Ubrogepant is Effective in the Acute Treatment of Migraine with Mild Pain

Richard B. Lipton,[1] David W. Dodick,[2] Peter J. Goadsby,[3] Rami Burstein,[4] Aubrey M. Adams,[5] Jeff Lai,[6] Sung Yun Yu,[6] Michelle Finnegan,[6] Joel M. Trugman[6]

[1]Albert Einstein College of Medicine and Montefiore Headache Center, Bronx, NY, USA; [2]Mayo Clinic, Phoenix, AZ, USA; [3]NIHR-Wellcome Trust King's Clinical Research Facility, King's College London, UK; [4]Harvard Medical School, Beth Israel Deaconess Medical Center, Boston, MA, USA; [5]AbbVie Inc., Irvine, CA, USA; [6]AbbVie Inc., Madison, NJ, USA

CONCLUSIONS

Treating migraine patients with ubrogepant when headaches are mild rather than moderate/severe doubles the likelihood of rendering them free of pain at 2 hours; associated symptom free rates are also substantially higher when headaches are treated during mild pain

RESULTS

Baseline Demographics and Clinical Characteristics

- A total of 1254 participants were randomized into the long-term extension trial
- The analysis population included 808 participants randomized to ubrogepant 50 mg (n=401) and ubrogepant 100 mg (n=407)
- Demographics were consistent with the typically reported migraine population (**Table 1**)

Table 1. Baseline Participant Demographics

	Ubrogepant 50 mg (N=401)	Ubrogepant 100 mg (N=407)
Age (years), mean (SD)	42.4 (12.2)	41.5 (11.2)
Female, n (%)	370 (92.3)	364 (89.4)
White, n (%)	347 (86.5)	336 (82.6)
BMI (kg/m^2), mean (SD)	29.4 (7.3)	30.1 (7.9)

Number of Treated Attacks

- Throughout the trial 21,454 migraine attacks were treated with 31,968 doses of ubrogepant
 - Ubrogepant 50 mg: 404 participants treated 10,323 migraine attacks with ≥1 dose for a total of 15,536 doses
 - Ubrogepant 100 mg: 409 participants treated 11,131 migraine attacks with ≥1 dose for a total of 16,432 doses
 - Average number of doses taken over 1-year trial period was 38.5 and 40.2 doses of ubrogepant 50 mg and ubrogepant 100 mg, respectively
- The analysis population consists of data from 19,255 migraine attacks treated with ubrogepant 50 mg or ubrogepant 100 mg (**Table 2**)

Table 2. Number of Treated Attacks by Headache Status (Analysis Population)

	Ubrogepant 50 mg	Ubrogepant 100 mg
All treated attacks, n (%)	n=9299	n=9956
Mild pain	1994 (21.4)	1911 (19.2)
Moderate/severe pain	7305 (78.6)	8045 (80.8)
First treated attack, n (%)	n=401	n=407
Mild pain	65 (16.2)	57 (14.0)
Moderate/severe pain	336 (83.8)	350 (86.0)

Efficacy in Mild Pain

- Across all efficacy endpoints reported, average percentage of treated attacks reporting freedom from pain or symptoms was significantly higher for attacks treated when pain was mild compared to those treated when pain was moderate or severe (**Figures 1-4**)
- Data for first treated attacks trended similarly

