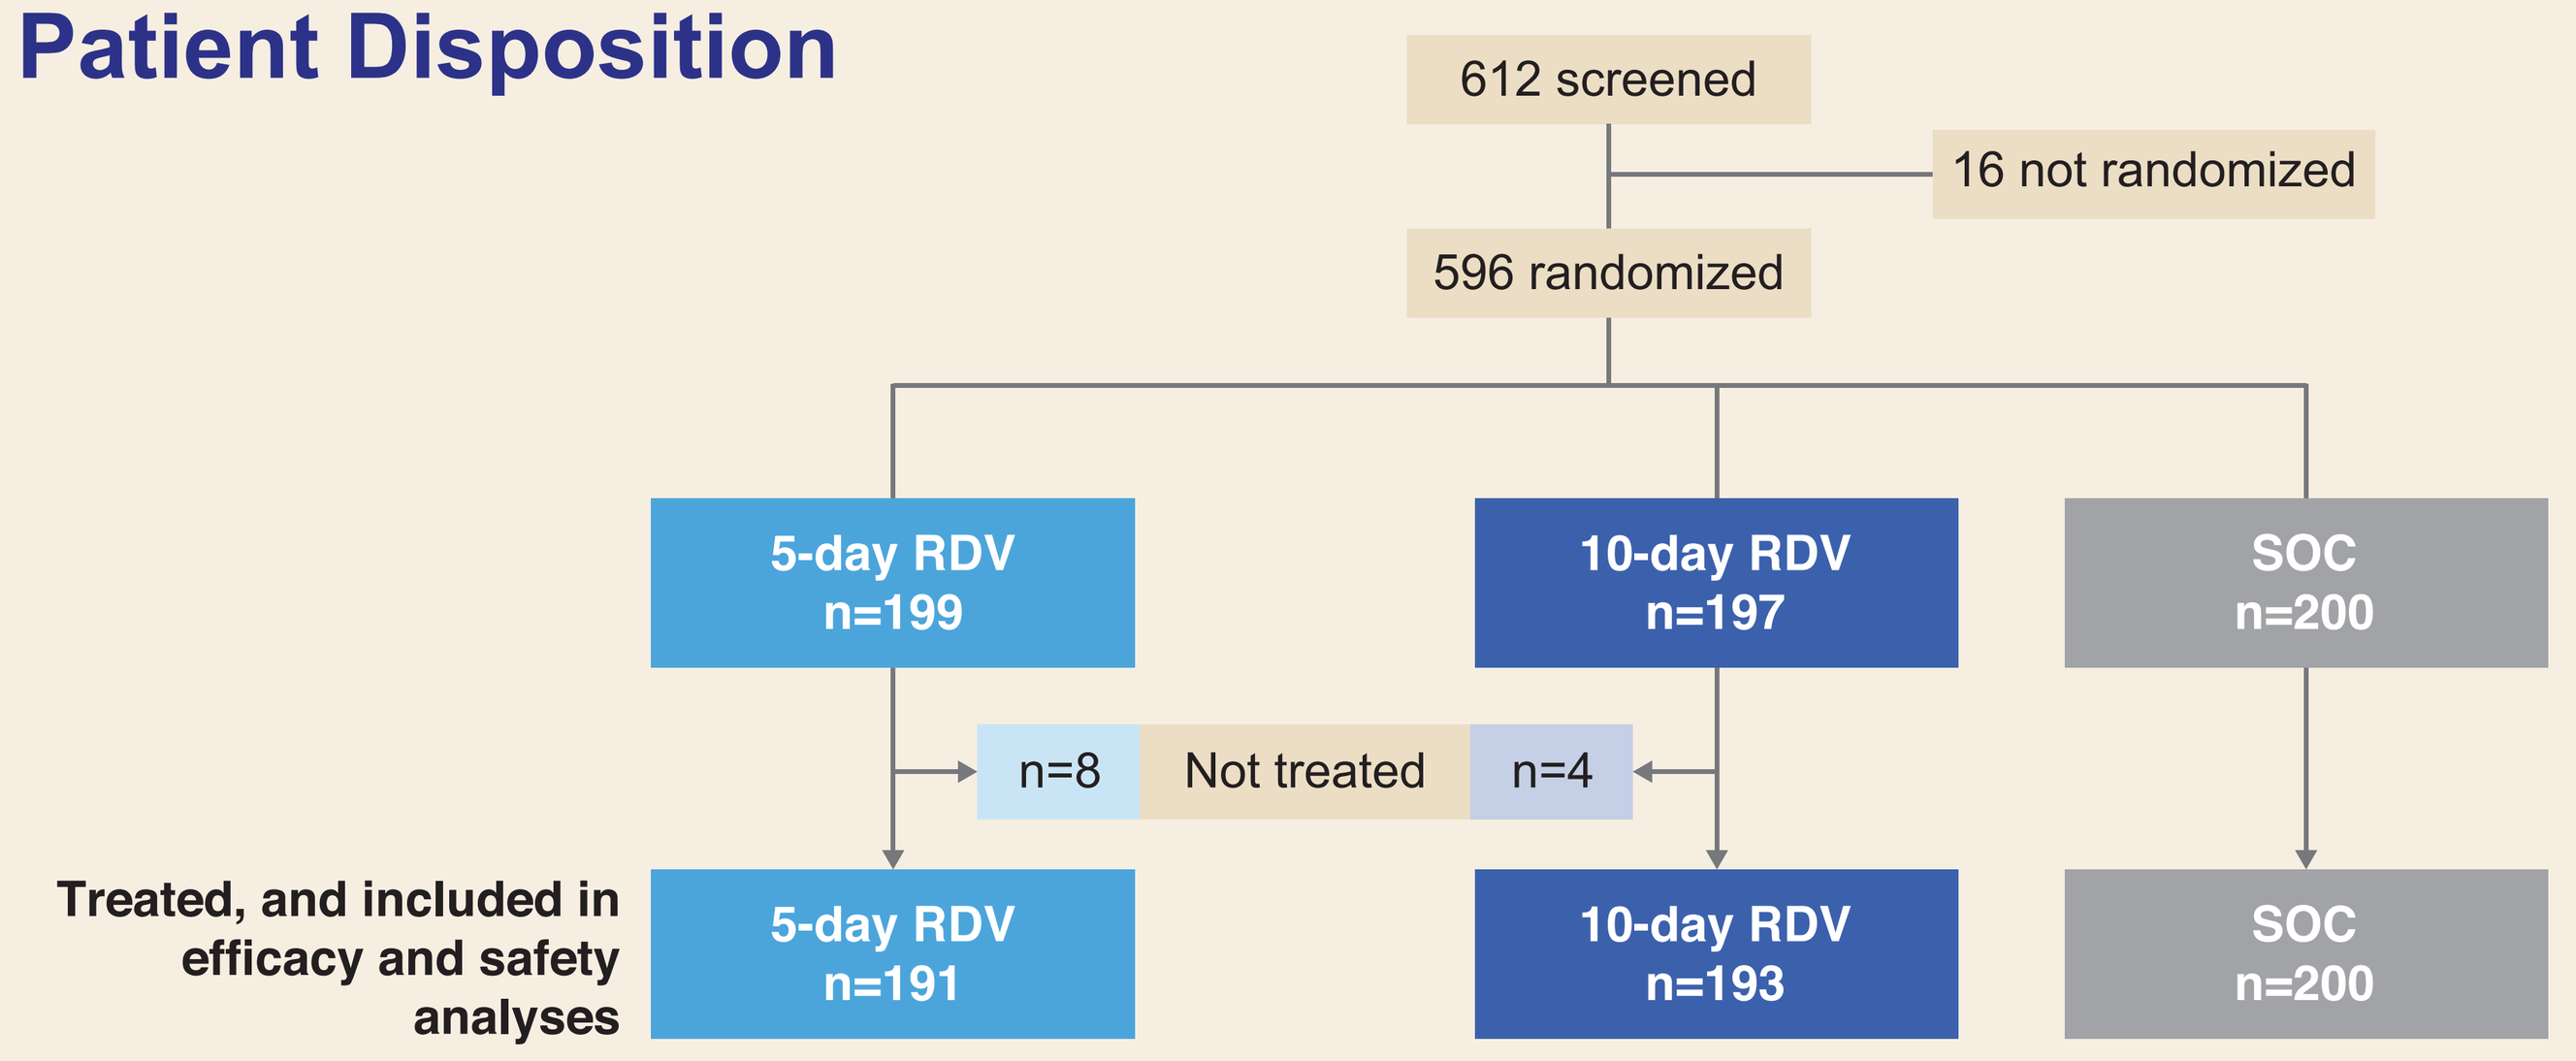

612 screened
16 not randomized
596 randomized
5-day RDV n=199
10-day RDV n=197
SOC n=200
n=8
Not treated
n=4
Treated, and included in efficacy and safety analyses
5-day RDV n=191
10-day RDV n=193
SOC n=200

Baseline Demographics and Characteristics

		5-Day RDV n=191	10-Day RDV n=193	SOC n=200
Median age, year (range)		58 (12–90)	56 (20–94)	57 (23–95)
Men, n (%)		114 (60)	118 (61)	125 (63)
Median body mass index, kg/m² (range)		27 (17–77)	28 (16–63)	27 (16–54)
Region, n (%)	USA	76 (40)	99 (51)	85 (43)
	Rest of world	115 (60)	94 (49)	115 (57)
Race, n/N (%)	White	109/186 (59)	107/188 (57)	112/193 (58)
	Black*	35/186 (19)	37/188 (20)	27/193 (14)
	Asian	34/186 (18)	31/188 (16)	37/193 (19)
	Other†	8/186 (4)	13/188 (7)	17/193 (9)
Clinical status on 7-point scale, n (%)				