Figure 1. Pain Freedom at 2 Hours by Headache Status Across All Treated Attacks

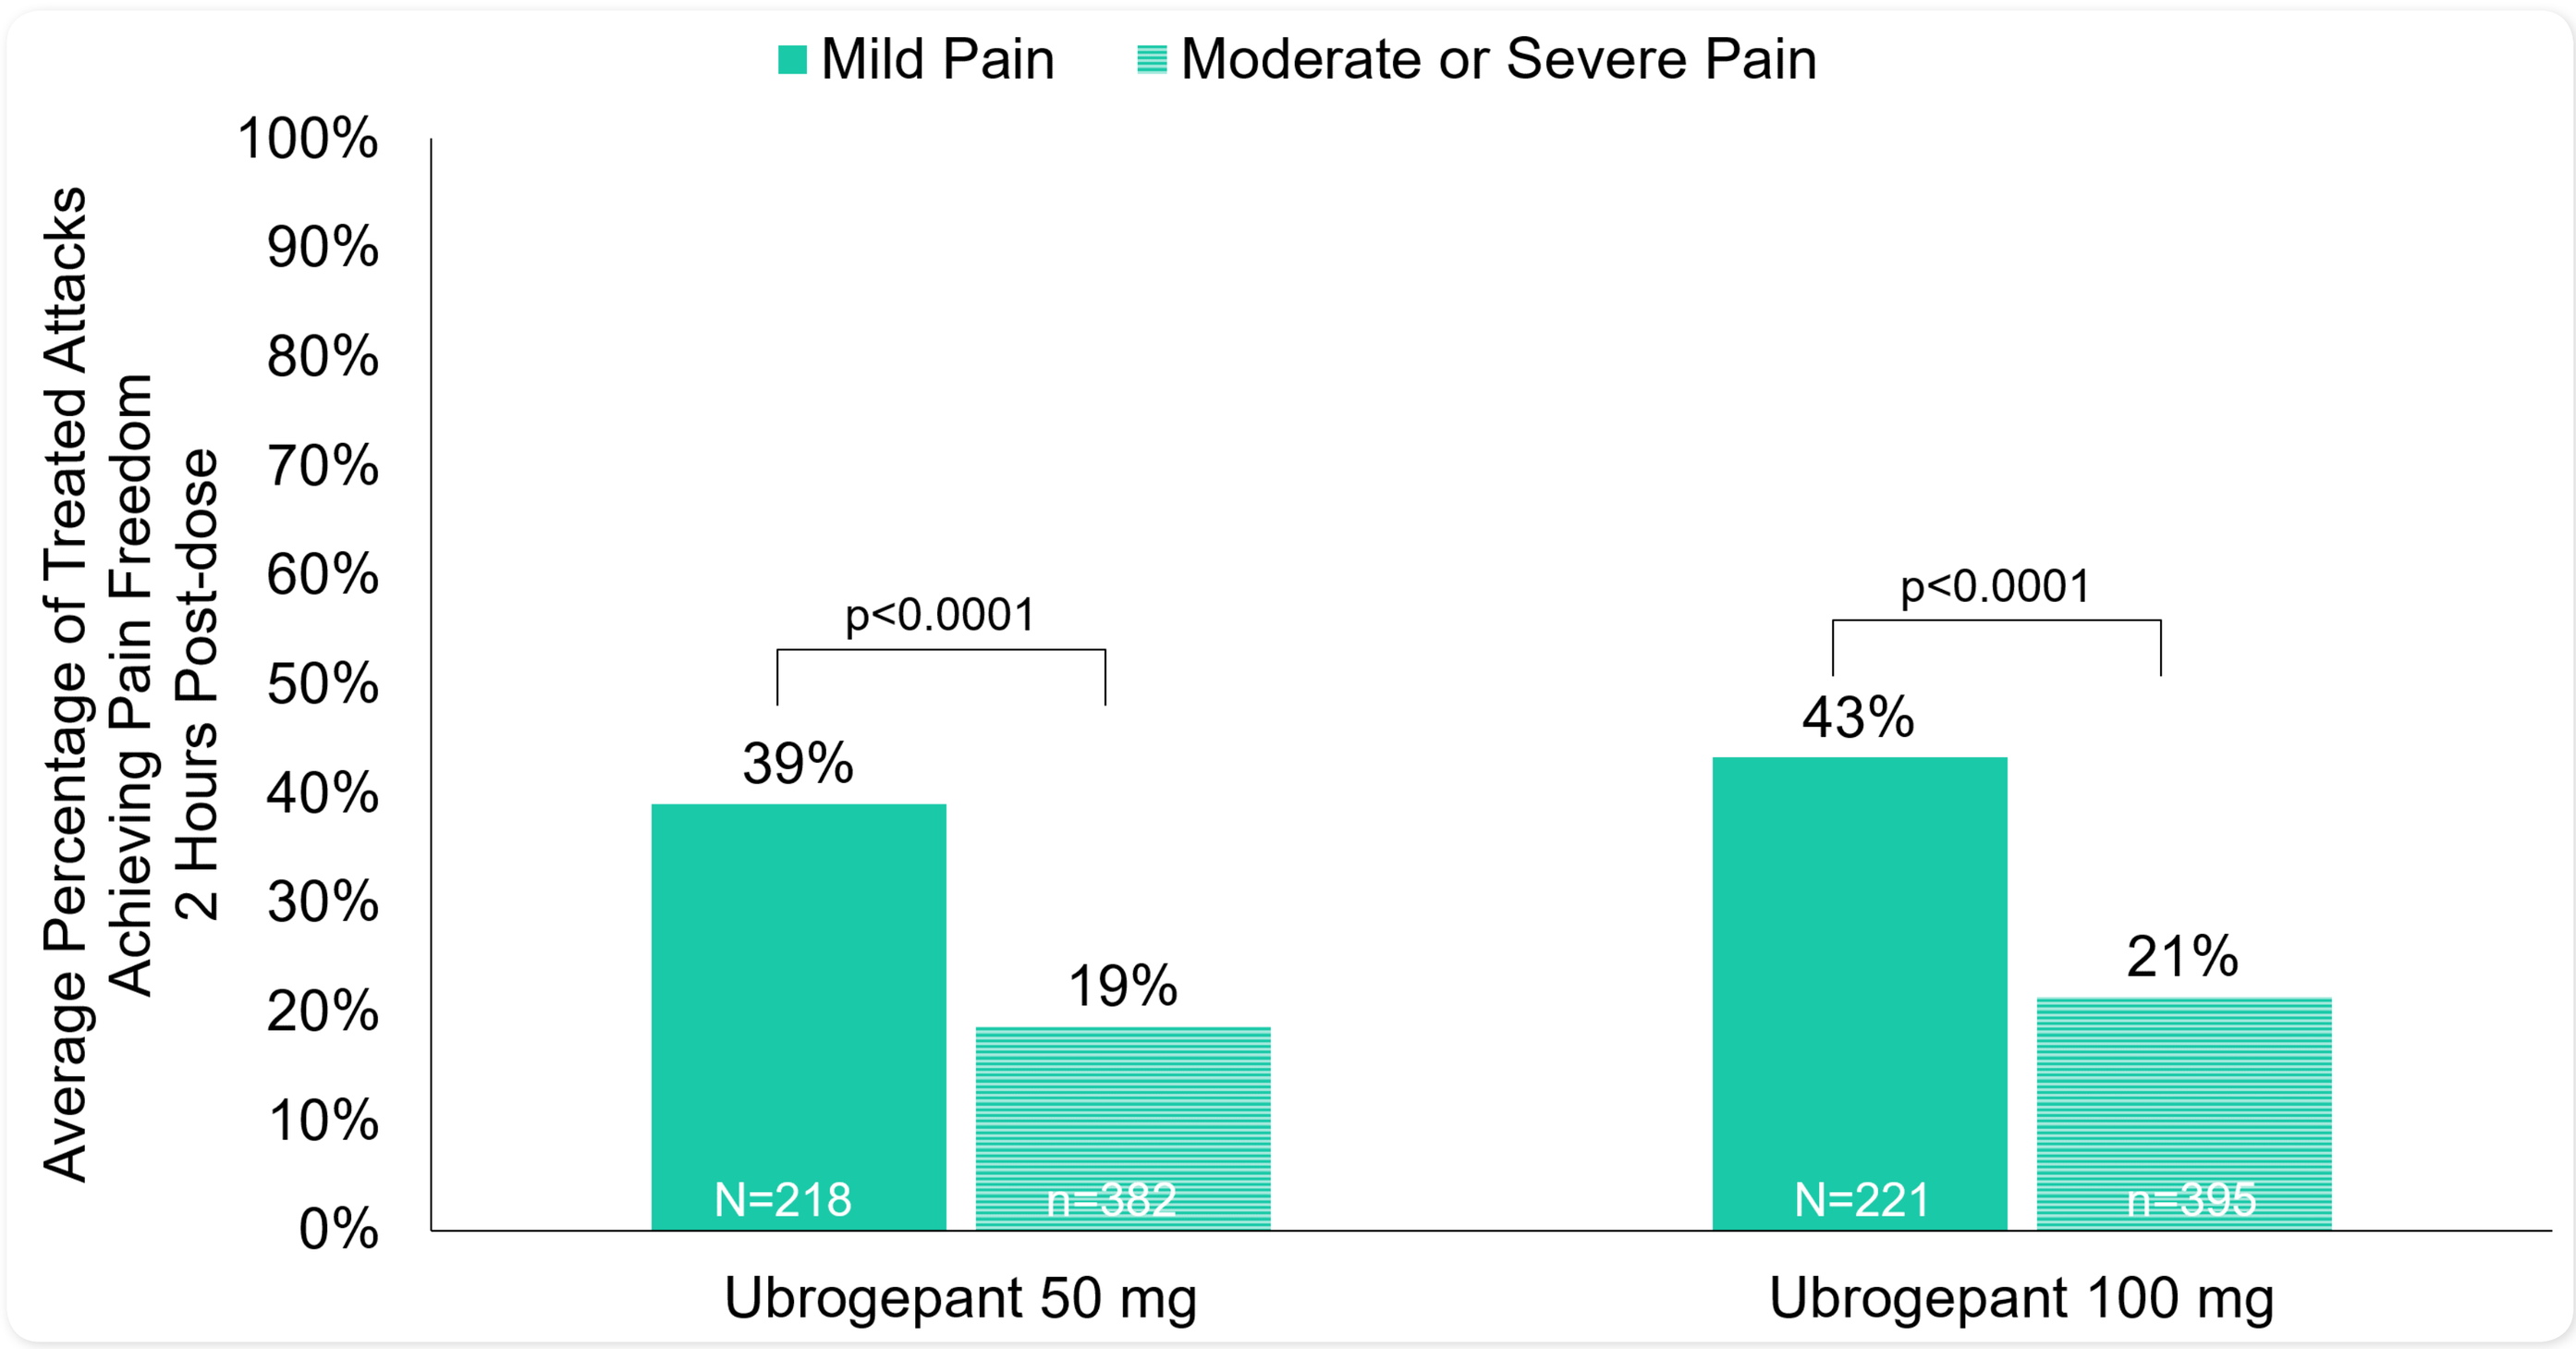
Mild Pain
Moderate or Severe Pain
Average Percentage of Treated Attacks Achieving Pain Freedom 2 Hours Post-dose
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
p<0.0001
39%
19%
N=218
n=382
p<0.0001
43%
21%
N=221
n=395
Ubrogepant 50 mg
Ubrogepant 100 mg

Analysis population is defined as all randomized patients who received at least 1 dose of IP (ubrogepant) and had at least 1 posttreatment efficacy assessment in the trial.
Pain Freedom (PF) is a reduction in headache severity from mild/moderate/severe at baseline to no headache pain.
All available data are used in the analysis. Multiple treated attacks of a subject may contribute multiple responder status

Figure 2. Absence of Phonophobia at 2 Hours by Headache Status Across All Treated Attacks

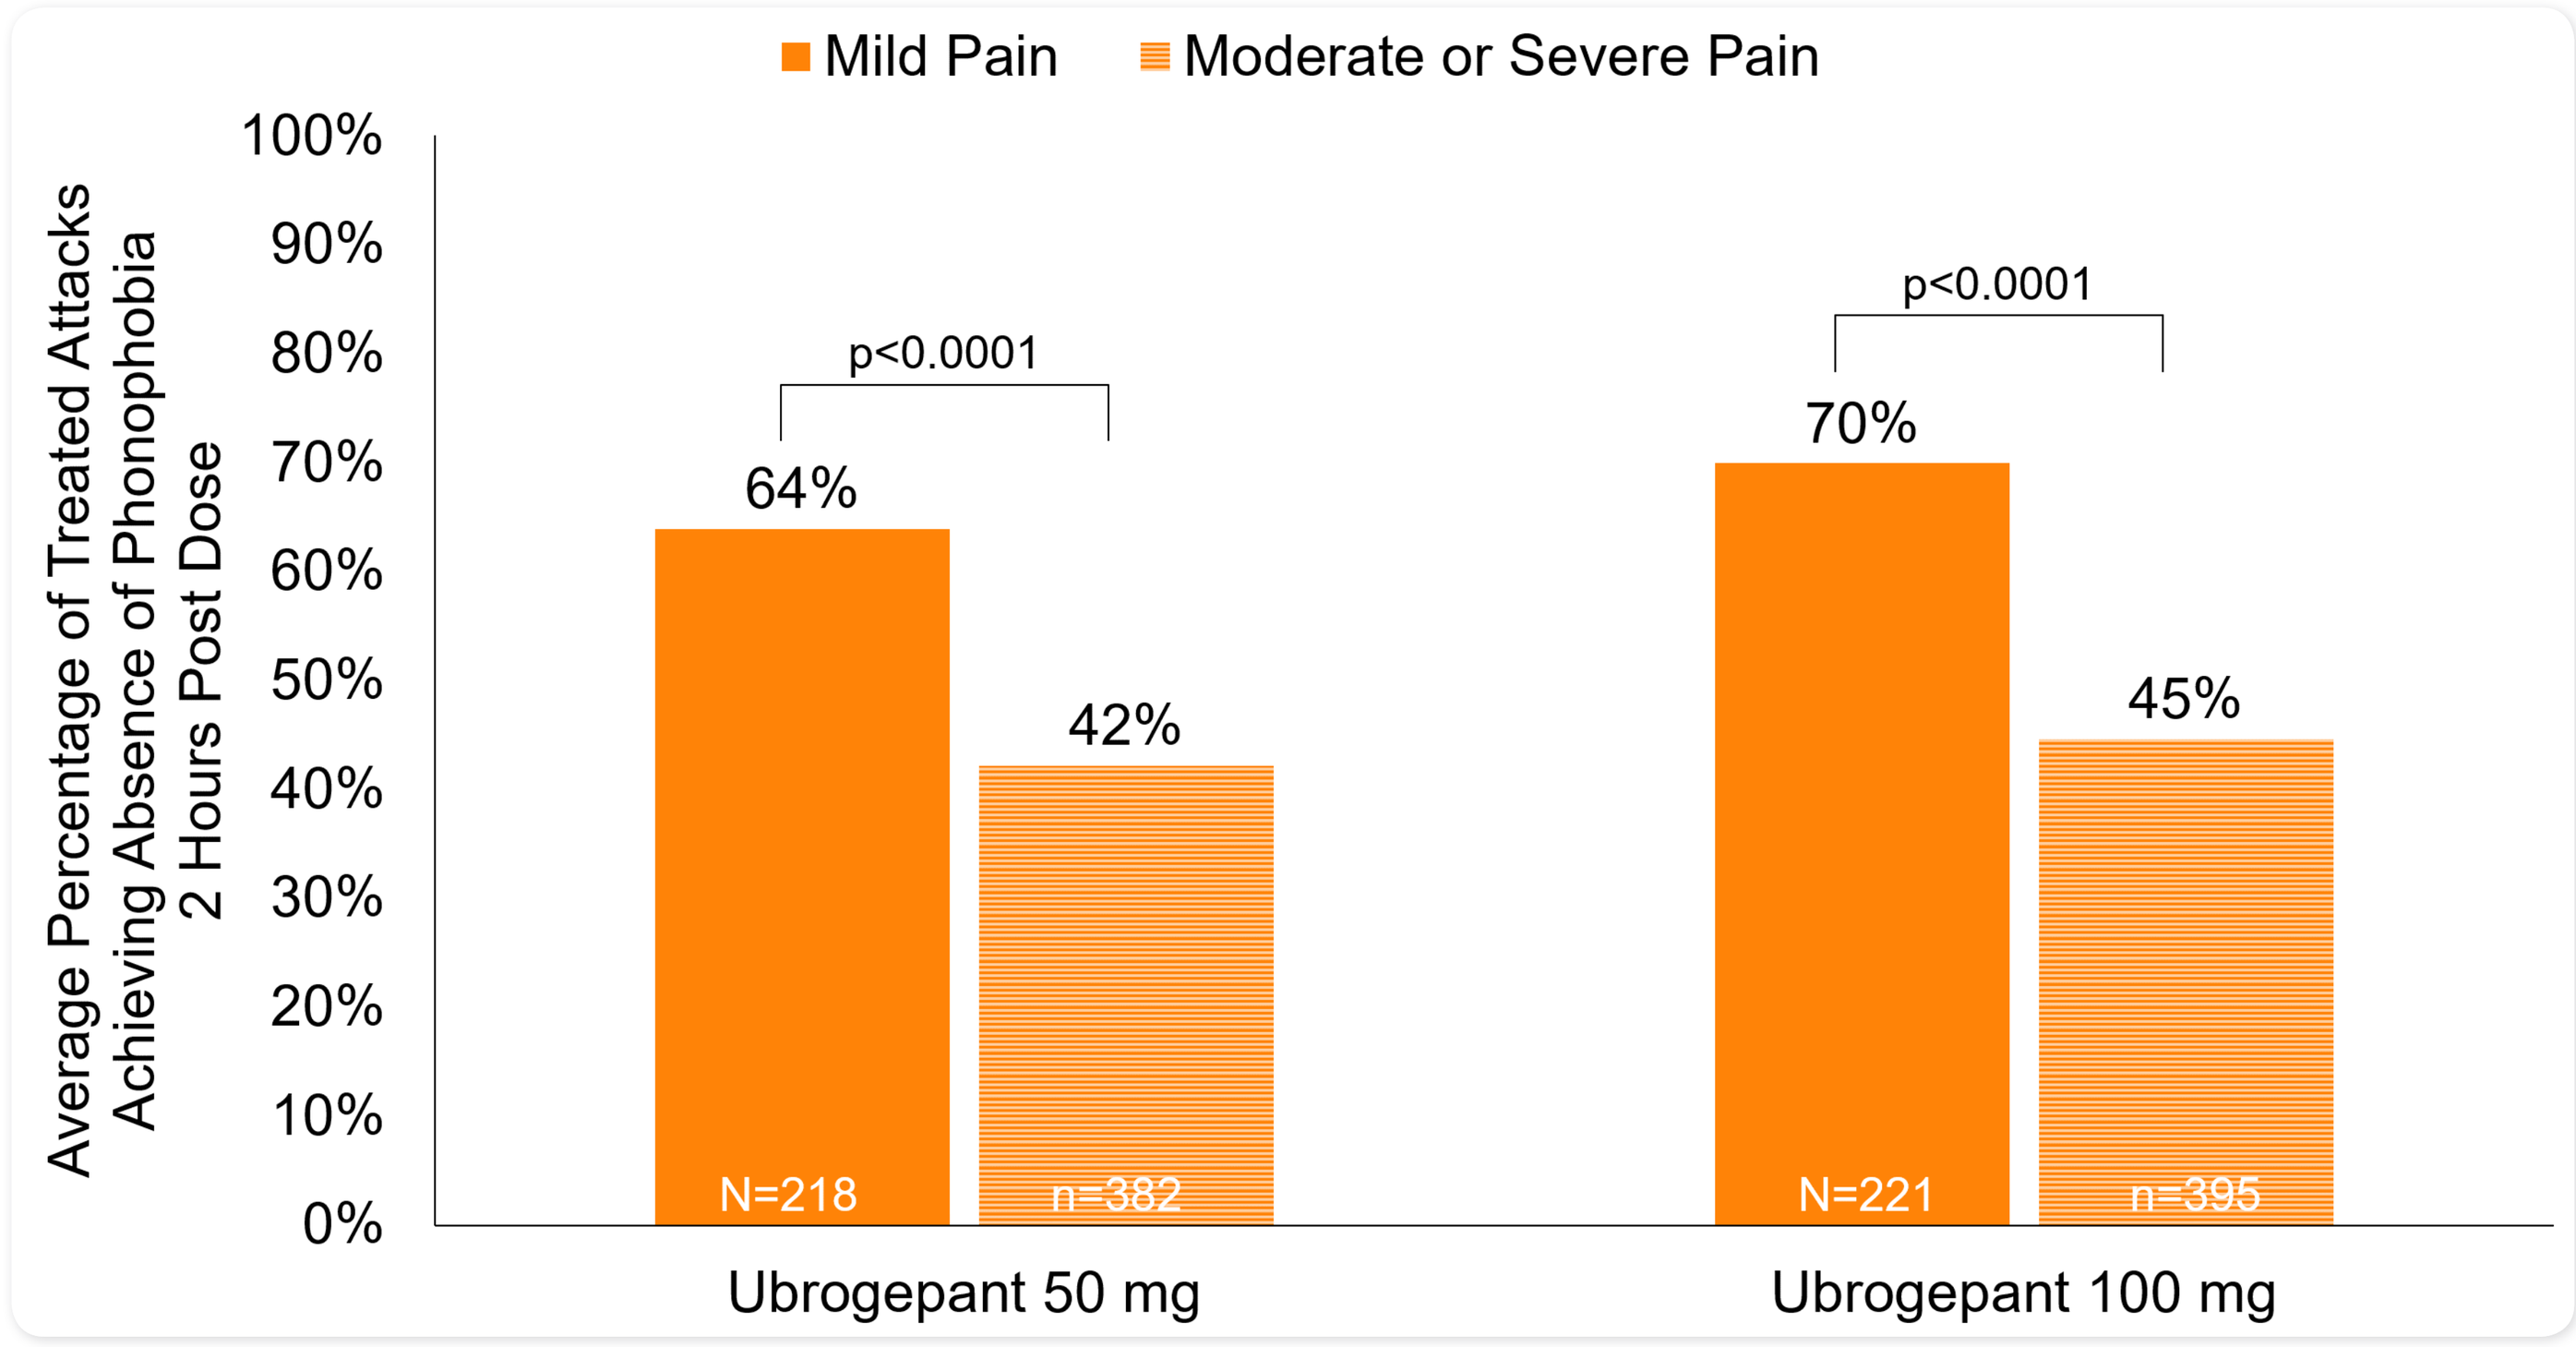
Mild Pain
Moderate or Severe Pain
Average Percentage of Treated Attacks Achieving Absence of Phonophobia 2 Hours Post Dose
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
p<0.0001
64%
42%
N=218
n=382
p<0.0001
70%
45%
N=221
n=395
Ubrogepant 50 mg
Ubrogepant 100 mg