3: Noninvasive ventilation or high-flow O_2		2 (1)	1 (1)	2 (1)
4: Low-flow O_2		29 (15)	23 (12)	36 (18)
5: Room air, but requiring medical care		160 (84)	163 (84)	160 (80)
6: Room air, no longer requiring ongoing medical care		0	6 (3)	2 (1)
Preexisting conditions, n (%)	Cardiovascular disease	111 (58)	111 (58)	107 (54)
	Hypertension	82 (43)	85 (44)	81 (41)
	Diabetes mellitus	71 (37)	85 (44)	76 (38)
	Asthma	22 (12)	31 (16)	28 (14)
Median duration of hospitalization before study Day 1, day (IQR)		2 (1–3)	2 (1–3)	2 (1–3)
Median duration of symptoms before study Day 1, day (IQR)		8 (5–11)	8 (5–11)	9 (6–11)

*33% (85/260) of all US patients; †Includes American Indian, Alaska Native, Native Hawaiian, Pacific Islander, and other. IQR, interquartile range.

Efficacy: Clinical Status at Days 11 and 28

- Patients treated with 5 days of RDV had significantly higher odds of better clinical status at Day 11 (odds ratio vs SOC 1.65; 95% confidence interval 1.09, 2.48; p=0.02)
 - The difference in clinical status at Day 11 between 10 days of RDV and SOC was not significantly different (p=0.18)
 - Detailed efficacy results are presented in Marty F, et al. IDWeek 2020, oral 72

Overall Safety

n (%)	5-Day RDV n=191	10-Day RDV n=193	SOC n=200
Any AE	98 (51)	113 (59)	93 (47)
Any Grade ≥3 AE	20 (10)	24 (12)	24 (12)
SAE	9 (5)	10 (5)	18 (9)
AE leading to discontinued treatment	4 (2)	8 (4)	NA
Death*	2 (1)	3 (2)	4 (2)
AEs occurring in >5% in any treatment group			
Nausea	19 (10)	18 (9)	6 (3)
Diarrhea	12 (6)	10 (5)	14 (7)
Hypokalemia	10 (5)	13 (7)	4 (2)
Headache	10 (5)	10 (5)	5 (3)

*None related to treatment. NA, not applicable; SAE, serious AE.