Analysis population is defined as all randomized patients who received at least 1 dose of IP (ubrogepant) and had at least 1 posttreatment efficacy assessment in the trial.
Absence of symptom includes data for all treated attacks regardless of presence/absence of the symptom for the treated attack.
All available data are used in the analysis. Multiple treated attacks of a subject may contribute multiple responder status

Figure 3. Absence of Photophobia at 2 Hours by Headache Status Across All Treated Attacks

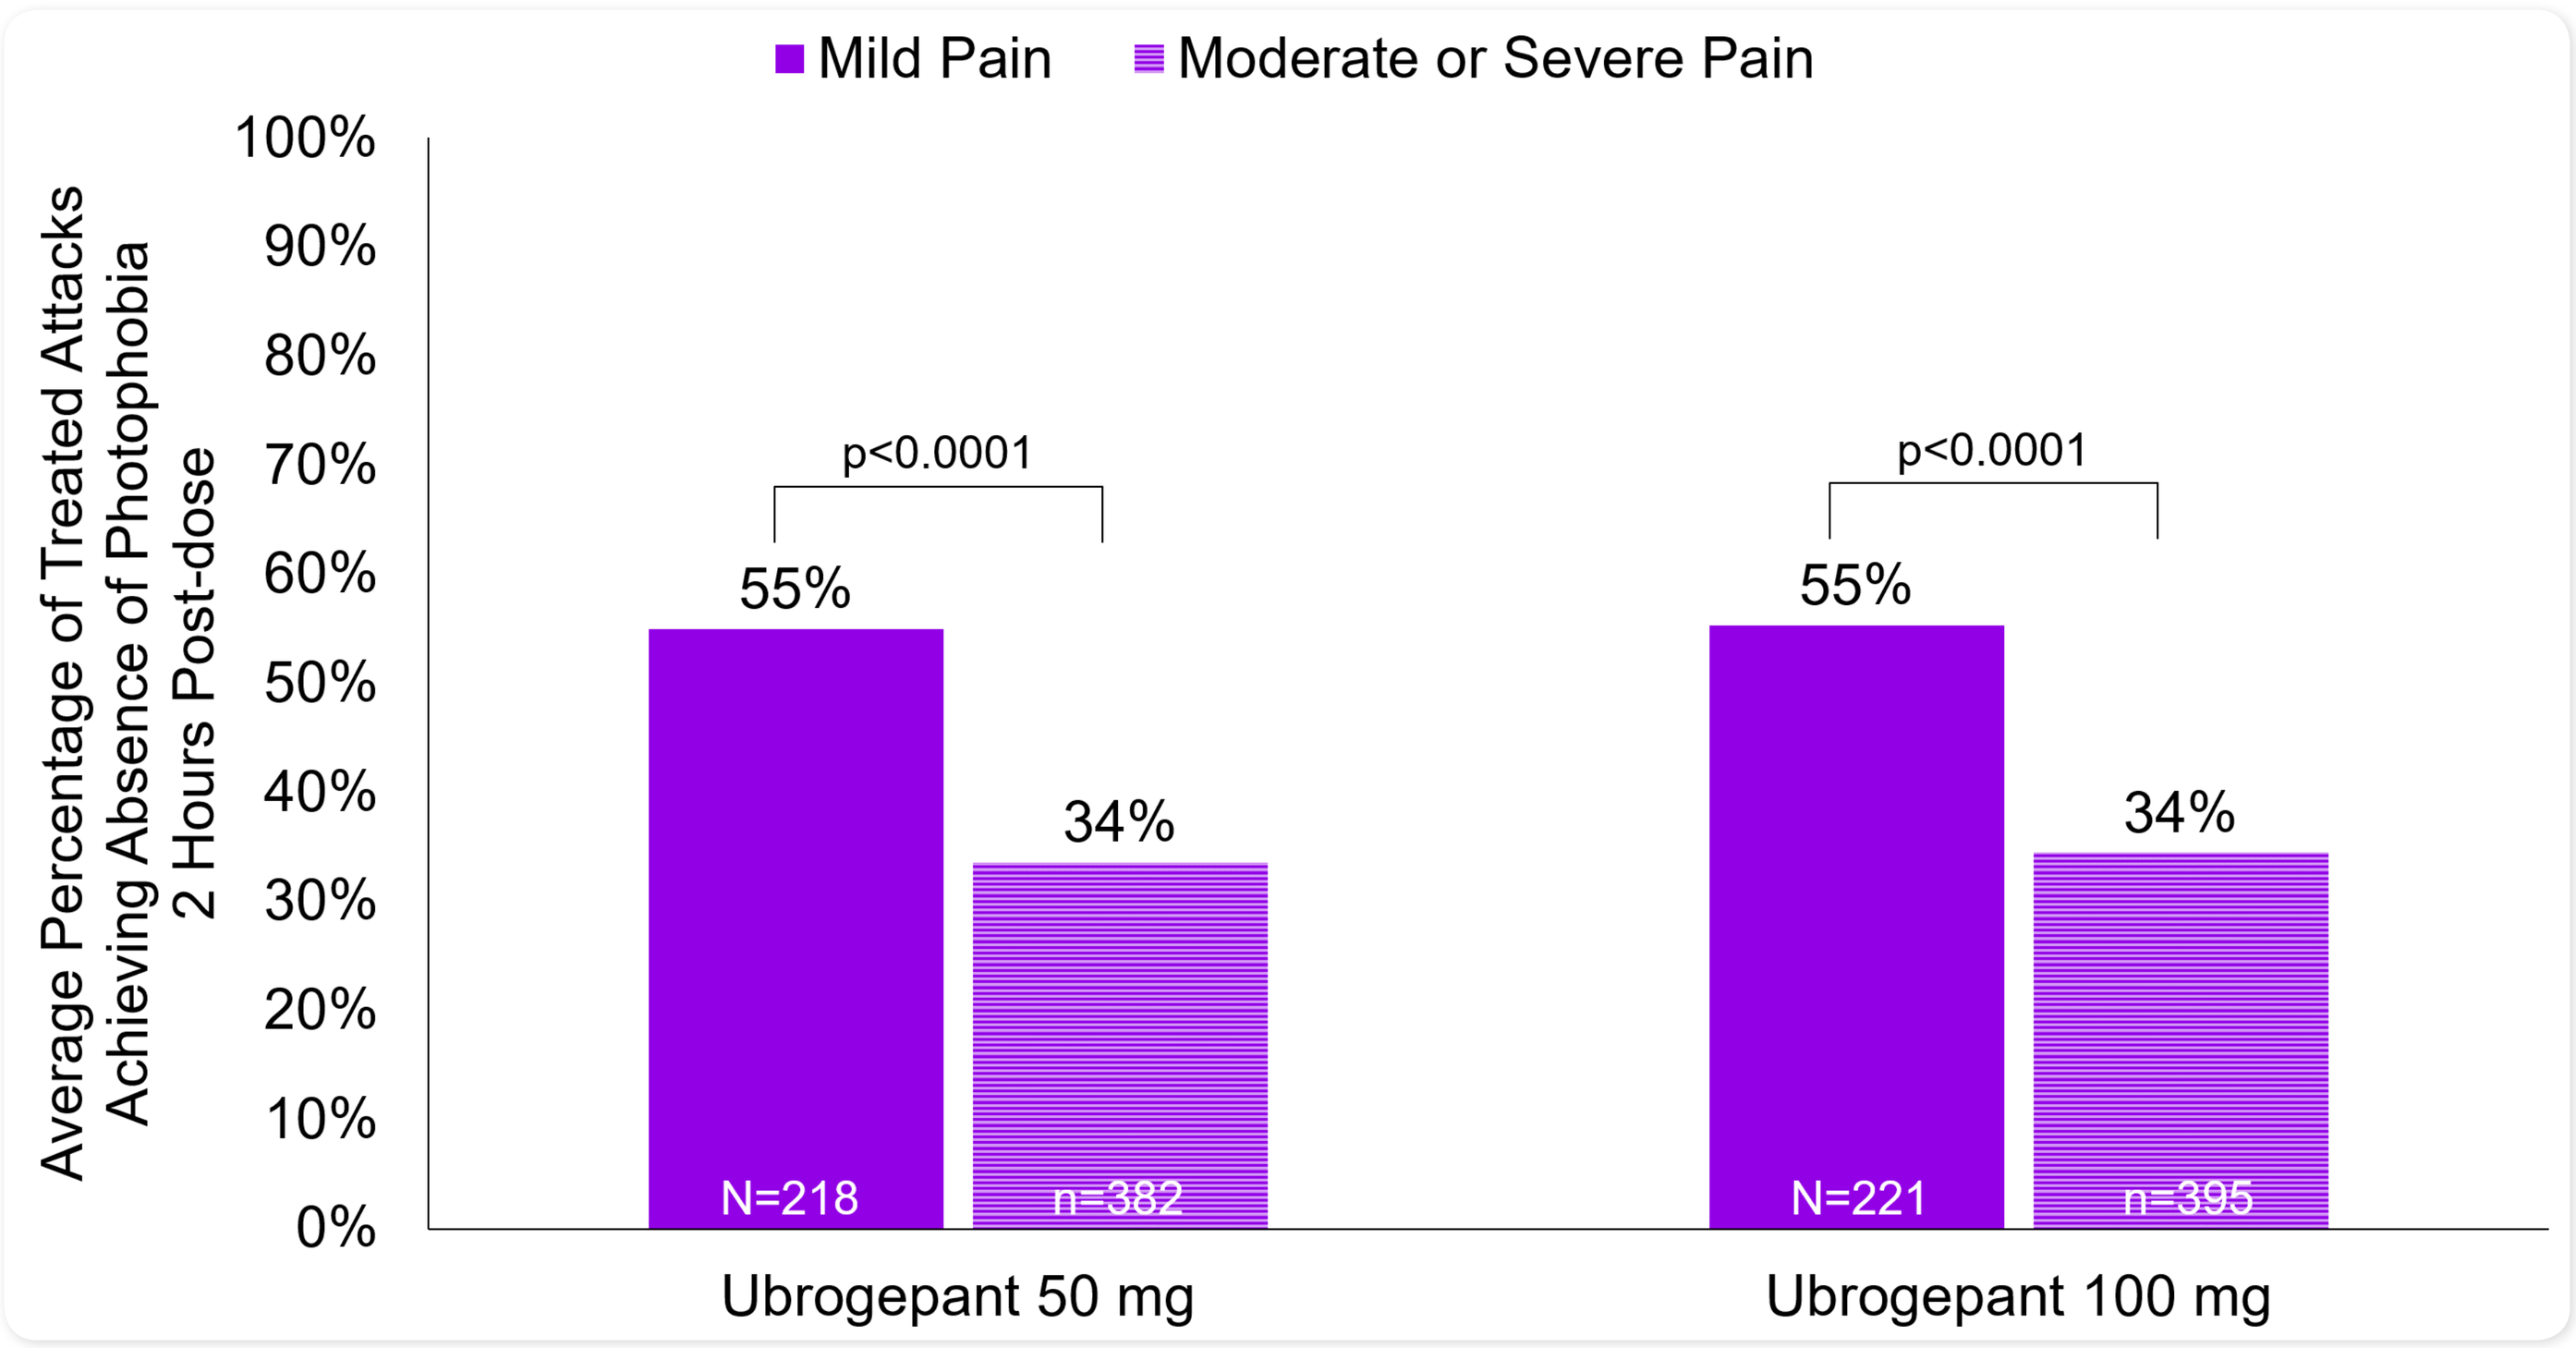
Mild Pain
Moderate or Severe Pain
Average Percentage of Treated Attacks Achieving Absence of Photophobia 2 Hours Post-dose
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
p<0.0001
55%
34%
N=218
n=382
p<0.0001
55%
34%
N=221
n=395
Ubrogepant 50 mg
Ubrogepant 100 mg

Analysis population is defined as all randomized patients who received at least 1 dose of IP (ubrogepant) and had at least 1 posttreatment efficacy assessment in the trial.
Absence of symptom includes data for all treated attacks regardless of presence/absence of the symptom for the treated attack.
All available data are used in the analysis. Multiple treated attacks of a subject may contribute multiple responder status

Figure 4. Absence of Nausea at 2 Hours by Headache Status Across All Treated Attacks

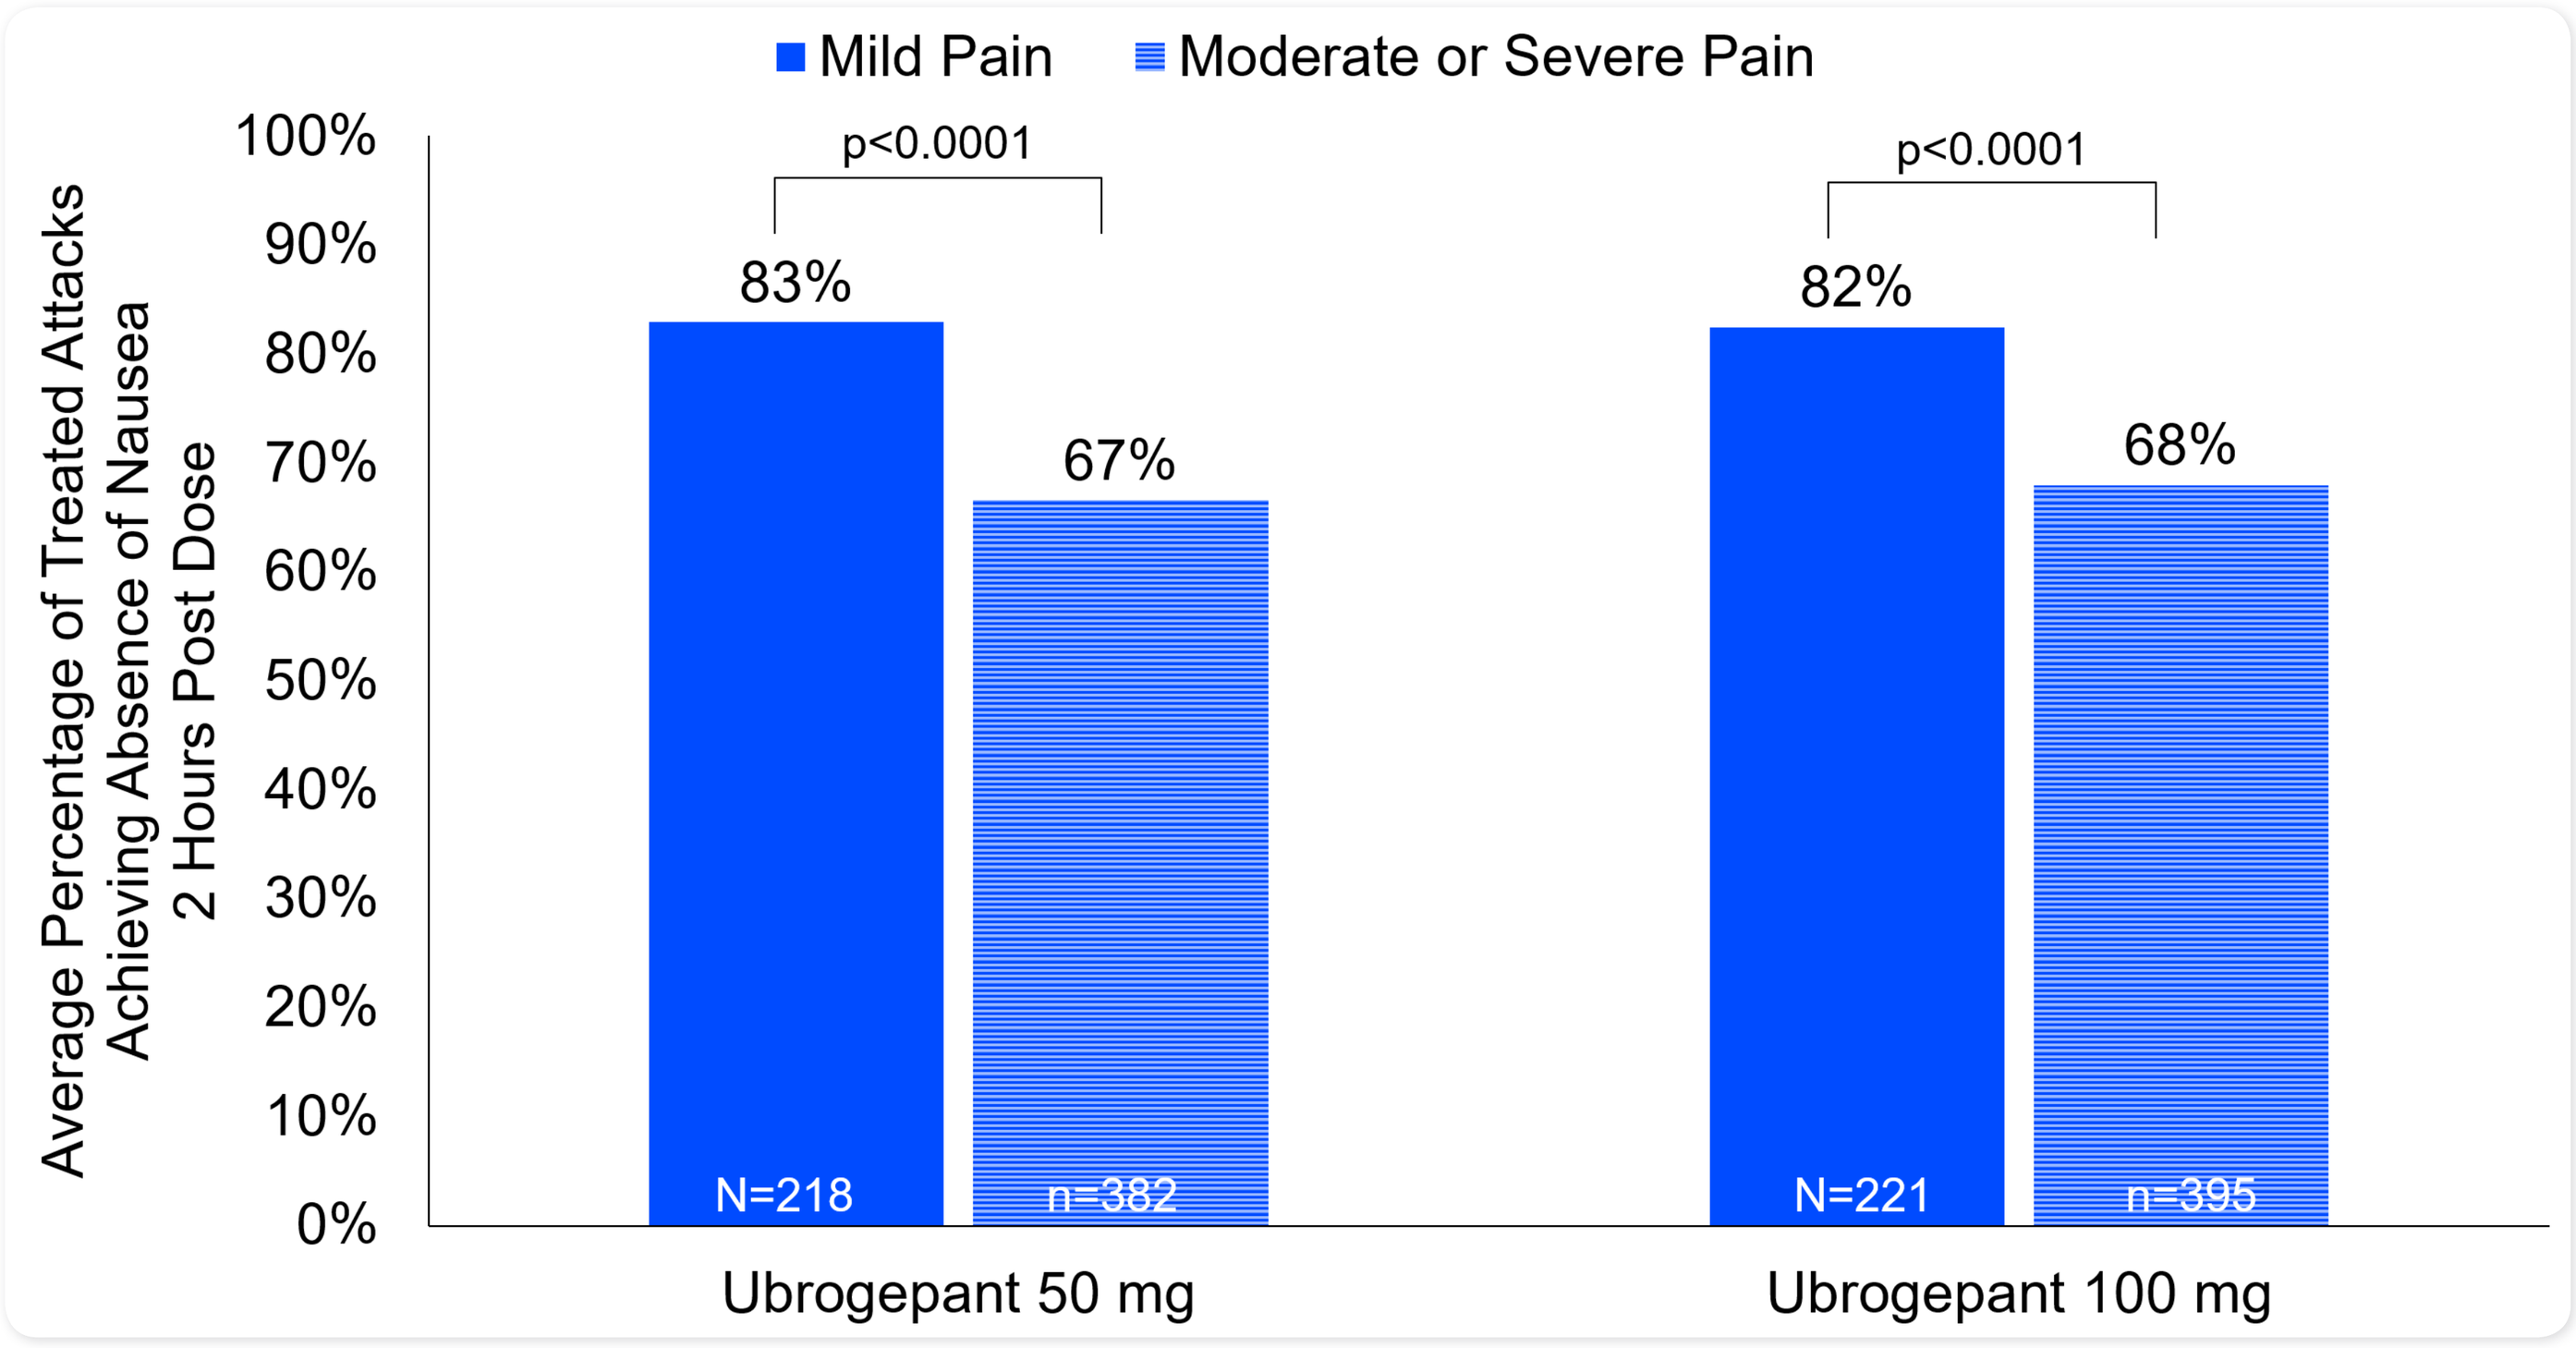
Mild Pain
Moderate or Severe Pain
Average Percentage of Treated Attacks Achieving Absence of Nausea 2 Hours Post Dose
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
p<0.0001
83%
67%
N=218
n=382
p<0.0001
82%
68%
N=221
n=395
Ubrogepant 50 mg
Ubrogepant 100 mg