- Overall, there were similar rates of Grade ≥3 AEs in the SOC and RDV arms, and higher rates of SAEs in the SOC arm

Adverse Events by Age, Sex, and Race

	<65 Years			≥65 Years		
n (%)	5-Day RDV n=142	10-Day RDV n=141	SOC n=142	5-Day RDV n=49	10-Day RDV n=52	SOC n=58
Any AE	69 (49)	80 (57)	58 (41)	29 (59)	33 (63)	35 (60)
Any Grade ≥3 AE	10 (7)	14 (10)	13 (9)	10 (20)	10 (19)	11 (19)
Study drug-related AE	27 (19)	20 (14)	NA	9 (18)	5 (10)	NA
SAE	4 (3)	5 (4)	7 (5)	5 (10)	5 (10)	11 (19)
AE leading to discontinued treatment	4 (3)	8 (6)	NA	0	0	NA
Death	0	0	1 (1)	2 (4)	3 (6)	3 (5)

	Men			Women		
n (%)	n=114	n=118	n=125	n=77	n=75	n=75
Any AE	52 (46)	68 (58)	56 (45)	46 (60)	45 (60)	37 (49)
Any Grade ≥3 AE	14 (12)	15 (13)	18 (14)	6 (8)	9 (12)	6 (8)
Study drug-related AE	17 (15)	14 (12)	NA	19 (25)	11 (15)	NA
SAE	7 (6)	7 (6)	14 (11)	2 (3)	3 (4)	4 (5)
AE leading to discontinued treatment	3 (3)	7 (6)	NA	1 (1)	1 (1)	NA
Death	2 (2)	1 (1)	3 (2)	0	2 (3)	1 (1)

	Asian			Black		
n (%)	n=34	n=31	n=37	n=35	n=37	n=27
Any AE	19 (56)	18 (58)	15 (41)	14 (40)	19 (51)	8 (30)
Any Grade ≥3 AE	2 (6)	3 (10)	0	3 (9)	6 (16)	4 (15)
Study drug-related AE	10 (29)	6 (19)	NA	2 (6)	3 (8)	NA
SAE	1 (3)	0	1 (3)	3 (9)	4 (11)	5 (19)
AE leading to discontinued treatment	2 (6)	0	NA	0	3 (8)	NA
Death	0	0	0	1 (3)	1 (3)	1 (4)

	White			Other		
n (%)	n=109	n=107	n=112	n=13	n=18	n=24
Any AE	60 (55)	66 (62)	58 (52)	5 (38)	10 (56)	12 (50)
Any Grade ≥3 AE	14 (13)	12 (11)	18 (16)	1 (8)	3 (17)	2 (8)
Study drug-related AE	21 (19)	15 (14)	NA	3 (23)	1 (6)	NA
SAE	5 (5)	4 (4)	10 (9)	0	2 (11)	2 (8)
AE leading to discontinued treatment	1 (1)	3 (3)	NA	1 (8)	2 (11)	NA
Death	1 (1)	2 (2)	2 (2)	0	0	1 (4)

- RDV was well tolerated compared with SOC and led to low discontinuation rates across patient populations irrespective of age, sex, or race

Deaths

	Age, Year	Sex	Race/ Ethnicity	Baseline Clinical Status	Cause of Death*
5-Day RDV	80	Man	Black	Room air	Respiratory failure
	73	Man	White	Room air	ARDS progression with worsening lung compliance
10-Day RDV	86	Man	White	Room air	COVID-19 pneumonia, blood stream infection, and ascending thoracic aortic aneurysm
	76	Woman	White	Room air	Acute hypoxic respiratory failure
	83	Woman	Black	Room air	Complete heart block
SOC	74	Man	White	Low-flow O_2	COVID-19
	89	Man	Black	Low-flow O_2	COVID-19
	89	Woman	White	Low-flow O_2	COVID-19
	64	Man	Other	Low-flow O_2	On BiPAP; likely hypoxic and thus went into cardiac arrest

*As assessed by investigator. ARDS, acute respiratory distress syndrome; BiPAP, bilevel positive airway pressure.