Analysis population is defined as all randomized patients who received at least 1 dose of IP (ubrogepant) and had at least 1 posttreatment efficacy assessment in the trial.
Absence of symptom includes data for all treated attacks regardless of presence/absence of the symptom for the treated attack.
All available data are used in the analysis. Multiple treated attacks of a subject may contribute multiple responder status

Summary of Ubrogepant Adverse Events

- Treatment-emergent adverse events (TEAEs) were reported by n=268 participants (66%) in the ubrogepant 50 mg group and n=297 participants (73%) in ubrogepant 100 mg
 - Most commonly reported was upper respiratory tract infection (**Table 3**)
- Treatment-related TEAE were reported by n=42 participants (10%) in ubrogepant 50 mg and n=43 participants (11%) in ubrogepant 100 mg
 - Most commonly reported were nausea, dizziness, and somnolence (all ≤2%)
- Serious TEAEs were reported by n=9 participants (2%) in ubrogepant 50 mg and n=12 participants (3%) in ubrogepant 100 mg; one was considered treatment-related
 - Ubrogepant 50 mg: n=1 (0.2%), exacerbation of sinus tachycardia
 - 41 year old female with medical history of supraventricular tachycardia with ablation; concomitant medications included propranolol, citalopram, and tizanadine
 - Reported to emergency room with racing heart, intermittent chest pain, feelings of stress, feeling flushed and hot, and numbness in left upper extremity
 - Physical exam (normal cardiovascular exam and negative blood work); ECG (normal)
 - The event of exacerbation of sinus tachycardia was treated with increasing propanolol dose and resolved; ubrogepant 50 mg was continued
- Discontinuations due to TEAEs were reported by 2%-3%; no deaths occurred during the trial