- No deaths were assessed as related to RDV

Renal-Related Adverse Events by Preferred Term

n (%)	5-Day RDV n=191	10-Day RDV n=193	SOC n=200
Any renal-related AE	3 (2)	4 (2)	4 (2)
Acute kidney injury	1 (1)	1 (1)	2 (1)
Blood creatinine increased	0	2 (1)	2 (1)
Creatinine renal clearance decreased	1 (1)	0	0
Glomerular filtration rate decreased	1 (1)	0	0
Urine output decreased	0	1 (1)	0

- The incidence of renal-related AEs was similar between patients treated with RDV and SOC
- There were no renal-related SAEs in the 5- or 10-day RDV groups, while 1 patient in the SOC group had ongoing Grade 4 acute kidney injury

Laboratory Abnormalities

n/N (%)		5-Day RDV n=191	10-Day RDV n=193	SOC n=200
Laboratory abnormalities	Any grade	131/180 (73)	128/179 (72)	136/186 (73)
	Grade 3	18/180 (10)	25/179 (14)	25/186 (13)
	Grade 4	5/180 (3)	4/179 (2)	9/186 (5)
ALT increase	Any grade	61/179 (34)	57/177 (32)	71/182 (39)
	Grade 3 (≥5–<10x ULN)	4/179 (2)	6/177 (3)	11/182 (6)
	Grade 4 (≥10x ULN)	0	0	3 (2)
AST increase	Any grade	56/177 (32)	56/175 (32)	60/182 (33)
	Grade 3 (≥5–<10x ULN)	3/177 (2)	2/175 (1)	6/182 (3)
	Grade 4 (≥10x ULN)	1/177 (1)	0	5/182 (3)
Creatinine clearance decrease	Any grade	26/178 (15)	45/176 (26)	55/183 (30)
	Grade 3 (30–<60 mL/min or 30–<50% decrease from baseline)	4/178 (2)	7/176 (4)	9/183 (5)
	Grade 4 (<30 mL/min or ≥50% decrease from baseline or dialysis needed)	0	2/176 (1)	5/183 (3)