Table 3. Common (≥2%) Adverse Events by Preferred Term

Preferred Term, n (%)	Ubrogepant 50 mg (N=404)	Ubrogepant 100 mg (N=409)
Nasopharyngitis	33 (8.2)	47 (11.5)
Upper respiratory tract infection	47 (11.6)	44 (10.8)
Sinusitis	28 (6.9)	26 (6.4)
Urinary tract infection	22 (5.4)	26 (6.4)
Influenza	17 (4.2)	25 (6.1)
Nausea	19 (4.7)	19 (4.6)
Bronchitis	13 (3.2)	18 (4.4)
Blood creatine phosphokinase increased	10 (2.5)	16 (3.9)
Alanine aminotransferase increased	9 (2.2)	15 (3.7)
Aspartate aminotransferase increased	7 (1.7)	14 (3.4)
Back pain	14 (3.5)	14 (3.4)
Nephrolithiasis	4 (1.0)	13 (3.2)
Dizziness	5 (1.2)	12 (2.9)
Anxiety	6 (1.5)	11 (2.7)
Cough	8 (2.0)	11 (2.7)
Diarrhea	10 (2.5)	11 (2.7)
Arthralgia	11 (2.7)	10 (2.4)
Gastroenteritis viral	7 (1.7)	10 (2.4)
Muscle strain	2 (0.5)	10 (2.4)
Depression	1 (3.2)	5 (1.2)
Epididymitis*	1 (3.2)	0 (0)
Testicular pain*	1 (3.2)	0 (0)

Only includes TEAEs reported in ≥2% of participants (after rounding) in any treatment group; AE dictionary: MedDRA Version 20.1
* For sex-specific AEs, percentages are relative to the number of participants of the appropriate sex.

INTRODUCTION

Background

- Ubrogepant demonstrated efficacy in treating migraine with moderate/severe pain in two phase 3 trials
- Clinical guidance recommends treatment of migraine when pain is mild, a strategy not previously studied for ubrogepant

Objective

- To evaluate the efficacy of ubrogepant in treating migraine attacks with mild pain

METHODS

Study Design

- Phase 3, open-label, 52-week extension trial (NCT02873221)
- Participants must have completed one of the lead-in trials:
 - ACHIEVE I, NCT02828020
 - ACHIEVE II, NCT02867709
- The trial included adults with a history of migraine with or without aura
- Participants were randomized 1:1:1 to usual care or in blinded fashion to ubrogepant 50 mg or ubrogepant 100 mg
- Participants were allowed to treat up to 8 migraine attacks of any pain severity within each 4 week time period over the course of 1 year

Efficacy Measures

- Efficacy measures were only collected for ubrogepant treatment groups and included
 - 2-hour pain freedom
 - 2-hour absence photophobia
 - 2-hour absence of phonophobia
 - 2-hour absence of nausea

Statistical Analysis

- Data were analyzed in two ways
 - Across all treated attacks, where data were averaged for each participant and then across participants, weighting individuals equally
 - For first treated attack

ADDL INFO

Thank you to all the participants and investigators who participated in this trial!

AUTHOR DISCLOSURES

This study was sponsored by Allergan plc, Dublin, Ireland (prior to acquisition by AbbVie Inc.). Writing and editorial assistance was provided to the authors by Amy Kuang, PhD of AbbVie Inc.. All authors met the ICMJE authorship criteria. Neither honoraria nor payments were made for authorship. Financial arrangements of the authors with companies whose products may be related to the present report are listed as declared by the authors.

RB Lipton reports the following conflicts from within the past 48 months: serves on the editorial boards of Neurology and Cephalalgia and as senior advisor to Headache. He has received research support from the NIH. He also receives support from the Migraine Research Foundation and the National Headache Foundation. He has reviewed for the NIA and NINDS; serves as consultant, advisory board member, or has received grant support or honoraria from Alder, Allergan (now AbbVie), Amgen, Autonomic Technologies, Avanir, Boston Scientific, Dr. Reddy's, Electrocore, Eli Lilly, eNeura Therapeutics, GlaxoSmithKline, Merck, Novartis, Teva, and Vedanta. He receives royalties from Wolff's Headache, 8th Edition, Oxford University Press, 2009, and Informa. He holds stock options in eNeura Therapeutics and Biohaven.

DW Dodick reports consulting: Amgen, Association of Translational Medicine, University Health Network, Daniel Edelman Inc., Autonomic Technologies, Axsome, Aural Analytics, Allergan (now AbbVie), Alder BioPharmaceuticals, Biohaven, Charleston Laboratories, Dr Reddy's Laboratories/Promius, Electrocore LLC, Eli Lilly, eNeura, Neurolief, Novartis, Ipsen, Impel, Satsuma, Supernus, Sun Pharma (India), Theranica, Teva, Vedanta, WL Gore, Nocira, PSL Group Services, University of British Columbia, XoC, Zosano, ZP Opco, Foresite Capital, Oppenheimer; Upjohn (Division of Pfizer), Pieris, Revance, Equinox, Salvia, Amzak Health. CME fees or royalty payments: HealthLogix, Medicom Worldwide, MedLogix Communications, Mednet, Miller Medical, PeerView, WebMD Health/Medscape, Chameleon, Academy for Continued Healthcare Learning, Universal Meeting Management, Haymarket, Global Scientific Communications, Global Life Sciences, Global Access Meetings, UpToDate (Elsevier), Oxford University Press, Cambridge University Press, Wolters Kluwer Health; Stock options: Aural Analytics, Healint, Theranica, Second Opinion/Mobile Health, Epien, GBS/Nocira, Matterhorn/Ontologics, King-Devick Technologies; Consulting without fee: Aural Analytics, Healint, Second Opinion/Mobile Health, Epien; Board of Directors: Epien, Matterhorn/Ontologics, King-Devick Technologies. Patent: 17189376.1-1466:vTitle: Botulinum Toxin Dosage Regimen for Chronic Migraine Prophylaxis without fee; Research funding: American Migraine Foundation, US Department of Defense, PCORI, Henry Jackson Foundation; Professional society fees or reimbursement for travel: American Academy of Neurology, American Brain Foundation, American Headache Society, American Migraine Foundation, International Headache Society, Canadian Headache Society.

PJ Goadsby reports personal fees from Allergan (now AbbVie), during the conduct of the study; personal fees from Alder Biopharmaceuticals, grants and personal fees from Amgen, personal fees from Biohaven Pharmaceuticals Inc., grants from Celgene, personal fees from Clexio, grants and personal fees from Eli Lilly and Company, personal fees from Electrocore LLC, personal fees from eNeura Inc, personal fees from Epalex, personal fees from Impel Neuropharma, personal fees from MundiPharma, personal fees from Novartis, personal fees from Pfizer, personal fees from Santara Therapeutics, personal fees from Teva Pharmaceuticals, other from Trigemina, personal fees from WL Gore, outside the submitted work; In addition, Dr. Goadsby has a patent Magnetic stimulation for headache licensed to eNeura without fee and fees for advice through Gerson Lehrman Group and Guidepoint, and fees for publishing from Oxford University Press, Massachusetts Medical Society, Wolters Kluwer and for medicolegal work.

R Burstein reports grant support, consulting, or advisory board with Alder, Allergan (now AbbVie), Amgen,ATI, Avanir, Biohaven, Depomed, Dr. Reddy Laboratories, Electrocore, Eli Lilly, Johnson&Johnson, Neurorelief, Pernix, Percept, Revance, Teva, Theranica, Trigemina and is an inventor and co-founder of Allay.

A Adams, J Lai, SY Yun, M Finnegan, and J Trugman are full-time employees of AbbVie Inc.

To obtain a PDF of this poster,
- Scan the QR code

OR

- Visit: www.allergancongressposters.com/256981

Charges may apply.
No personal information is stored.

Presentation for the 2020 PainWeek Live Virtual Congress

abbvie