- Rates of laboratory abnormalities were similar in the RDV and SOC arms

Creatinine Clearance Over Time*

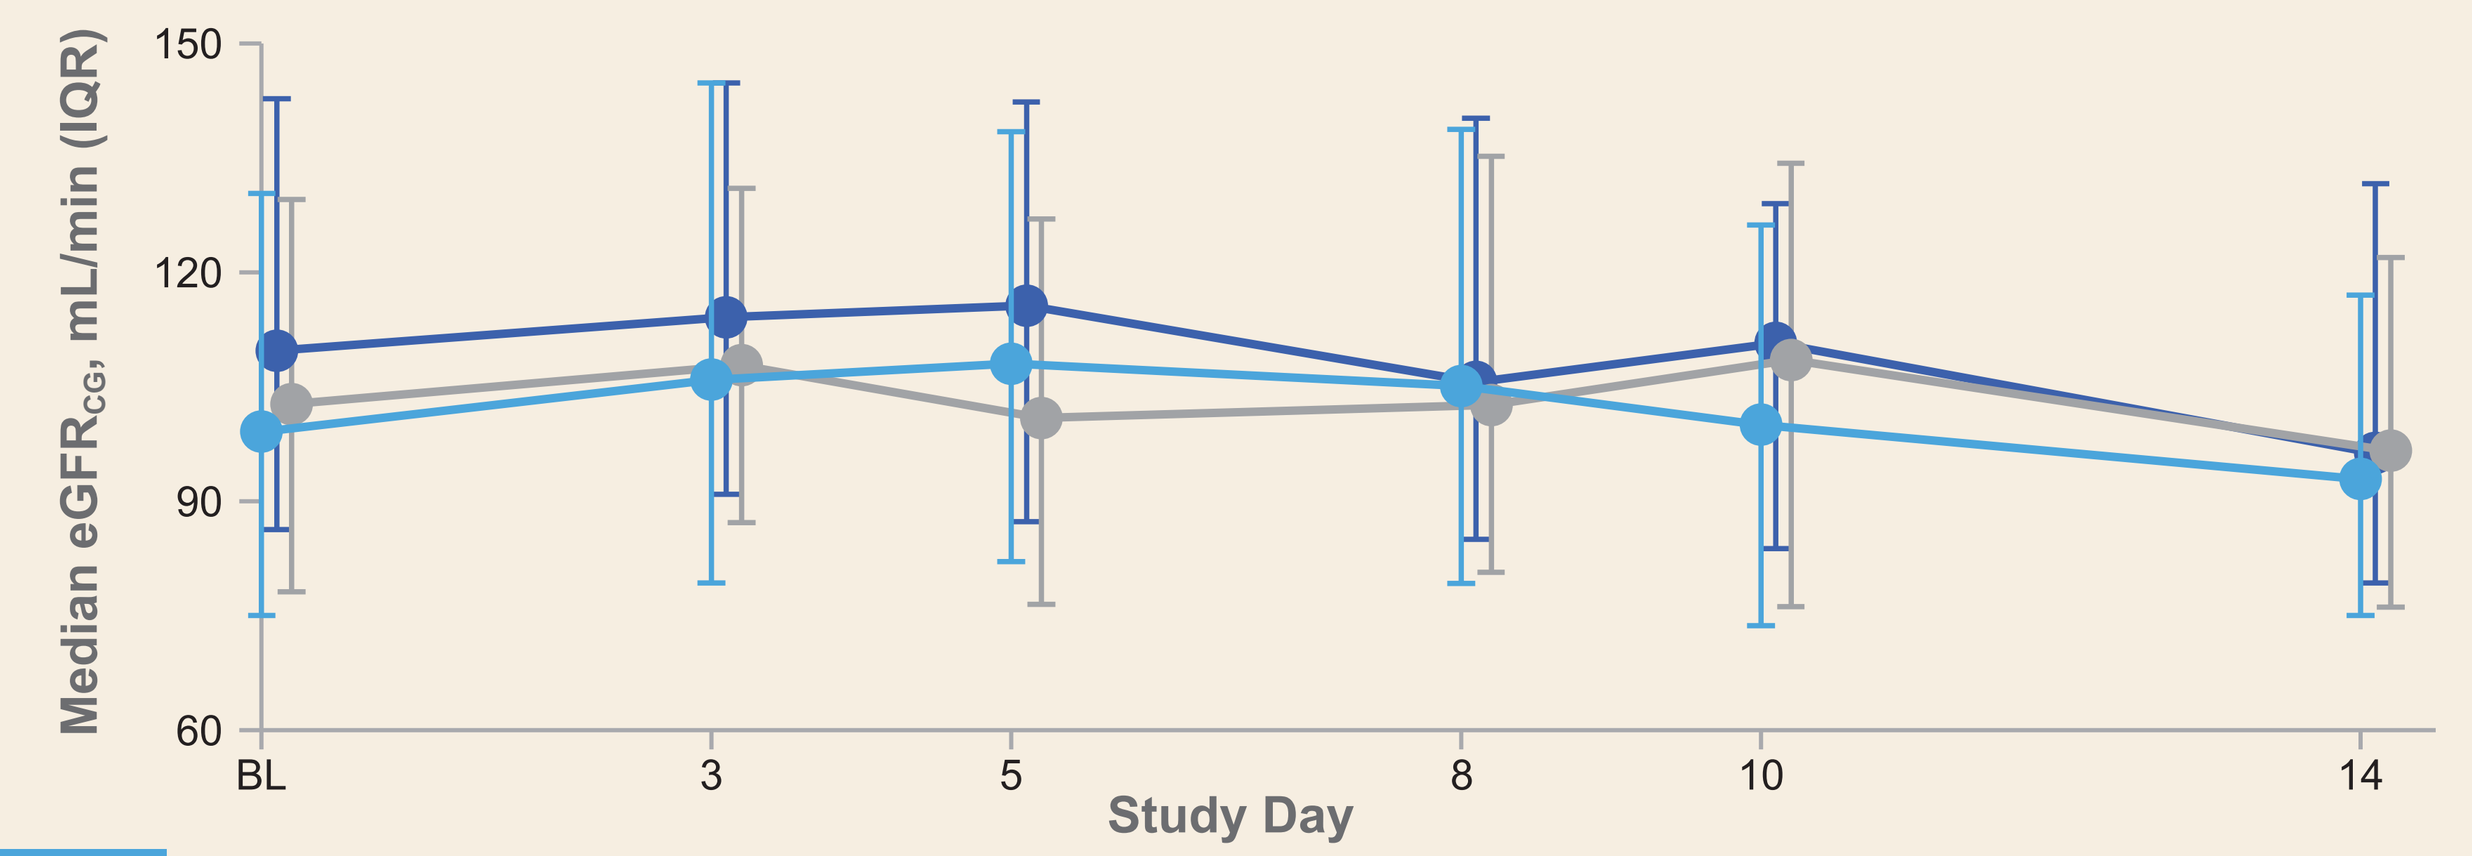

Median eGFRCG, mL/min (IQR)
150
120
90
60
BL
3
5
8
10
14
Study Day

n=	BL	3	5	8	10	14
5-Day RDV	188	174	152	79	53	43
10-Day RDV	190	168	139	95	78	48
SOC	199	176	136	82	69	50

*Data not scheduled for collection past Day 14. $eGFR_{CG}$, estimated glomerular filtration rate by Cockcroft-Gault.

- There were no significant changes in creatinine clearance in patients treated with RDV vs SOC

ALT and AST Over Time*

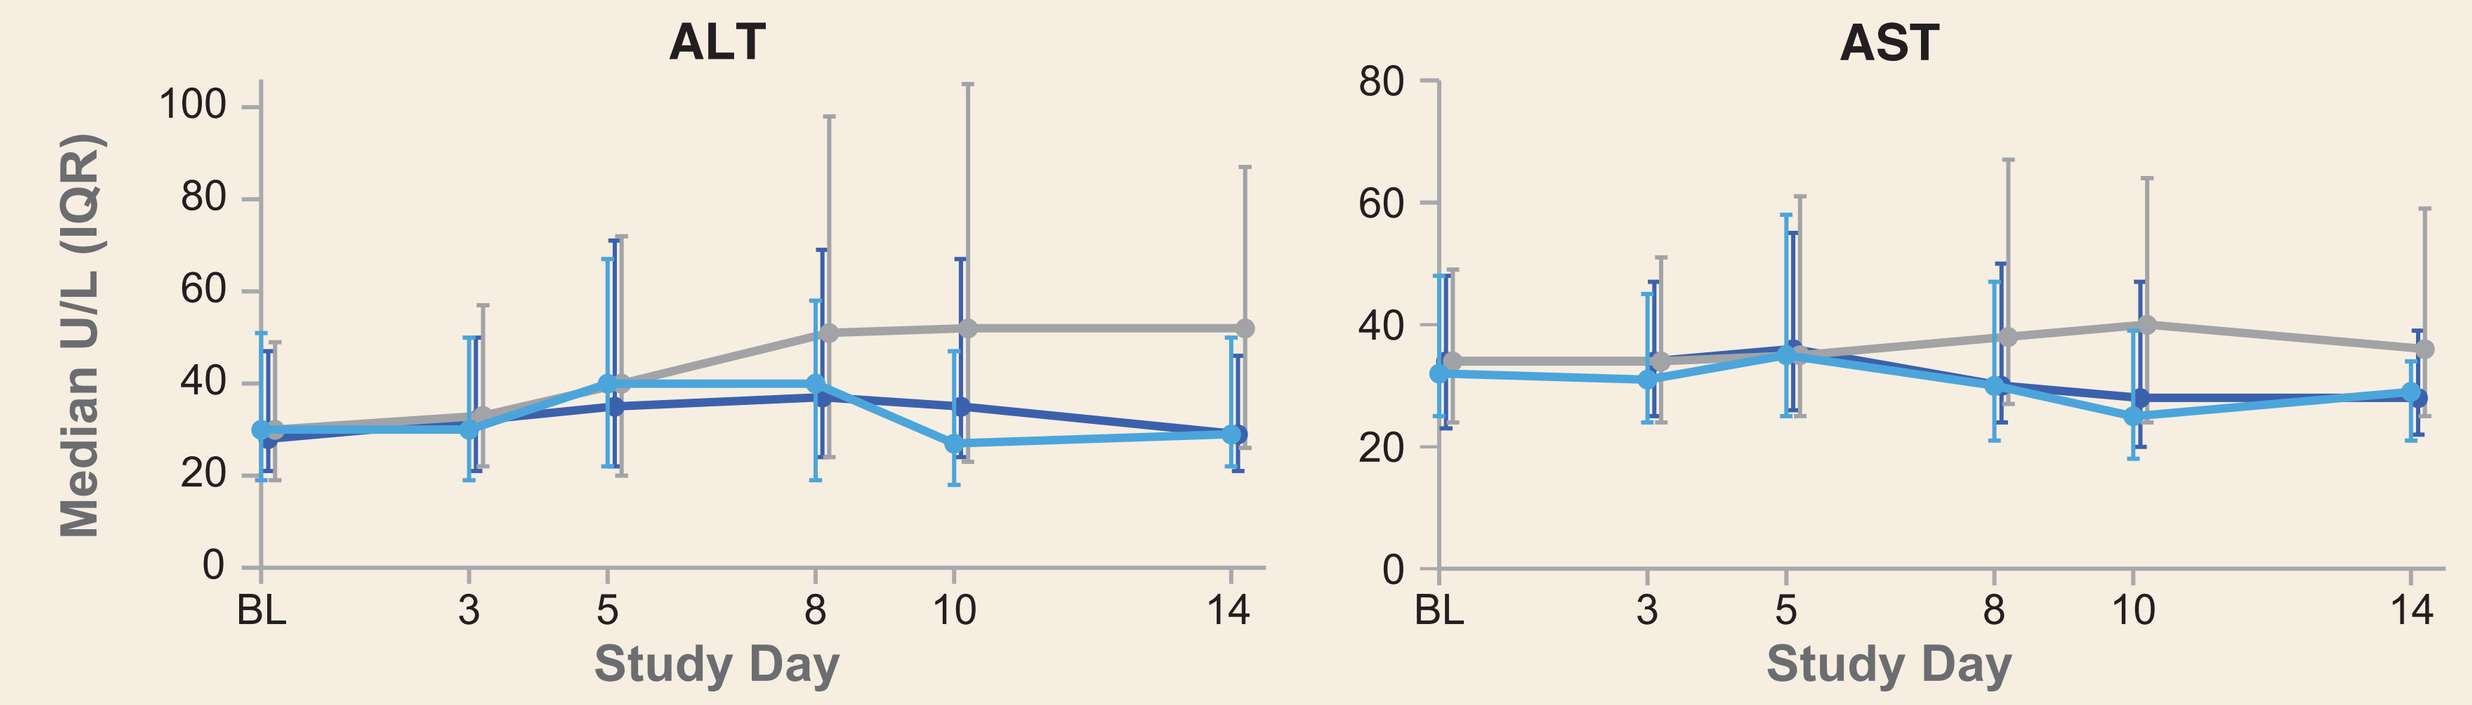

ALT
AST
Median U/L (IQR)
100
80
60
40
20
0
BL
3
5
8
10
14
Study Day

n=	ALT BL	3	5	8	10	14	AST BL	3	5	8	10	14
5-Day RDV	191	172	152	76	54	42	186	167	148	74	53	42
10-Day RDV	193	169	141	93	81	47	187	164	136	89	75	44
SOC	200	170	131	83	66	47	193	165	131	82	66	48

*Data not scheduled for collection past Day 14.

- There were no significant changes in ALT or AST in patients treated with RDV vs SOC

Conclusions

- RDV given for 5 or 10 days was generally safe and well tolerated in patients with moderate COVID-19
 - Renal-related AEs and laboratory abnormalities occurred in similar or higher numbers of patients treated with SOC vs RDV
 - Liver-related laboratory abnormalities occurred in similar or higher numbers of patients receiving SOC vs RDV
- No clinically relevant safety signals were observed in patients receiving RDV

References: **1.** Pizzorno A, et al. Antiviral Res 2020;181:104878; **2.** Sheahan TP, et al. Sci Transl Med. 2017;9:eaal3653; **3.** Williamson BN, et al. Nature 2020;585:273-6; **4.** Spinner CD, et al. JAMA 2020 Aug 21;e2016349.

Acknowledgments: We express our solidarity with those who are or have been ill with COVID-19, their families, and the healthcare workers on the frontlines of this pandemic. We extend our thanks and appreciation to the GS-US-540-5774 study participants, their partners and families, the frontline healthcare workers caring for them, the study staff, and the study investigators. This study was funded by Gilead Sciences, Inc.

Presented at IDWeek 2020, October 21–25, 2020

©2020 Gilead Sciences, Inc. All rights reserved.